7058

Navigating the Post-Covalent Bruton Tyrosine Kinase Inhibitor Landscape in Mantle Cell Lymphoma: Real-World Insights on Treatment Patterns, Discontinuation, and Healthcare Resource Utilization

Alvaro Alencar,[1] Swetha Challagulla,[2] Dong Yuan,[2] Qianhong Fu,[2] Keri Yang,[2] Toby A. Eyre[3]

[1]University of Miami Sylvester Comprehensive Cancer Center, Miami, FL, USA; [2]BeOne Medicines, Ltd, San Carlos, CA, USA; [3]Oxford Cancer and Haematology Centre, Churchill Hospital, Headington, Oxford, UK

CONCLUSIONS

- In this real-world analysis, patients with MCL who previously received cBTK inhibitors were most frequently re-treated with other cBTK inhibitors, highlighting the need for novel treatment options in the post-cBTK inhibitor setting
- rwTTD was longest in patients who received cBTK inhibitors or ncBTK inhibitors and shortest in those treated with BCL2 inhibitors
- HCRU burden was lowest for patients who received cBTK inhibitors and highest for those treated with CAR-T therapy
- Further research is warranted to confirm these findings, including investigation into the reasons for re-exposure to cBTK inhibitors

INTRODUCTION

- Mantle cell lymphoma (MCL) is an aggressive B-cell malignancy[1-3]
 – Covalent Bruton tyrosine kinase (cBTK) inhibitors are a standard treatment for relapsed/refractory MCL and are increasingly used in earlier lines of treatment
- Real world evidence on treatment patterns and clinical and economic outcomes following cBTK inhibitor therapy remain limited[3]
- This study aimed to evaluate treatment patterns, real-world time to treatment discontinuation (rwTTD), and healthcare resource utilization (HCRU) among patients with MCL previously treated with cBTK inhibitors

METHODS

Data Source

- A retrospective observational study was conducted using claims data from Symphony Integrated Dataverse® (Symphony Health, Blue Bell, PA) a comprehensive, longitudinal, open claims database including medical, hospital, and pharmacy claims for patients in the United States[4]

Study Design

- Adult patients (aged ≥18 years) with an MCL diagnosis who received a cBTK inhibitor in any line of therapy (LOT) between January 1, 2020, and August 31, 2025, were identified
- The treatment initiation date was identified from both medical and prescription claims and was considered the index date, with the cBTK inhibitor regimen at treatment initiation considered the index regimen
- Patients were excluded if they did not have 30 days of continuous enrollment prior to and following the index date, or if they participated in a clinical trial during the study period

Study Cohorts and Treatment Groups

- Patients were categorized into mutually exclusive treatment groups based on the index regimen of acalabrutinib, ibrutinib, or zanubrutinib
- Post-cBTK inhibitor regimens were categorized as B-cell lymphoma 2 (BCL2) inhibitors, cBTK inhibitors, noncovalent BTK (ncBTK) inhibitors, chimeric antigen receptor T-cell therapy (CAR-T), chemotherapy ± immunotherapy (C±IT), and other
- rwTTD and HCRU, including per-patient per-month (PPPM) outpatient, inpatient, and other medical services, were assessed for regimens received post cBTK inhibitor

RESULTS

Baseline Characteristics of Patients Who Initiated Post-cBTK Inhibitor LOT

- A total of 10,519 patients with MCL treated with a cBTK inhibitor in any line of therapy were identified
- Median age at initiation of post-cBTK inhibitor therapy was 72.0 years, with 77% of patients aged ≥65 years (**Table 1**)
- Patients were predominantly male (74%) and White, non-Hispanic (67%); together, Black, non-Hispanic and Hispanic patients accounted for ≈10% of the study population
- The most common payer was Medicare (62%), consistent with an older patient population

Table 1. Baseline Characteristics

Characteristic	All (N=571)	cBTK inhibitor (n=215)	ncBTK inhibitor[a] (n=182)	C±IT (n=62)	BCL2 inhibitor[b] (n=53)	CAR-T (n=12)	Other[c] (n=47)
Sex, n (%)							
Female	151 (26.4)	59 (27.4)	49 (26.9)	17 (27.4)	12 (22.6)	1 (8.3)	13 (27.7)
Male	420 (73.6)	156 (72.6)	133 (73.1)	45 (72.6)	41 (77.4)	11 (91.7)	34 (72.3)
Age at index							
Mean (SD), years	70.23 (8.44)	69.46 (8.78)	70.75 (8.40)	71.71 (7.97)	70.47 (7.63)	70.92 (4.48)	69.30 (9.12)
Median (IQR), years	72 (65-77)	71 (64-76)	73 (66-78)	75 (67-78)	74 (65-77)	72 (68-75)	72 (62-77)
≥65 years, n (%)	441 (77.2)	154 (71.6)	147 (80.8)	53 (85.5)	41 (77.4)	11 (91.7)	35 (74.5)
Race and ethnicity, n (%)							
White, non-Hispanic	382 (66.9)	144 (67.0)	122 (67.0)	43 (69.4)	36 (67.9)	6 (50.0)	31 (66.0)
Black, non-Hispanic	28 (4.9)	14 (6.5)	8 (4.4)	1 (1.6)	3 (5.7)	1 (8.3)	1 (2.1)
Hispanic	37 (6.5)	15 (7.0)	8 (4.4)	5 (8.1)	3 (5.7)	1 (8.3)	5 (10.6)
Other/ unknown	124 (21.7)	42 (19.6)	44 (24.2)	13 (21.0)	11 (20.7)	4 (33.3)	10 (21.3)
Payer type, n (%)							
Medicare	353 (61.8)	131 (60.9)	121 (66.5)	37 (59.7)	36 (67.9)	5 (41.7)	23 (48.9)
Commercial	183 (32.0)	71 (33.0)	48 (26.4)	23 (37.1)	14 (26.4)	7 (58.3)	20 (42.6)
Medicaid	19 (3.3)	7 (3.3)	8 (4.4)	2 (3.2)	0 (0.0)	0 (0.0)	2 (4.3)
Other/ unknown	16 (2.8)	6 (2.8)	5 (2.7)	0 (0.0)	3 (5.7)	0 (0.0)	2 (4.3)
Charleson Comorbidity Index							
Mean (SD)	2.17 (2.24)	2.21 (2.39)	1.76 (2.05)	1.97 (1.47)	2.25 (1.75)	4.00 (3.57)	3.23 (2.66)

[a]Pirtobrutinib was the only ncBTK inhibitor treatment used by study patients. [b]Venetoclax was the only BCL2 inhibitor used by study patients. [c]Any other drug class not explicitly listed, such as the PI bortezomib.
Abbreviations: BCL2, B-cell lymphoma 2; CAR-T, chimeric antigen receptor T-cell; cBTK, covalent Bruton tyrosine kinase; C±IT: chemotherapy ± immunotherapy; ncBTK, noncovalent Bruton tyrosine kinase; PI, proteasome inhibitor.

Treatment Sequencing

- Among all identified patients, 3031 (29.0%) had been previously treated with a cBTK inhibitor at index (zanubrutinib, n=1007; acalabrutinib, n=1302; ibrutinib, n=722)
 – cBTK inhibitors were most frequently received in the second line (n=1720; 56.7%), followed by first line (n=950; 31.3%), third line (n=288; 9.5%), fourth line (n=54; 1.8%), fifth line (n=15; 0.5%), and sixth line (n=4; 0.1%)

- Among patients with MCL who received cBTK inhibitors, 571 (19.0%) received subsequent LOTs (**Figure 1**)
 – cBTK inhibitor-based regimens were the most commonly used regimen immediately following initial cBTK inhibitor therapy (37.7%), followed by ncBTK inhibitor-based regimens (31.9%), C±IT (10.9%), BCL2 inhibitor (9.3%; venetoclax monotherapy, 7%), other (7.8%), and CAR-T (2.1%)

Figure 1. Post-cBTK Inhibitor Treatment Regimens by cBTK Inhibitor at Index

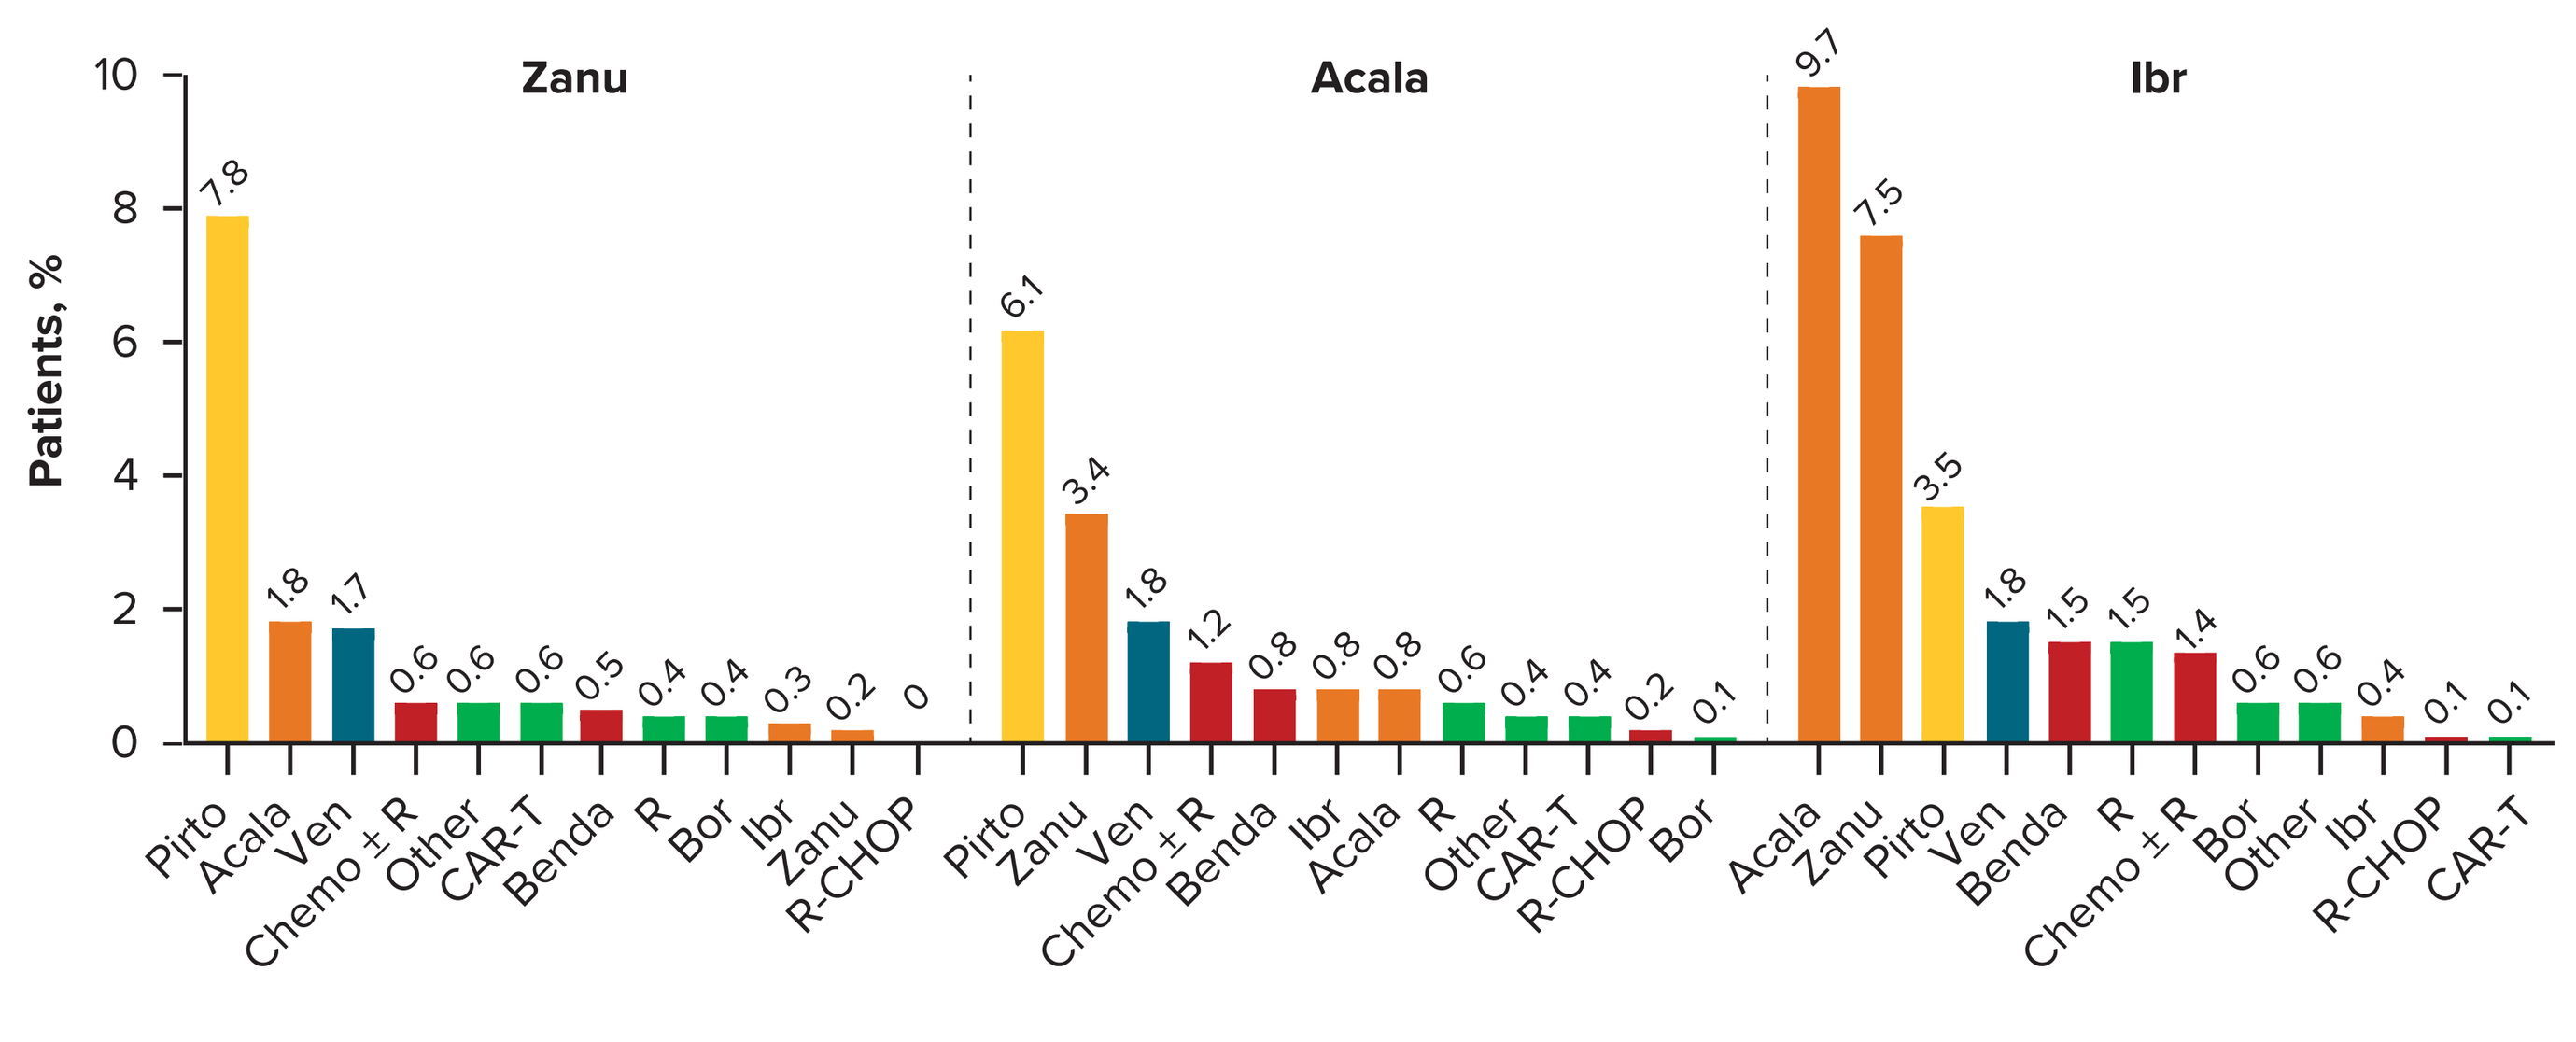
BCL2 inhibitor
cBTK inhibitor
C±IT
ncBTK inhibitor
Other
Patients, %
Zanu
Acala
Ibr
Pirto 7.8, Acala 1.8, Ven 1.7, Chemo ± R 0.6, Other 0.6, CAR-T 0.6, Benda 0.5, R 0.4, Bor 0.4, Ibr 0.3, Zanu 0.2, R-CHOP 0
Pirto 6.1, Zanu 3.4, Ven 1.8, Chemo ± R 1.2, Benda 0.8, Ibr 0.8, Acala 0.8, R 0.6, Other 0.4, CAR-T 0.4, R-CHOP 0.2, Bor 0.1
Acala 9.7, Zanu 7.5, Pirto 3.5, Ven 1.8, Benda 1.5, R 1.5, Chemo ± R 1.4, Bor 0.6, Other 0.6, Ibr 0.4, R-CHOP 0.1, CAR-T 0.1

Abbreviations: acala, acalabrutinib; benda, bendamustine; bor, bortezomib; CAR-T, chimeric antigen receptor T-cell; cBTK, covalent Bruton tyrosine kinase; chemo, chemotherapy; ibr, ibrutinib; pirto, pirtobrutinib; R, rituximab; R-CHOP, rituximab, cyclophosphamide, doxorubicin, vincristine, and prednisone; ven, venetoclax; zanu, zanubrutinib.

rwTTD

- Among patients receiving treatment post cBTK inhibitor, median rwTTD was 222 days (95% CI, 181-281); rwTTD differed significantly across treatment regimens (log-rank P<.001)
- The longest median rwTTD was observed for cBTK inhibitors (365 days), followed by ncBTK inhibitors (206 days)
 – Shorter median rwTTDs were observed with C±IT (169 days) and BCL2 inhibitors (140 days)
- At 12 months, approximately 49% of patients receiving cBTK inhibitors discontinued treatment compared with 62% receiving ncBTK inhibitors and 84% receiving BCL2 inhibitors (**Figure 2**)

Figure 2. rwTTD For Post cBTK Inhibitor Treatments

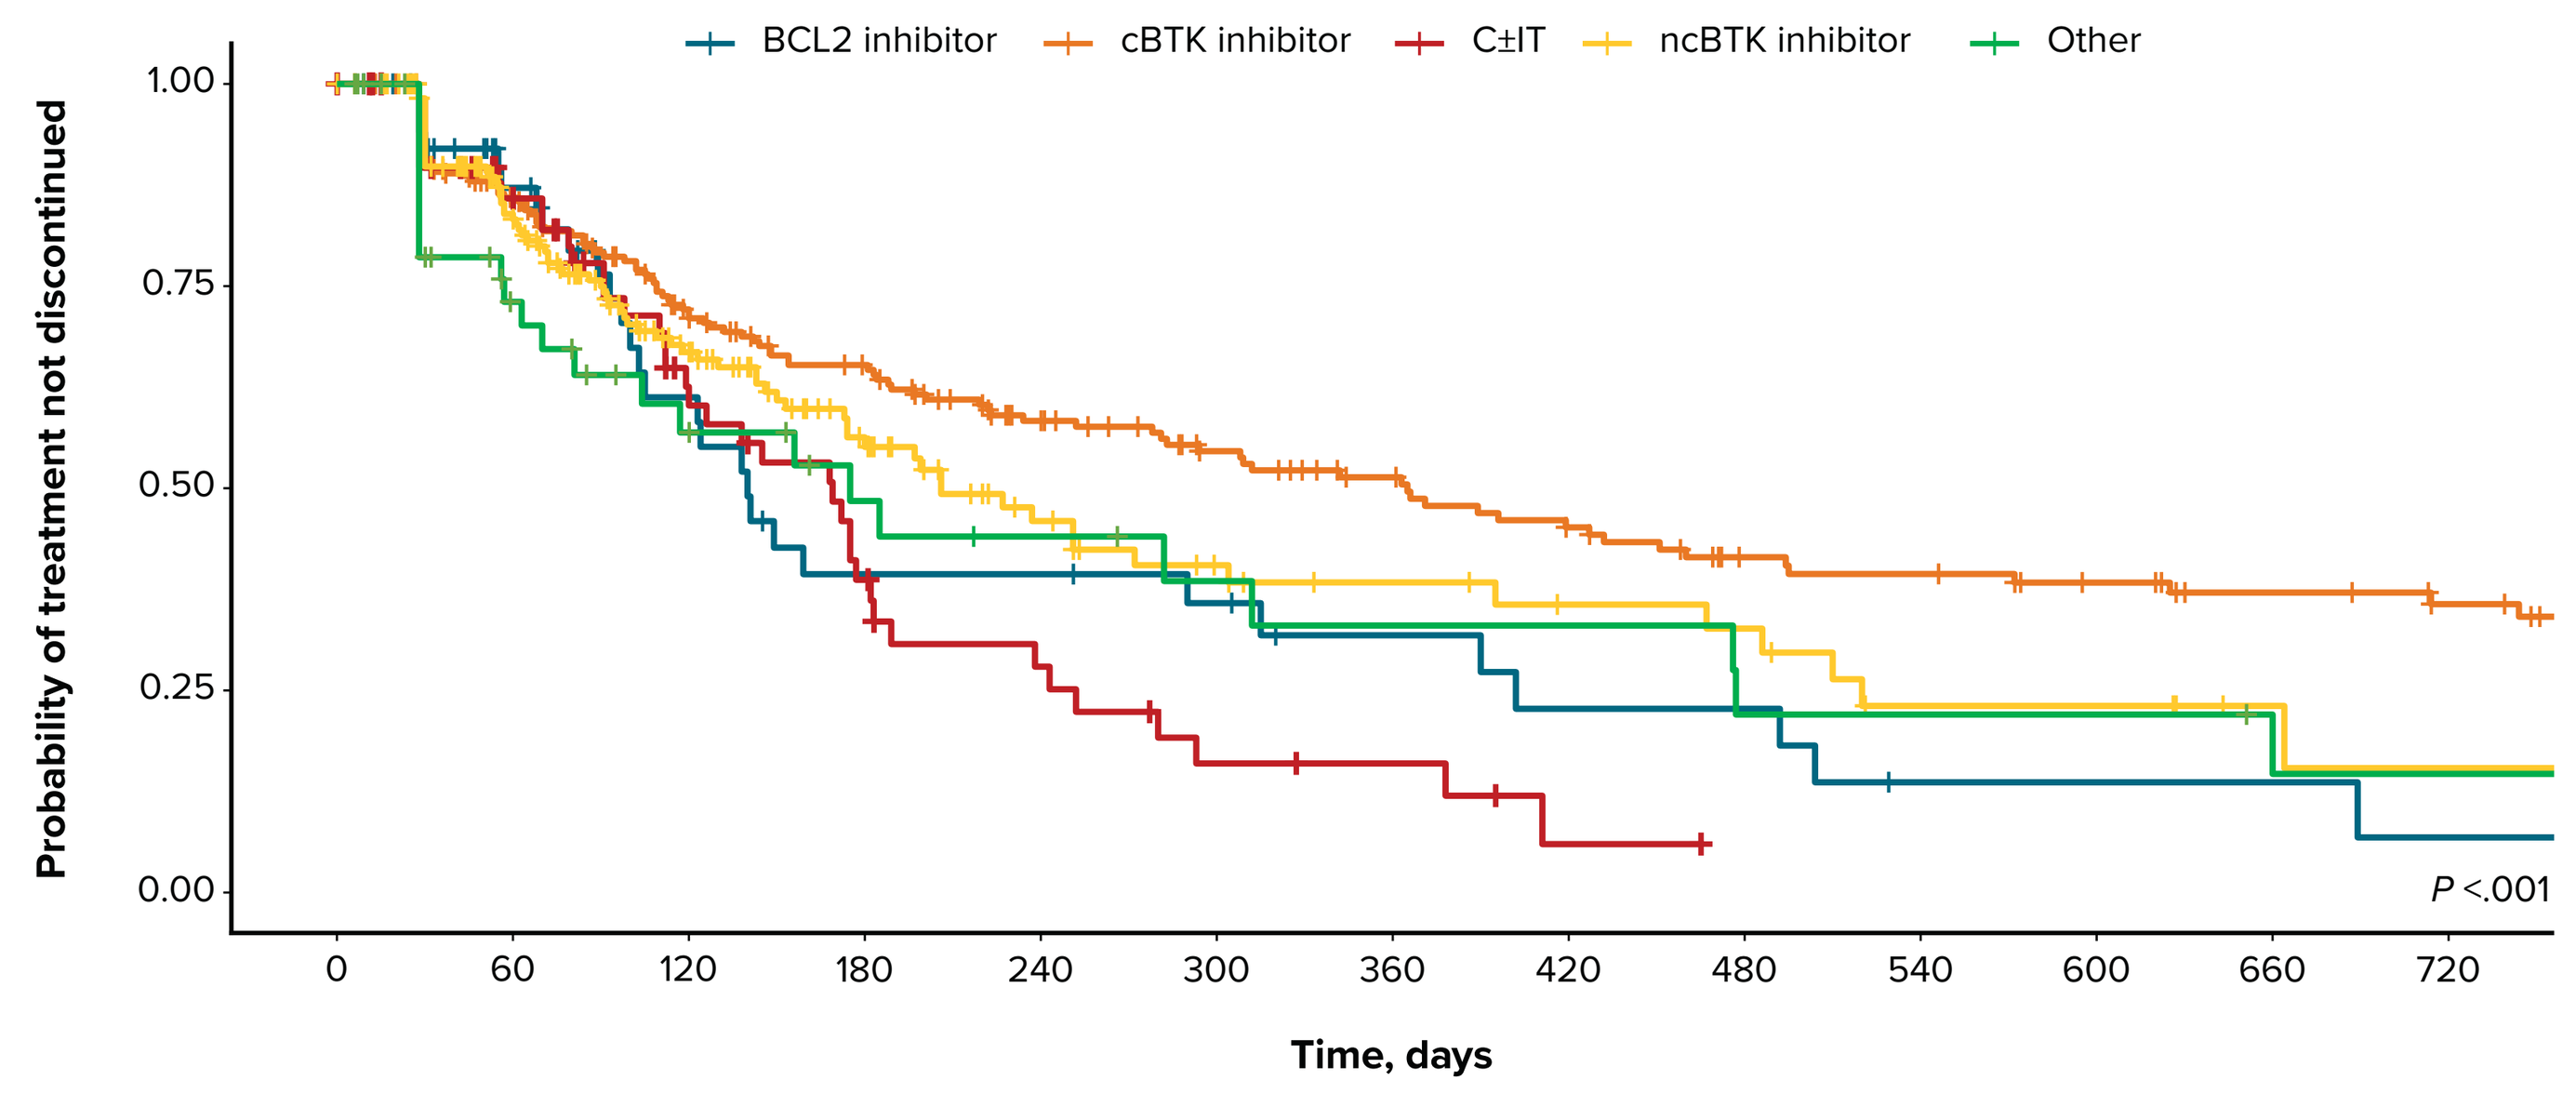
BCL2 inhibitor
cBTK inhibitor
C±IT
ncBTK inhibitor
Other
Probability of treatment not discontinued
Time, days
P <.001

Number at risk	0	60	120	180	240	300	360	420	480	540	600	660	720
BCL2 inhibitor	53	36	20	12	12	10	7	5	5	2	2	2	1
cBTK inhibitor	215	169	130	108	85	69	59	50	40	38	33	28	24
C±IT	62	45	27	16	10	5	4	1	0	0	0	0	0
ncBTK inhibitor	182	128	75	47	27	19	15	12	11	6	6	3	2
Other	47	25	16	11	9	7	6	6	4	4	4	3	2

Abbreviations: BCL2, B-cell lymphoma 2; cBTK, covalent Bruton tyrosine kinase; C±IT: chemotherapy ± immunotherapy; ncBTK, noncovalent Bruton tyrosine kinase; rwTTD, real-world time to treatment discontinuation.

HCRU

- Outpatient visit utilization differed substantially by treatment regimen, with cBTK inhibitor-based regimens demonstrating the lowest outpatient visit burden (mean PPPM, 1.2) and CAR-T therapy corresponding with the highest outpatient utilization (mean PPPM, 10.6; **Figure 3**)
- Utilization of inpatient services also varied notably across regimens, with cBTK inhibitor therapy associated with the lowest inpatient use (mean PPPM, 0.2) and CAR-T therapy with the highest inpatient use (mean PPPM, 1.9)
- ncBTK inhibitor-based regimens were associated with moderate outpatient and inpatient utilization (mean PPPM, 1.5 and 0.3, respectively), which was lower than utilization of all other treatment regimens except for cBTK inhibitor therapy
- Utilization of other medical services was lowest among cBTK inhibitor-treated patients (mean PPPM, 1.1) and highest with for those treated with C±IT regimens (mean PPPM, 2.5), with modest variation observed across other targeted-therapy approaches

Figure 3. HCRU by MCL Treatment Regimens Received Post cBTK Inhibitor

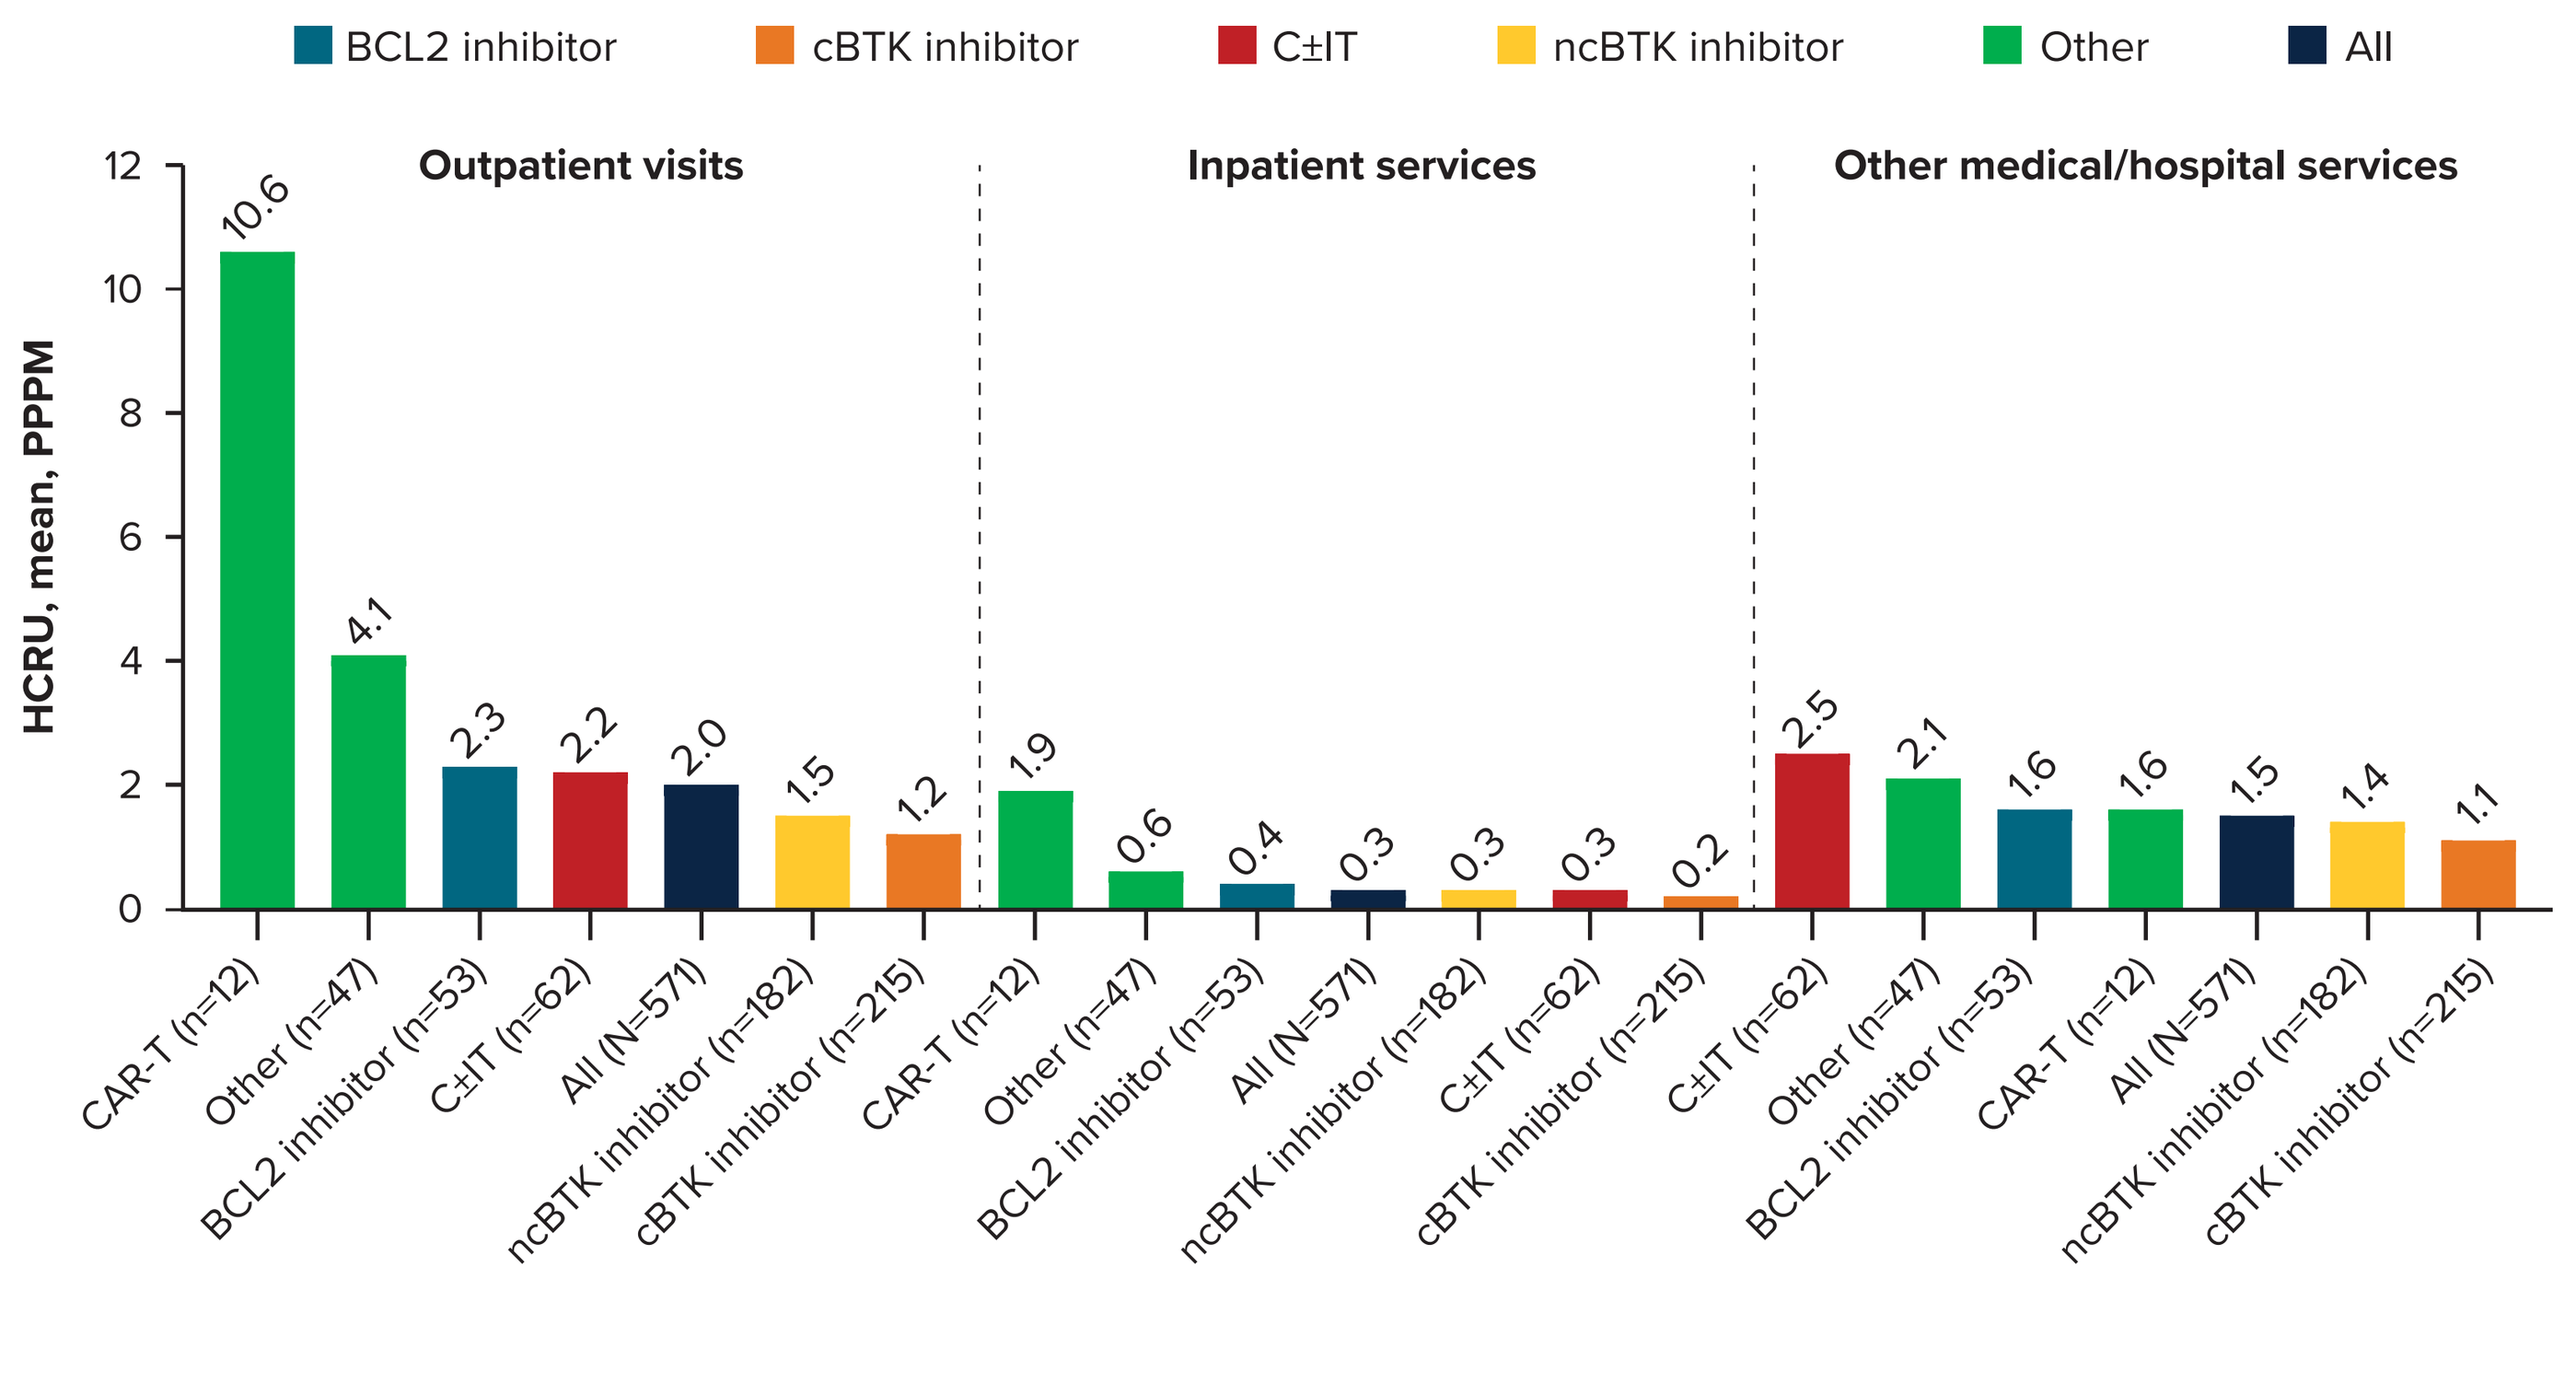
BCL2 inhibitor
cBTK inhibitor
C±IT
ncBTK inhibitor
Other
All
HCRU, mean, PPPM
Outpatient visits
Inpatient services
Other medical/hospital services
CAR-T (n=12) 10.6, Other (n=47) 4.1, BCL2 inhibitor (n=53) 2.3, C±IT (n=62) 2.2, All (N=571) 2.0, ncBTK inhibitor (n=182) 1.5, cBTK inhibitor (n=215) 1.2
CAR-T (n=12) 1.9, Other (n=47) 0.6, BCL2 inhibitor (n=53) 0.4, All (N=571) 0.3, ncBTK inhibitor (n=182) 0.3, C±IT (n=62) 0.3, cBTK inhibitor (n=215) 0.2
C±IT (n=62) 2.5, Other (n=47) 2.1, BCL2 inhibitor (n=53) 1.6, CAR-T (n=12) 1.6, All (N=571) 1.5, ncBTK inhibitor (n=182) 1.4, cBTK inhibitor (n=215) 1.1

Abbreviations: BCL2, B-cell lymphoma 2; CAR-T, chimeric antigen receptor T-cell; cBTK, covalent Bruton tyrosine kinase; C±IT, chemotherapy ± immunotherapy; HCRU, healthcare resource utilization; MCL, mantle cell lymphoma; ncBTK, noncovalent Bruton tyrosine kinase; PPPM, per-patient per-month.

DISCUSSION

- In the post-cBTK inhibitor setting, retreatment with cBTK inhibitors was the most common subsequent treatment strategy, highlighting both limited available options and ongoing unmet need for effective therapies in this patient population
- These findings highlight that there are important trade-offs between treatment durability and HCRU in the post-cBTK inhibitor landscape and support the need for further research into optimal sequencing strategies and drivers of cBTK inhibitor re-exposure in patients with MCL

REFERENCES

1. Silkenstedt E, Dreyling M. *Hematol Oncol.* 2023;41(suppl 1):36-42.
2. Grainger BT, Cheah CY. *Haematologica.* 2025;110(3):576-587.
3. Sancho JM, et al. *J Blood Med.* 2024;15:239-254.
4. Integrated Dataverse and Source Solutions. Symphony Health. Accessed April 17, 2026. https://www.iconplc.com/solutions/real-world-intelligence/symphony-health/integrated-dataverse-and-source-solutions

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Editorial assistance was provided by Nucleus Global, an Inizio company, and supported by BeOne Medicines.

DISCLOSURES

AA: Consulting or advisory role: Amgen, Lilly, Genentech, AbbVie, BeOne Medicines, Ltd, ADC Therapeutics, Incyte.

CORRESPONDENCE: Alvaro Alencar; aalencar@med.miami.edu

Presented at the 2026 American Society of Clinical Oncology (ASCO) Annual Meeting; May 29-June 2, 2026; Chicago, IL, USA