Evaluation of PET-CT Metrics and Pharmacokinetics in Adults Receiving Tislelizumab for Relapsed/Refractory Classical Hodgkin Lymphoma: Ancillary Analyses of LYSA Phase 2 TIRHOL Study BGB-A317-210

PS2028

Hervé Ghesquières,[1] Krimo Bouabdallah,[2] Salim Kanoun,[3] Marc André,[4] Philippe Quittet,[5] Cécile Borel,[6] Aspasia Stamatoullas Bastard,[7] Michael Gilbertson,[8] Fabien Le Bras,[9] Catherine Thieblemont,[10] Baptiste Delapierre,[11] Mohamed Touati,[12] Pierre Feugier,[13] Loïc Renaud,[14] Nadine Morineau,[15] Thomas Gastinne,[16] Isabelle Chaillol,[17] Rod Ramchandren,[18] Harsh Shah,[19] Dipenkumar Modi,[20] Heather Allewelt,[21] Pierre Fustier,[22] Xiangyu Liu,[21] Jun Zhang,[21] Richard Delarue,[22] Franck Morschhauser,[23] Cédric Rossi[24]

[1]Lyon Sud Hospital, Pierre Bénite, France; [2]Hôpital Haut-Lévêque, CHU Bordeaux, Pessac, France; [3]Institut Universitaire du Cancer de Toulouse, Toulouse, France; [4]CHU UCL Namur, Yvoir, Belgium; [5]CHU Montpellier, Montpellier, France; [6]IUCT Oncopole, Toulouse, France; [7]Centre Henri Becquerel, Rouen, France; [8]Monash Health, Melbourne, VIC, Australia; [9]CHU Mondor, Creteil, France; [10]AP-HP, Hôpital Saint-Louis, Hemato-oncology, Paris University Paris Cité, Paris, France; [11]Caen University Hospital, Caen, France; [12]CHU Limoges, Limoges, France; [13]CHU Nancy, Nancy, France; [14]Gustave Roussy, Department of Hematology, Villejuif, France; [15]CHD de Vendee, La Roche Sur Yon, France; [16]CHU de Nantes, Nantes, France; [17]Lymphoma Academic Research Organisation, Lyon, France; [18]UPMC Hillman Cancer Center, Pittsburgh, PA, USA; [19]University of Utah, Salt Lake City, UT, USA; [20]Karmanos Cancer Institute, Wayne State University, Detroit, MI, USA; [21]BeOne Medicines, Ltd, San Carlos, CA, USA; [22]BeOne Medicines, Ltd, Basel, Switzerland; [23]CHU de Lille, Lille, France; [24]CHU Dijon, Dijon, France

CONCLUSIONS

- Tislelizumab is an effective therapeutic option for patients with R/R cHL
- Analyses of centrally reviewed PET-CT imaging demonstrated a trend for underestimation of local response assessments, and therefore in ORR, likely due to the presence of non-lymphomatous inflammatory lesions
- Tislelizumab PK and efficacy outcomes did not differ based on baseline PET-CT metrics
- OS was lower in patients without metabolic response at week 12 compared with patients who had CMR or PMR
- Variations in PET-CT parameters during anti–PD-1 therapy should be interpreted with caution to avoid premature treatment discontinuation

INTRODUCTION

- The phase 2 TIRHOL study (NCT04318080) was designed to evaluate the efficacy of the PD-1 inhibitor tislelizumab in patients with relapsed/refractory (R/R) classical Hodgkin lymphoma (cHL) in Western countries[1]
 - Overall, 45 patients with R/R cHL were treated with tislelizumab every 3 weeks until progressive disease, unacceptable toxicity, or study withdrawal; disease response was assessed by the investigator every 12 weeks per Lugano 2014 positron emission tomography–computed tomography (PET-CT) classification
 - The study met its primary endpoint, with an overall response rate (ORR) of 66.7% (95% CI, 51%-80%) and a complete response rate of 31%
 - Pharmacokinetics (PK) profiles were comparable between BGB-A317-210 and the prior Chinese study BGB-A317-203[2,3]
- Conventional methods using PET-CT to assess lymphoma response are suboptimal in patients treated with PD-1 inhibitors, and specialized criteria are required to prevent premature treatment discontinuation due to inflammation unrelated to disease progression

METHODS

- In this correlative, ancillary analysis of the TIRHOL study, PET-CT imaging was evaluated post hoc by a central, independent reviewer
- The prognostic role of baseline PET-CT metrics was evaluated with correlation of TMTV4, TLG4, Dmax4, SUVmean, SUVmax, and:
 - First and best response assessments (complete metabolic response [CMR], partial metabolic response [PMR], stable disease [SD], and progressive disease [PD])
 - Outcome: progression-free survival (PFS) and overall survival (OS)
- The longitudinal evolution of TMTV4 during tislelizumab treatment was evaluated
- Tislelizumab PK profiles (cycle 1 postdose and cycle 2 predose concentrations) were evaluated against baseline clinical and PET-CT characteristics

RESULTS

Clinical Characteristics

- PET-CT imaging at baseline and during follow-up was available for central review for 37 of 45 patients included in TIRHOL
- Patient clinical characteristics are shown in **Table 1**

Table 1. Patient Clinical Characteristics

Parameters	Central PET-CT cohort n=37	TIRHOL study cohort n=45
Age, median (range), years	53 (18-87)	64 (18-87)
≥45 years, n (%)	25 (67.6)	31 (68.9)
Male, n (%)	26 (70.3)	30 (66.7)
Time since initial diagnosis, median (range), months	23.5 (6-326)	24.7 (6-326)
ECOG PS 1, n (%)	13 (35.1)	17 (37.8)
IPS ≥3, n (%)	17 (47.2)	22 (50.0)
Refractory status at enrollment, n (%)	12 (32.4)	13 (28.9)
No. of prior lines, median (range)	2 (1-4)	2 (1-4)
Prior therapy, n (%)		
Monoclonal antibody	29 (78.4)	34 (75.6)
Chemotherapy	37 (100)	45 (100)
Radiotherapy	7 (18.9)	10 (22.2)
ASCT	12 (32.4)	14 (31.1)
Other anticancer therapy	0	2 (4.4)
Cohort 1 (prior ASCT and BV)	12 (32.4)	14 (31.1)
Cohort 2 (not candidate for ASCT)	25 (67.6)	31 (68.9)

Abbreviations: ASCT, autologous stem cell transplant ; BV, brentuximab vedotin; ECOG PS, Eastern Cooperative Oncology Group performance status; IPS, International Prognostic Score; PET-CT, positron emission tomography–computed tomography.

Tislelizumab PK Profiles

- No differences in PK results were observed based on key baseline categorical variables (sex, age, B-symptoms, inclusion in cohort 1 or 2, Ann Arbor stage, bulk, refractory vs relapsed disease, International Prognostic Score, TMTV4, albumin level) (**Figure 1**)

Figure 1. Tislelizumab Concentration and TMTV4 at Cycle 1 Postdose and Cycle 2 Predose

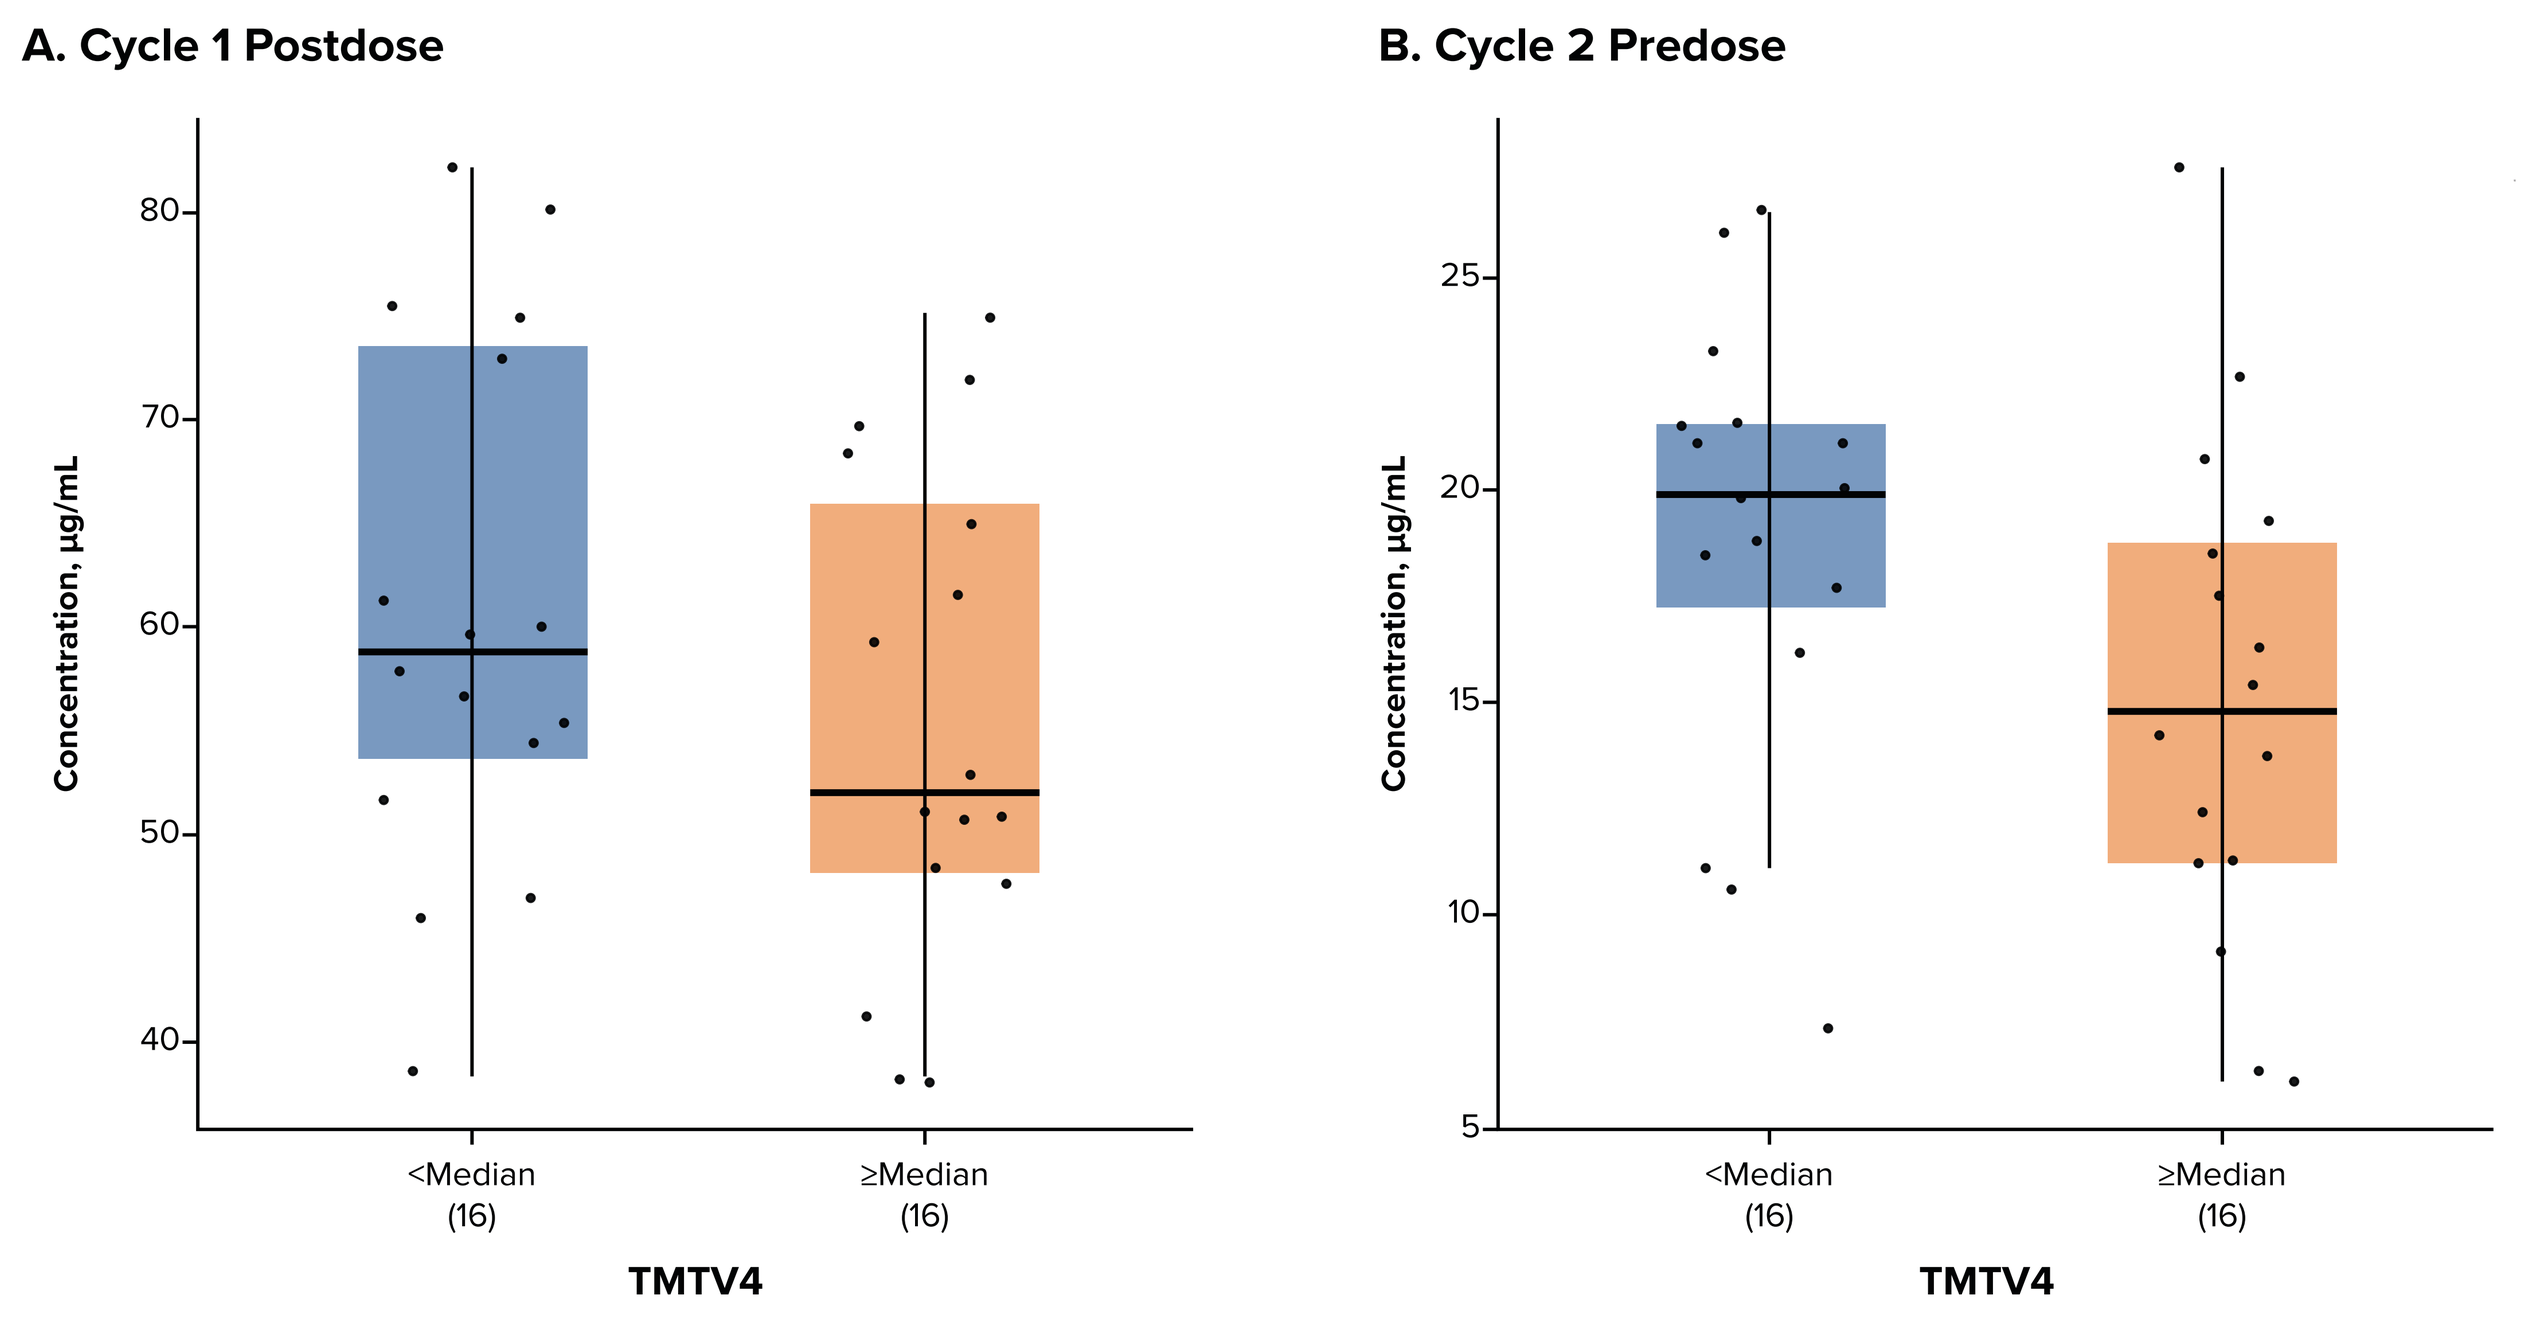
A. Cycle 1 Postdose
B. Cycle 2 Predose
Concentration, µg/mL
<Median (16)
≥Median (16)
TMTV4

Abbreviation: TMTV4, total metabolic tumor volume with standardized uptake value ≥4.

Response Assessments: Local Investigators vs Central Review

- Analyses of PET-CT imaging demonstrated a trend for underestimation of responses assessed by local investigator vs responses by central review in the ORR (66.7% vs 70%) and CMR rate (31% vs 41%), likely due to the presence of non-lymphomatous inflammatory lesions (**Table 2**)
- CMR was achieved in 10 of 45 (22%) included patients at first evaluation at week 12 per local investigator; on central review, the CMR rate was 27% (**Table 3**)

Table 2. Best Response (Primary Endpoint)

n (%)	Best response, local investigators					
Best response, central review	**CMR n=11**	**PMR n=13**	**SD n=1**	**PD n=11**	**NE n=1**	**All n=37**
CMR	10 (90.9)	5 (38.5)	0	0	0	**15 (40.5)**
PMR	1 (9.1)	7 (53.8)	1 (100)	2 (18.2)	0	**11 (29.7)**
SD	0	0	0	1 (9.1)	0	1 (2.7)
PD	0	1 (7.7)	0	8 (72.7)	0	9 (24.3)
NE	0	0	0	0	1 (100)	1 (2.7)

Abbreviations: CMR, complete metabolic response; NE, not evaluated; PD, progressive disease; PMR, partial metabolic response; SD, stable disease.

Table 3. First Response at 12 Weeks

n (%)	First response, local investigators					
First response, central review	**CMR n=9**	**PMR n=11**	**SD n=1**	**PD n=14**	**NE n=1**	**All n=37**
CMR	7 (77.8)	2 (18.2)	1 (50.0)	0	0	**10 (27)**
PMR	2 (22.2)	7 (63.6)	1 (50.0)	3 (21.4)	0	**13 (35)**
SD	0	0	0	1 (7.1)	0	1 (2.7)
PD	0	2 (18.2)	0	10 (71.4)	0	12 (32.4)
NE	0	0	0	0	1 (100)	1 (2.7)

Abbreviations: CMR, complete metabolic response; NE, not evaluated; PD, progressive disease; PMR, partial metabolic response; SD, stable disease.

Baseline PET-CT Metrics and Response

- Median baseline PET-CT metrics were comparable across best metabolic response categories (**Figure 2A-E**)
- Similar results were observed with week 12 metabolic response

Figure 2. Baseline PET-CT Metrics and Best Overall Metabolic Response

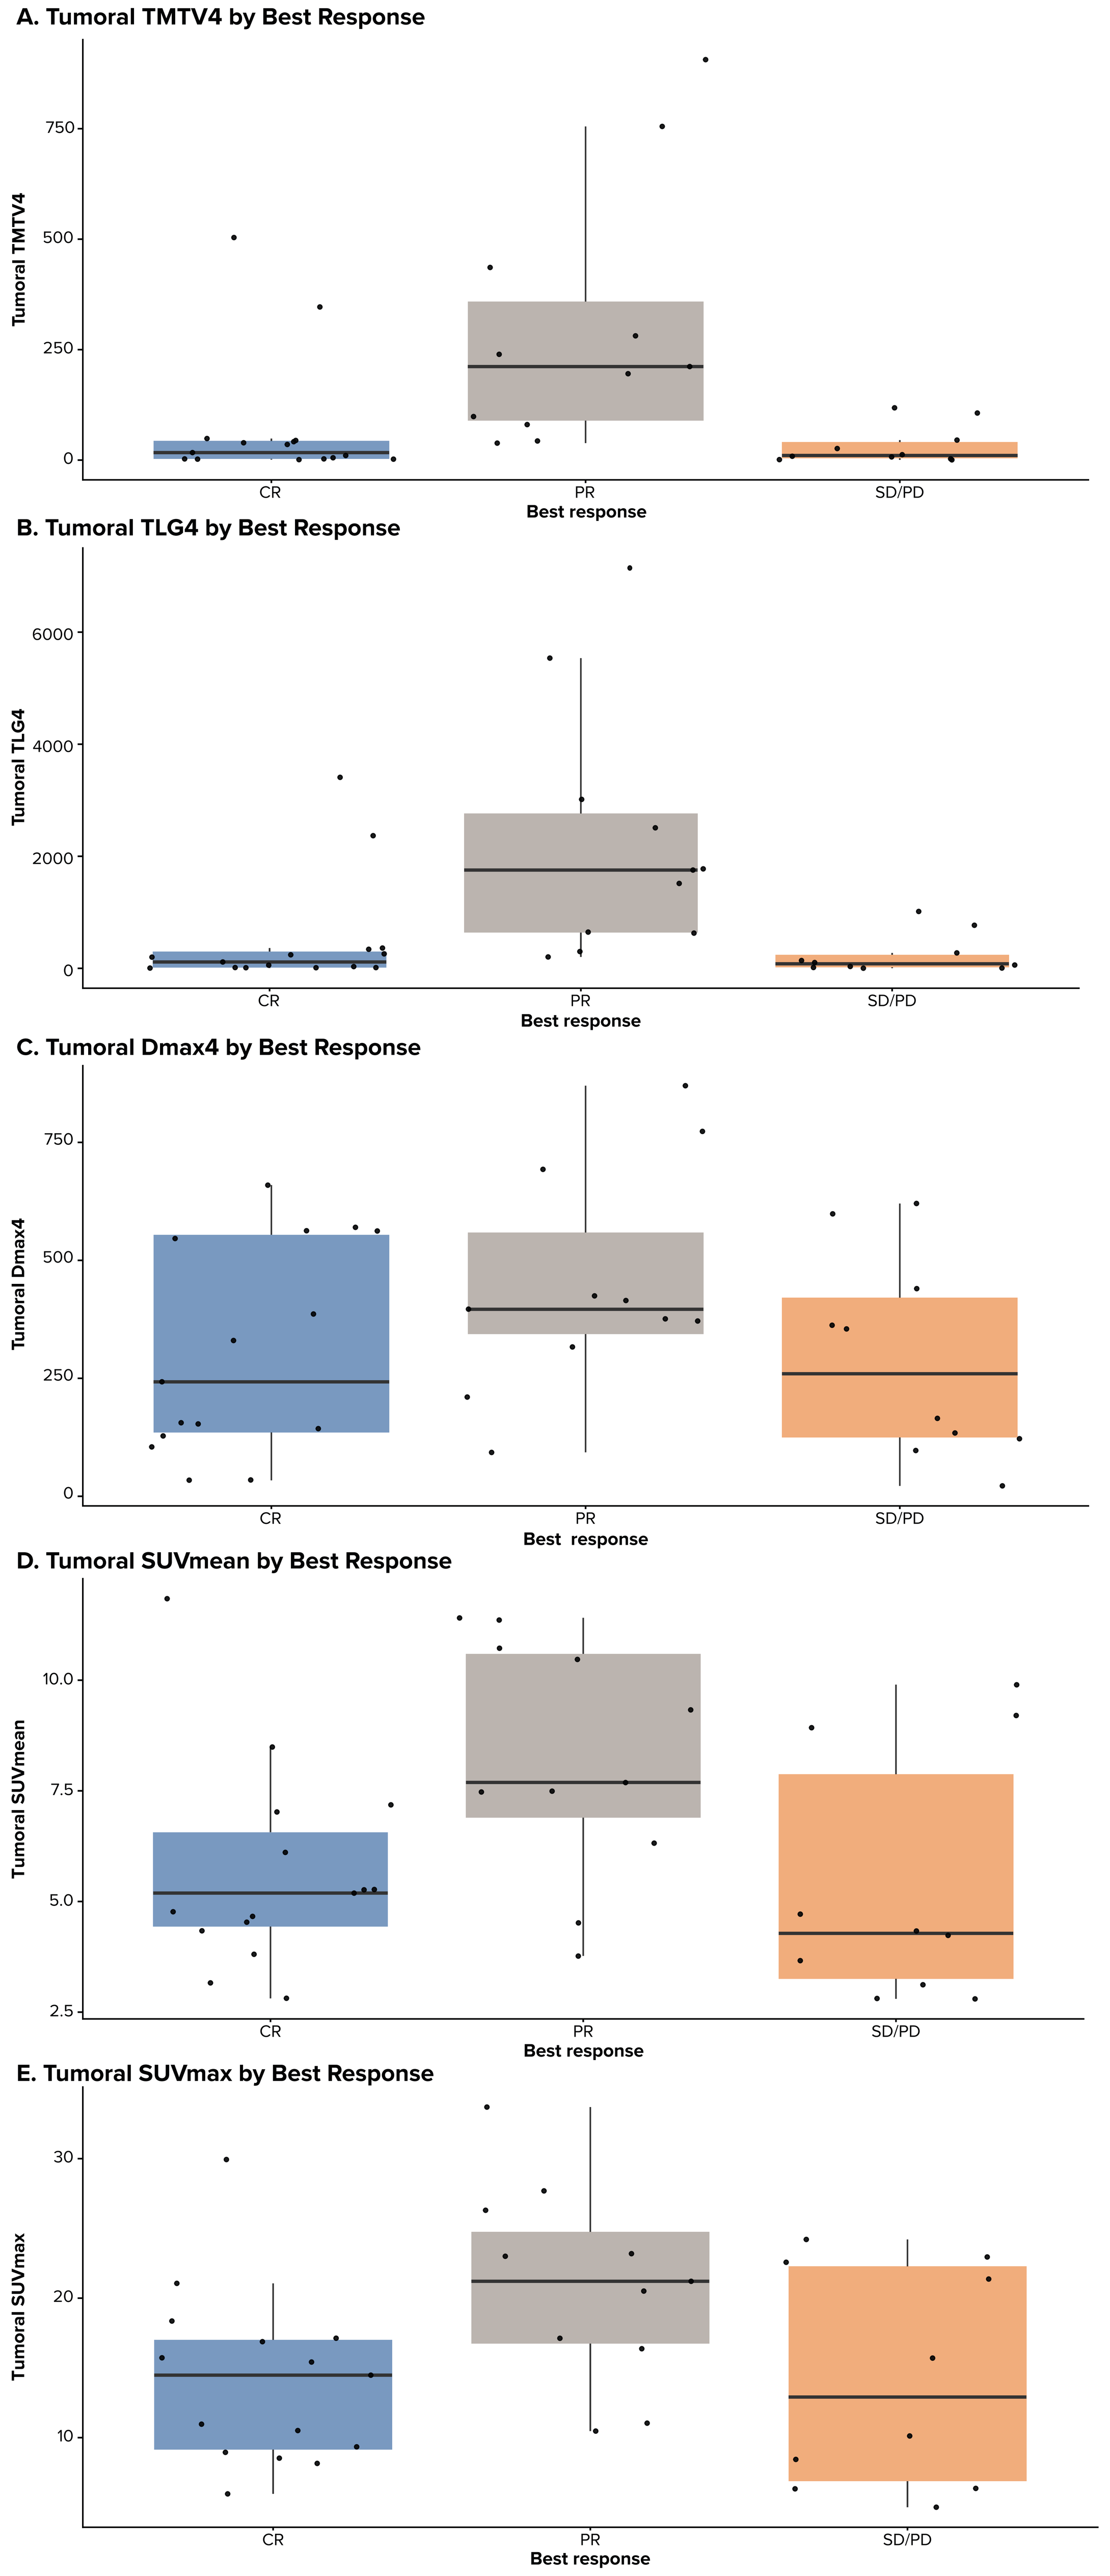
A. Tumoral TMTV4 by Best Response
Tumoral TMTV4
B. Tumoral TLG4 by Best Response
Tumoral TLG4
C. Tumoral Dmax4 by Best Response
Tumoral Dmax4
D. Tumoral SUVmean by Best Response
Tumoral SUVmean
E. Tumoral SUVmax by Best Response
Tumoral SUVmax
CR
PR
SD/PD
Best response

Abbreviations: CR, complete response; Dmax4, maximum distance between lesions with SUV ≥4; PD, progressive disease; PET-CT, positron emission tomography–computed tomography; PR, partial response; SD, stable disease; SUV, standardized uptake value; TLG4, total lesion glycolysis with SUV ≥4; TMTV4; total metabolic tumor volume with SUV ≥4.

PET-CT Metrics and Outcome

- The depth of metabolic response at first evaluation (week 12) influenced PFS and OS and was associated with 1-year PFS rates of 60%, 26%, and 8% (**Figure 3A**) and 1-year OS rates of 100%, 100%, and 77% (**Figure 3B**) in patients with best response of CMR (n=10), PMR (n=13), and NMR (n=13), respectively

Figure 3. PFS and OS by First Metabolic Response

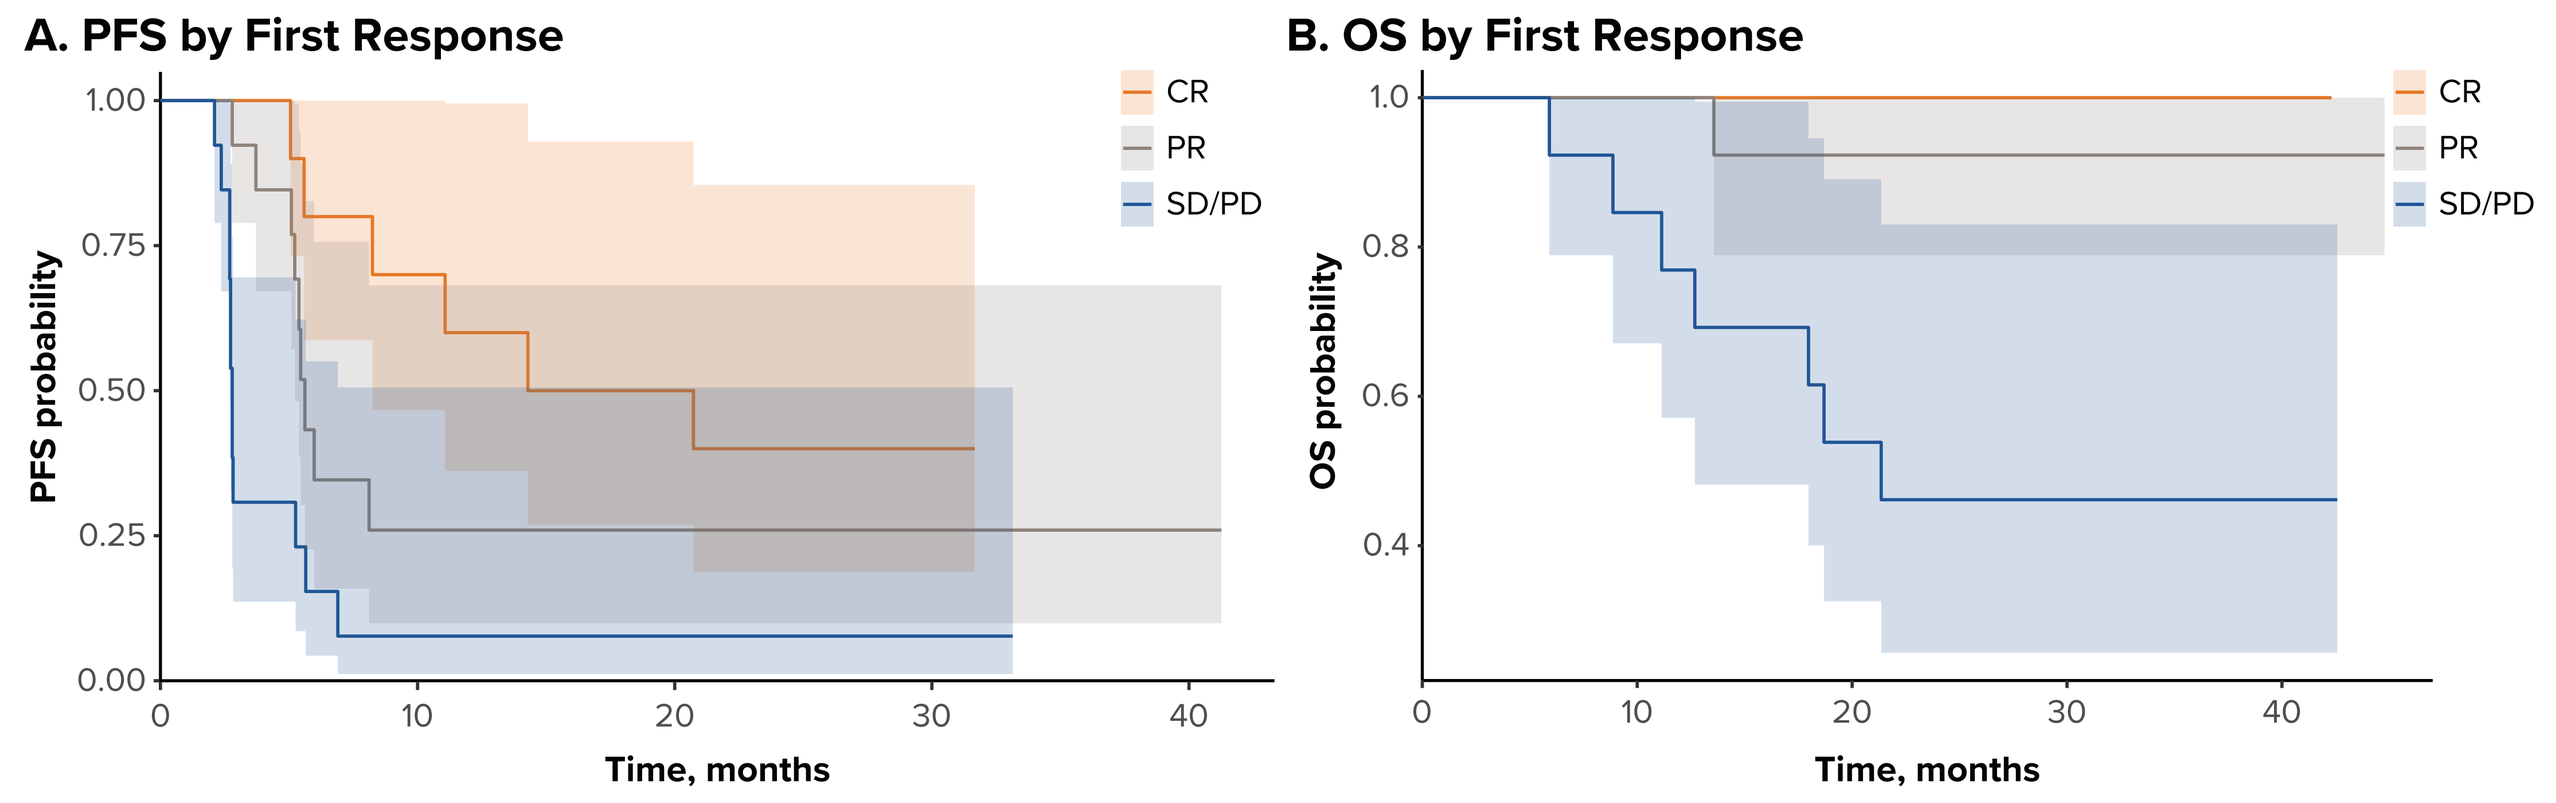
A. PFS by First Response
PFS probability
B. OS by First Response
OS probability
CR
PR
SD/PD
Time, months

Abbreviations: CR, complete response; OS, overall survival; PD, progressive disease; PET-CT, positron emission tomography–computed tomography; PFS, progression-free survival; PR, partial response; SD, stable disease.

Longitudinal Evaluations of PET-CT Metrics

- Longitudinal evaluations of TMTV4 (**Figure 4**) and SUVmean (**Figure 5**) showed a rapid decrease in values at first evaluation in patients with CMR and PMR at week 12
- Patterns of TMTV4 and SUV variations during tislelizumab treatment differed between patients with best metabolic response of CMR, PMR, SD, and PD

Figure 4. TMTV4 (Change From Baseline)

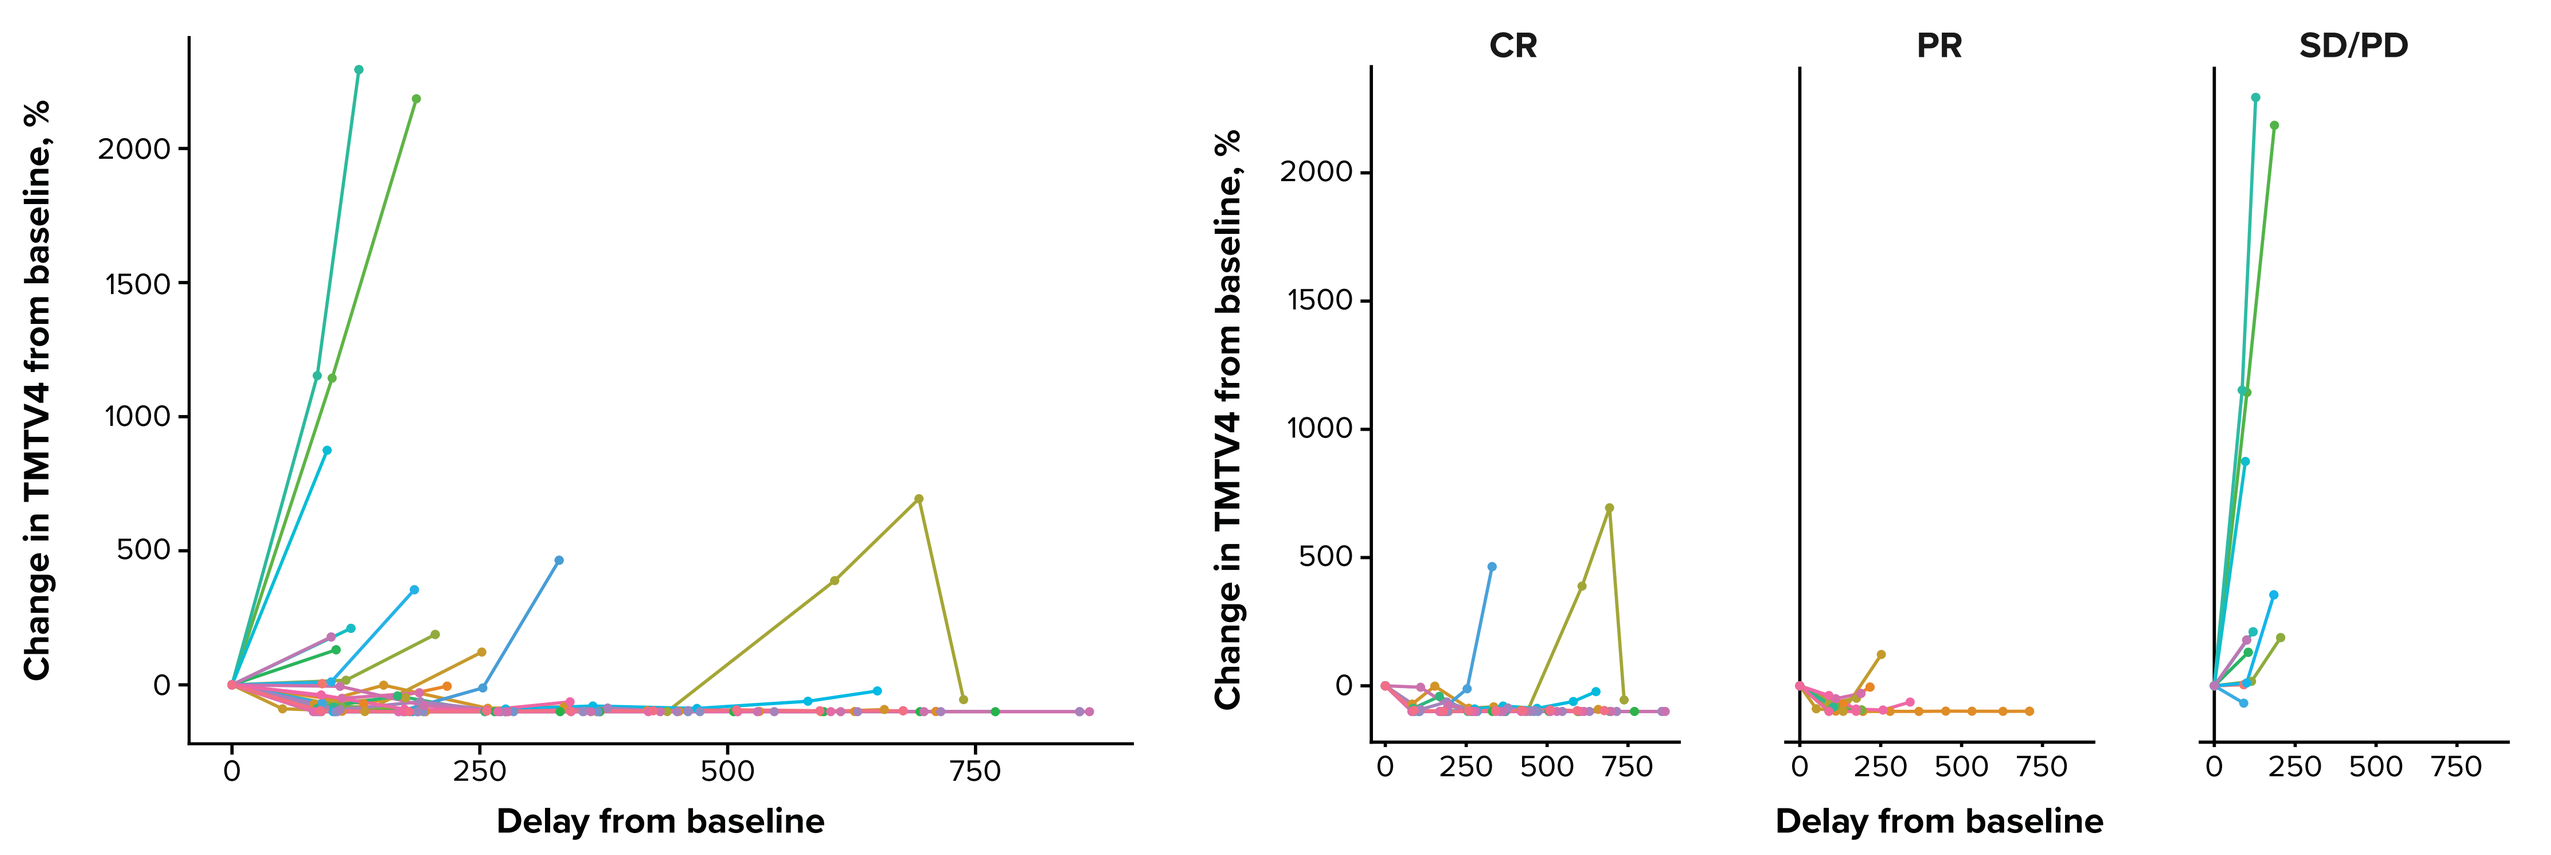
Change in TMTV4 from baseline, %
Delay from baseline
CR
PR
SD/PD

Abbreviations: CR, complete response; PD, progressive disease; PR, partial response; SD, stable disease; TMTV4; total metabolic tumor volume with standardized uptake value ≥4.

Figure 5. SUVmean (Change From Baseline)

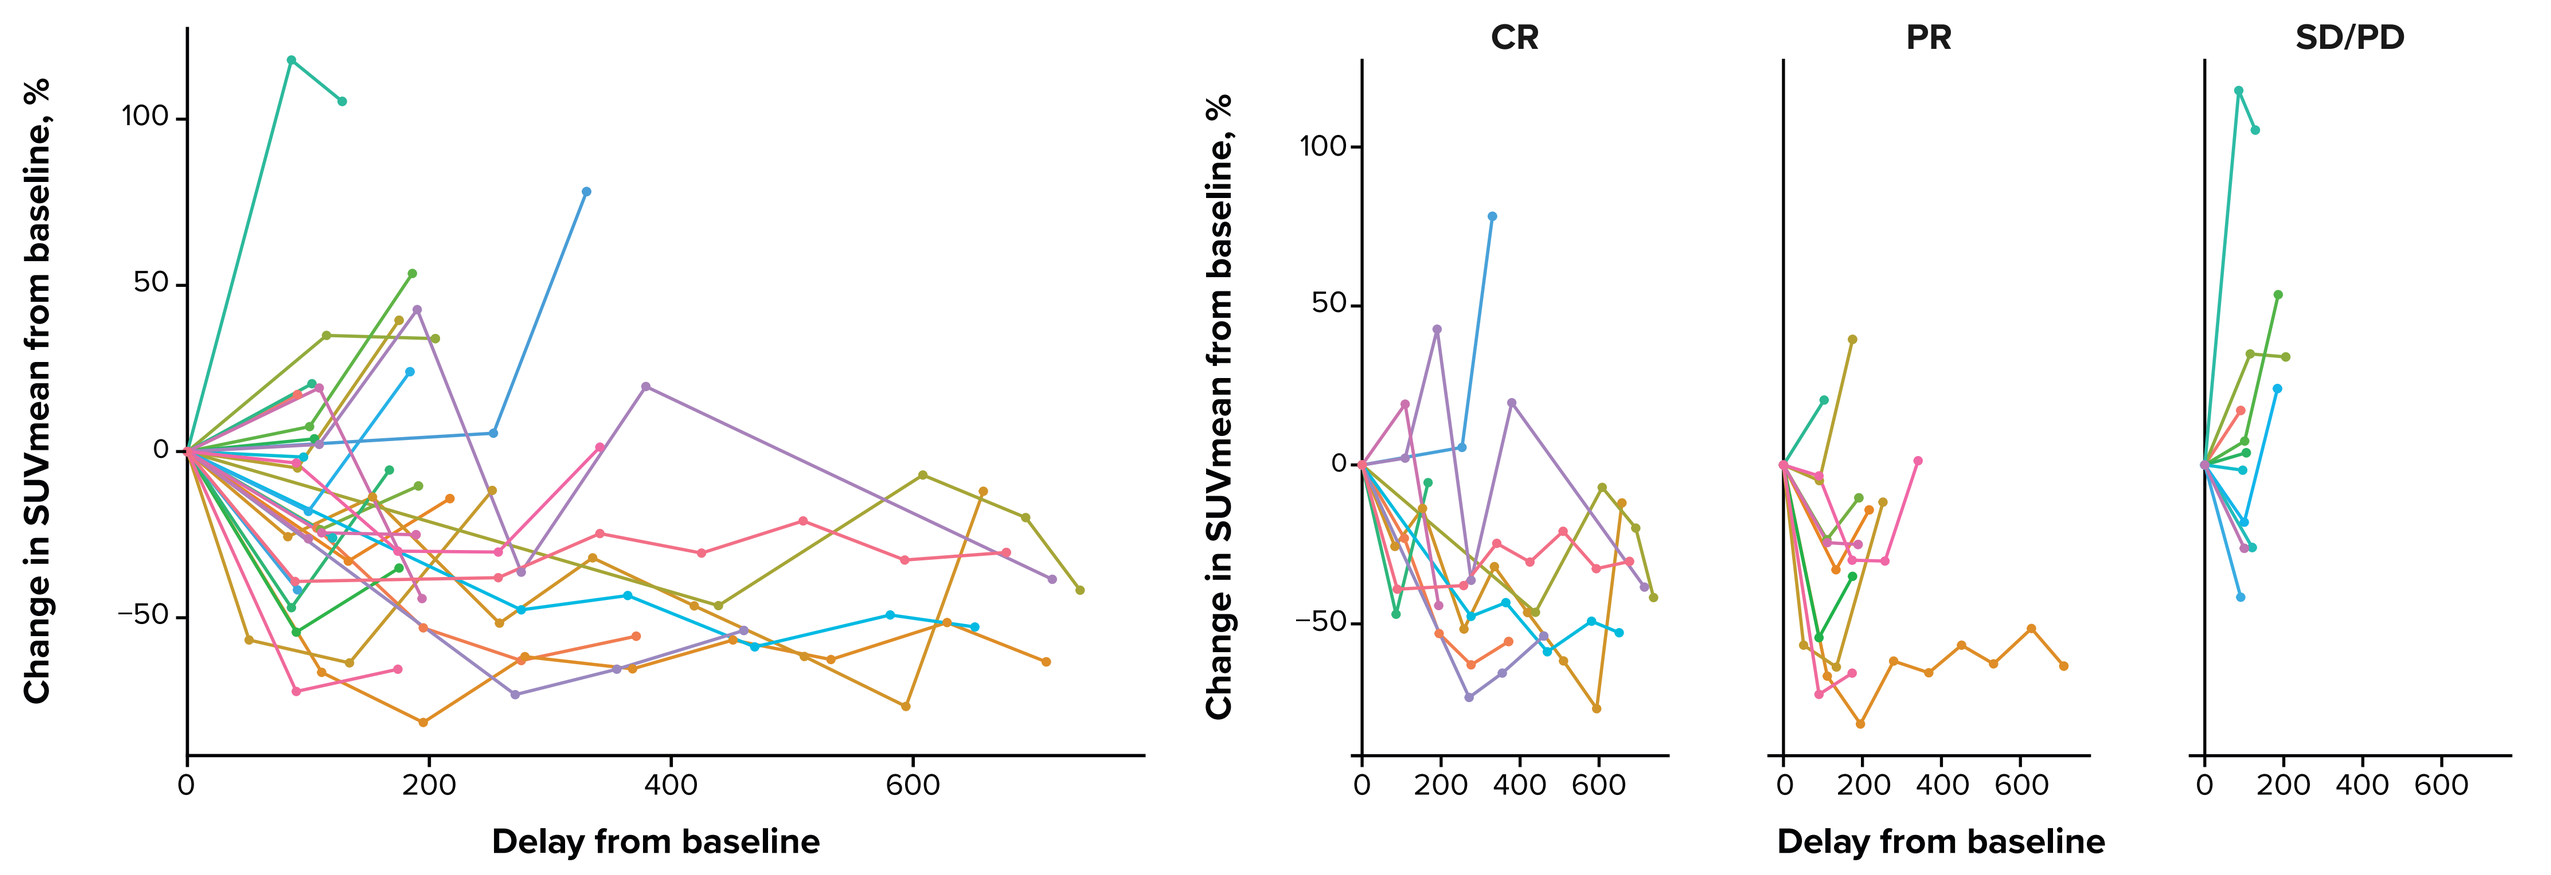
Change in SUVmean from baseline, %
Delay from baseline
CR
PR
SD/PD

Abbreviations: CR, complete response; PD, progressive disease; PR, partial response; SD, stable disease; SUVmean, mean standardized uptake value.

REFERENCES

1. Ghesquières H, et al. ICML 2025. Abstract 130.
2. Budha N, et al. CPT *Pharmacometrics Syst Pharmacol.* 2022;12(1):95-109.
3. Ghesquières H, et al. ASH 2023. Abstract 1717.

DISCLOSURES

HG: Consultant: Roche, BMS, Takeda; Honoraria: Gilead, Roche, BMS, AbbVie, Takeda. **KB:** Honoraria: Takeda, BeOne Medicines, Ltd, Kite Pharma/Gilead, AstraZeneca; Consulting or advisory role: BeOne Medicines, Ltd, Takeda, Lilly, AstraZeneca; Research funding: Takeda; Travel, accommodations, expenses: Takeda, Lilly, Pierre Fabre, AstraZeneca. **SK:** Other relationship: Founder of Pixilib (Imaging CRO). PQ: Honoraria and speakers bureau: Novartis. **ASB:** Travel, accommodations, expenses: Takeda, Johnson & Johnson. **CT:** Consulting or advisory role, travel, accommodations, expenses: Novartis, Roche, BMS, Takeda, AbbVie. **MT:** Honoraria: Janssen, Amgen, Roche, AbbVie, BMS, BeOne Medicines, Ltd; Consulting or advisory role: Janssen, Takeda, Amgen; Travel, accommodations, expenses: Amgen, Janssen, BeOne Medicines, Ltd. **LR:** Consulting or advisory role: Janssen, Takeda, BMS, AbbVie. **TG:** Employment, stock or other ownership: Sanofi; Honorari a: Gilead, Kyte, Takeda, BMS; Consulting or advisory role: Gilead, BMS, Takeda; Expert testimony: Takeda, Gilead; Travel, accommodations, expenses: Roche, Gilead, Takeda. **RR:** Speakers bureau: GSK. **HS:** Honoraria: Curio Science, Epizyme, BMS, Seattle Genetics, ADC Therapeutics. **DM:** Honoraria: Regeneron; Consulting or advisory role: AstraZeneca, Daiichi Sankyo, BMS, ADC Therapeutics, Genmab; Research funding (payable to institution): Karyopharm Therapeutics, Genentech, Genmab, AstraZeneca. **HA:** Employment and may own stock: BeOne Medicines, Ltd, Nkarta Therapeutics; Patents and royalties: St. Jude Children's Research Hospital; Writing support: BeOne Medicines, Ltd; Travel support: BeOne Medicines, Ltd, Spouse: Nkarta, Inc.; Advisory board: BeOne Medicines, Ltd; Leadership role: Board Secretary: Fix Our Ferals, dba Animal Fix Clinic (non-profit surgical clinic for cats and dogs, unrelated to employment or publications), Spouse: Nkarta, Inc. **PF:** Employment: BeOne Medicines, Ltd; Stock or other ownership: BeOne Medicines, Ltd, BMS. **XL, JZ, RD:** Employment and may own stock: BeOne Medicines, Ltd. FM: Consultant: Roche, BMS, Gilead; Honoraria or speakers bureau: Takeda, Roche, Chugai; Data safety monitoring board or advisory board: Gilead, BMS, AbbVie. **MA, CB, MG, FLB, BD, PF, NM, IC, CR:** No disclosures.

ACKNOWLEDGMENTS

This study was co-sponsored by BeOne Medicines, Ltd and LYSARC. Editorial support was provided by Nucleus Global, an Inizio company, and supported by BeOne Medicines.

CORRESPONDENCE: Hervé Ghesquières, herve.ghesquieres@chu-lyon.fr

Presented at the EHA2026 Congress; June 11-14, 2026; Stockholm, Sweden