Combination of Zanubrutinib + Venetoclax for Treatment-naive CLL/SLL: Results in SEQUOIA Arm D

PS1566

Mazyar Shadman,[1-2] Talha Munir,[3] Shuo Ma,[4] Masa Lasica,[5] Monica Tani,[6] Tadeusz Robak,[7] Ian W. Flinn,[8] Jennifer R. Brown,[9] Paolo Ghia,[10-11] Emmanuelle Ferrant,[12] Constantine S. Tam,[13] Wojciech Janowski,[14] Wojciech Jurczak,[15] Linlin Xu,[16] Tian Tian,[16] Stephanie Agresti,[16] Jamie Hirata,[16] Alessandra Tedeschi[17]

[1]Fred Hutchinson Cancer Center, Seattle, WA, USA; [2]University of Washington, Seattle, WA, USA; [3]Leeds Teaching Hospitals NHS Trust, Leeds, UK; [4]Robert H. Lurie Comprehensive Cancer Center, Northwestern University Feinberg School of Medicine, Chicago, IL, USA; [5]St Vincent's Hospital Melbourne, Melbourne, VIC, Australia; [6]Santa Maria delle Croci Hospital, Ravenna, Italy; [7]Copernicus Memorial Hospital, Medical University of Łódź, Łódź, Poland; [8]Tennessee Oncology/OneOncology, Nashville, TN, USA; [9]Dana-Farber Cancer Institute, Boston, MA, USA; [10]Università Vita-Salute San Raffaele, Milano, Italy; [11]IRCCS Ospedale San Raffaele, Milano, Italy; [12]CHU de Lyon-Sud, Lyon-Sud, France; [13]Alfred Hospital and Monash University, Melbourne, VIC, Australia; [14]Calvary Mater Newcastle Hospital, Waratah, NSW, Australia; [15]Maria Sklodowska-Curie National Research Institute of Oncology, Kraków, Poland; [16]BeOne Medicines Ltd, San Carlos, CA, USA; [17]ASST Grande Ospedale Metropolitano Niguarda, Milano, Italy

CONCLUSIONS

- In SEQUOIA Arm D, zanubrutinib + venetoclax in TN CLL/SLL showed robust efficacy with deep and durable responses, regardless of del(17p)/*TP53* mutational status
 - In patients without del(17p) and *TP53* mutation, the 24-month PFS was 89%; in patients with del(17p) and/or *TP53* mutation, the 24-month PFS was 94% and maintained at 36-months (88%)
- Best uMRD in the peripheral blood was achieved in 59% of patients
 - uMRD was achieved in 43% by Cycle 16 and 60% by Cycle 28 for patients without del(17p) and *TP53* mutation
- The safety profile of zanubrutinib + venetoclax was tolerable and no unexpected safety signals were identified
 - Rates of atrial fibrillation/flutter were low and no cardiac- or COVID-19-related deaths occurred on study
- Zanubrutinib + venetoclax combination compares favorably with currently available fixed-duration regimens for patients with TN CLL/SLL
- These data highlight the potential for an all oral, time-limited therapy, with zanubrutinib as a backbone, to drive meaningful disease control regardless of del(17p)/*TP53* mutation status

INTRODUCTION

- Zanubrutinib is a highly potent and selective next-generation Bruton tyrosine kinase (BTK) inhibitor that was designed to provide complete and sustained target inhibition and is the only BTK inhibitor to demonstrate superiority over ibrutinib in a head-to-head phase 3 trial, including high risk del(17p)[1-4]
- Fixed-duration therapies with BTK and BCL2 inhibitors are emerging as a new treatment option but there are limitations due to efficacy or safety concerns, especially in high-risk populations with del(17p)/*TP53* mutation
- Most previous studies either excluded or only included a small percentage of patients with del(17p)/*TP53* mutation[5-7]
- Furthermore, optimal duration of treatment to achieve deep and durable remission has yet to be determined
- SEQUOIA (NCT03336333) is a phase 3 study that evaluated zanubrutinib in a broad range of patients with treatment-naive chronic lymphocytic leukemia/small lymphocytic lymphoma (CLL/SLL), including those with high-risk features (**Figure 1**)[8,9]
- Here, results from SEQUOIA Arm D are presented for zanubrutinib + venetoclax in patients with del(17p) and/or *TP53* mutation or without both

METHODS

Study design

- Arm D is a nonrandomized cohort of SEQUOIA, in which patients with del(17p) and/or *TP53* mutation or without both received zanubrutinib + venetoclax (**Figure 1**); treatment schedule is shown in **Figure 2**

Assessments

- Study endpoints are shown in **Figure 1**
- Progression-free survival (PFS) and overall survival (OS) were assessed in the intention-to-treat population (ITT)
- Overall response rate (ORR) was assessed by investigator per the 2008 International Workshop on Chronic Lymphocytic Leukemia (iwCLL) guidelines[10] with modification for treatment-related lymphocytosis[11] in patients with CLL and per Lugano criteria[12] in patients with small lymphocytic lymphoma (SLL)
 - ORR was defined as achievement of partial response with lymphocytosis (PR-L) or better

Figure 1. SEQUOIA Study Design

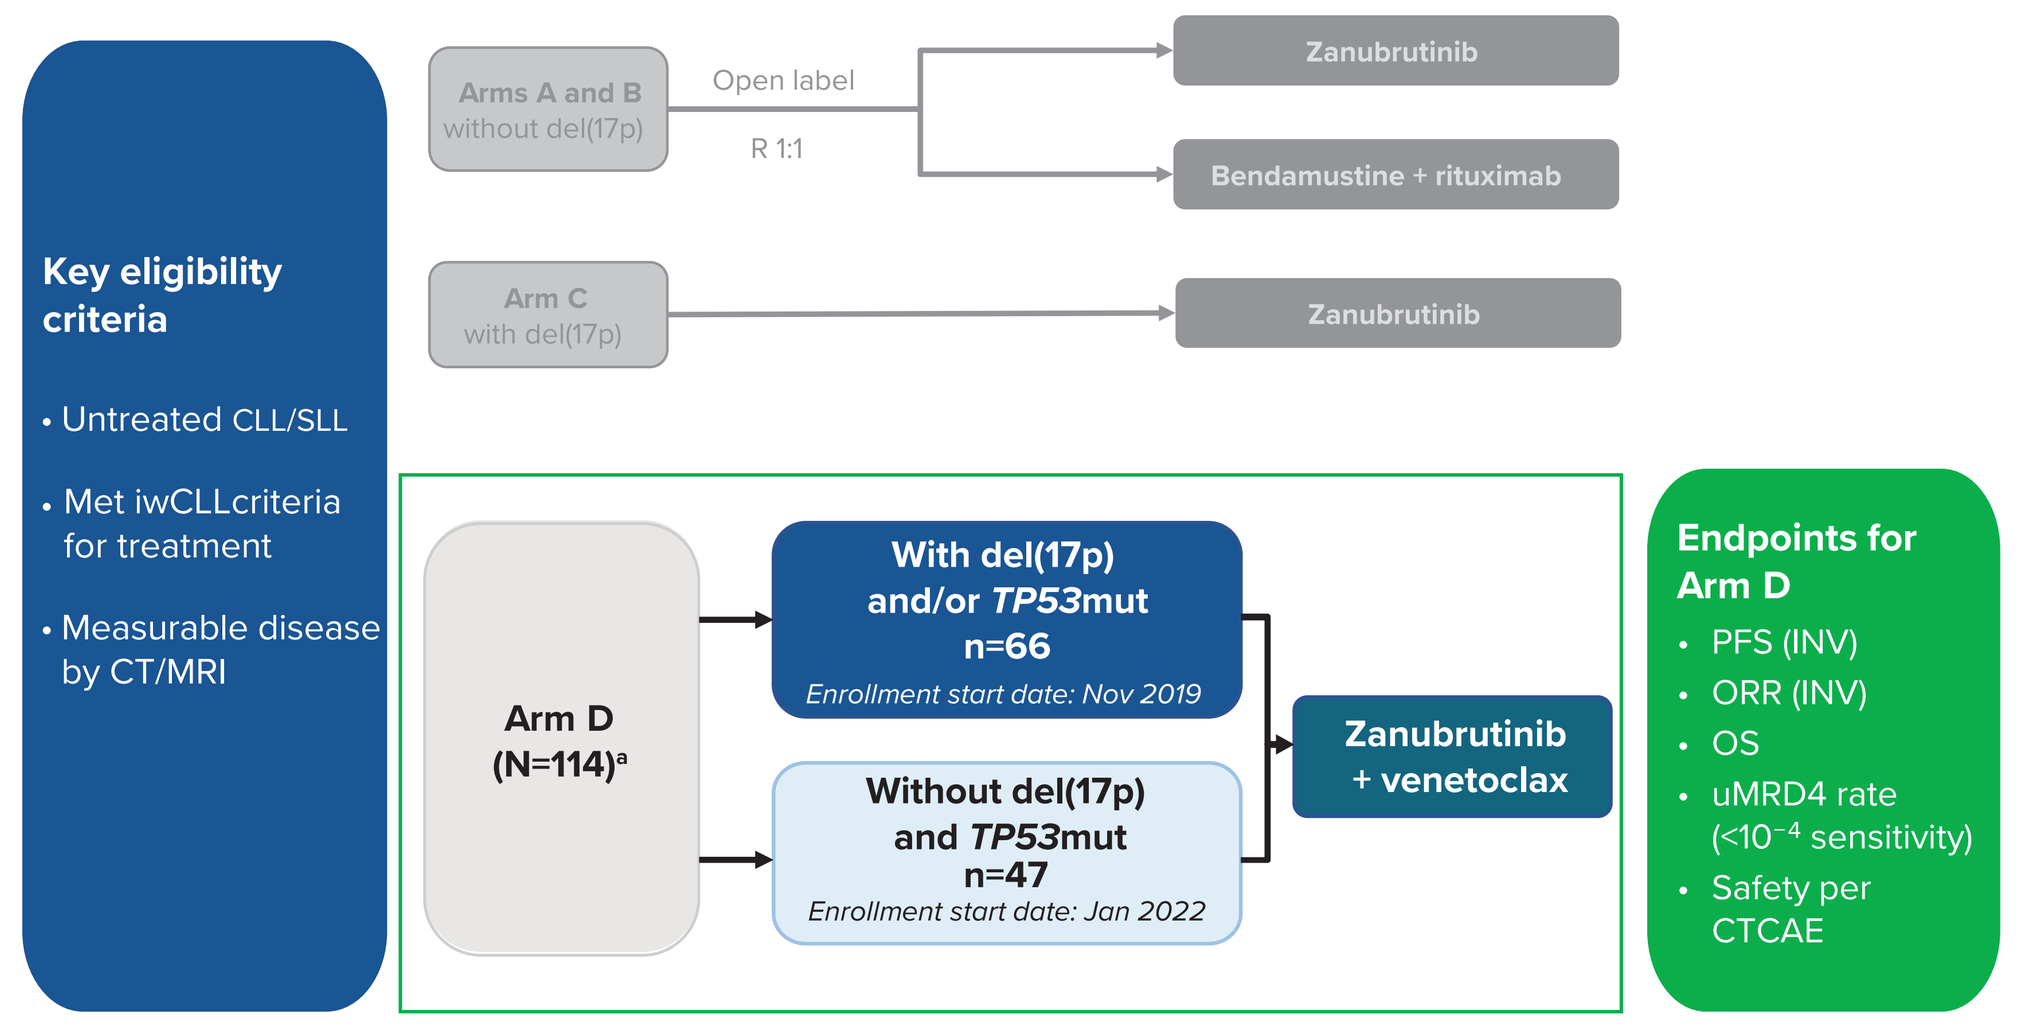
Key eligibility criteria
• Untreated CLL/SLL
• Met iwCLLcriteria for treatment
• Measurable disease by CT/MRI
Arms A and B without del(17p)
Open label
R 1:1
Zanubrutinib
Bendamustine + rituximab
Arm C with del(17p)
Zanubrutinib
Arm D (N=114)a
With del(17p) and/or TP53mut n=66
Enrollment start date: Nov 2019
Without del(17p) and TP53mut n=47
Enrollment start date: Jan 2022
Zanubrutinib + venetoclax
Endpoints for Arm D
• PFS (INV)
• ORR (INV)
• OS
• uMRD4 rate (<10⁻⁴ sensitivity)
• Safety per CTCAE

[a]One patient had a missing *TP53* result (via central laboratory).
Abbreviations: CLL, chronic lymphocytic leukemia; CT, computed tomography; CTCAE, Common Terminology Criteria for Adverse Events; INV, investigator-assessed; iwCLL, International Workshop on Chronic Lymphocytic Leukemia; MRI, magnetic resonance imaging; mut, mutation; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PR-L, partial response with lymphocytosis; R, randomized; SLL, small lymphocytic lymphoma; uMRD, undetectable minimal residual disease.

Figure 2: Arm D Treatment Schedule

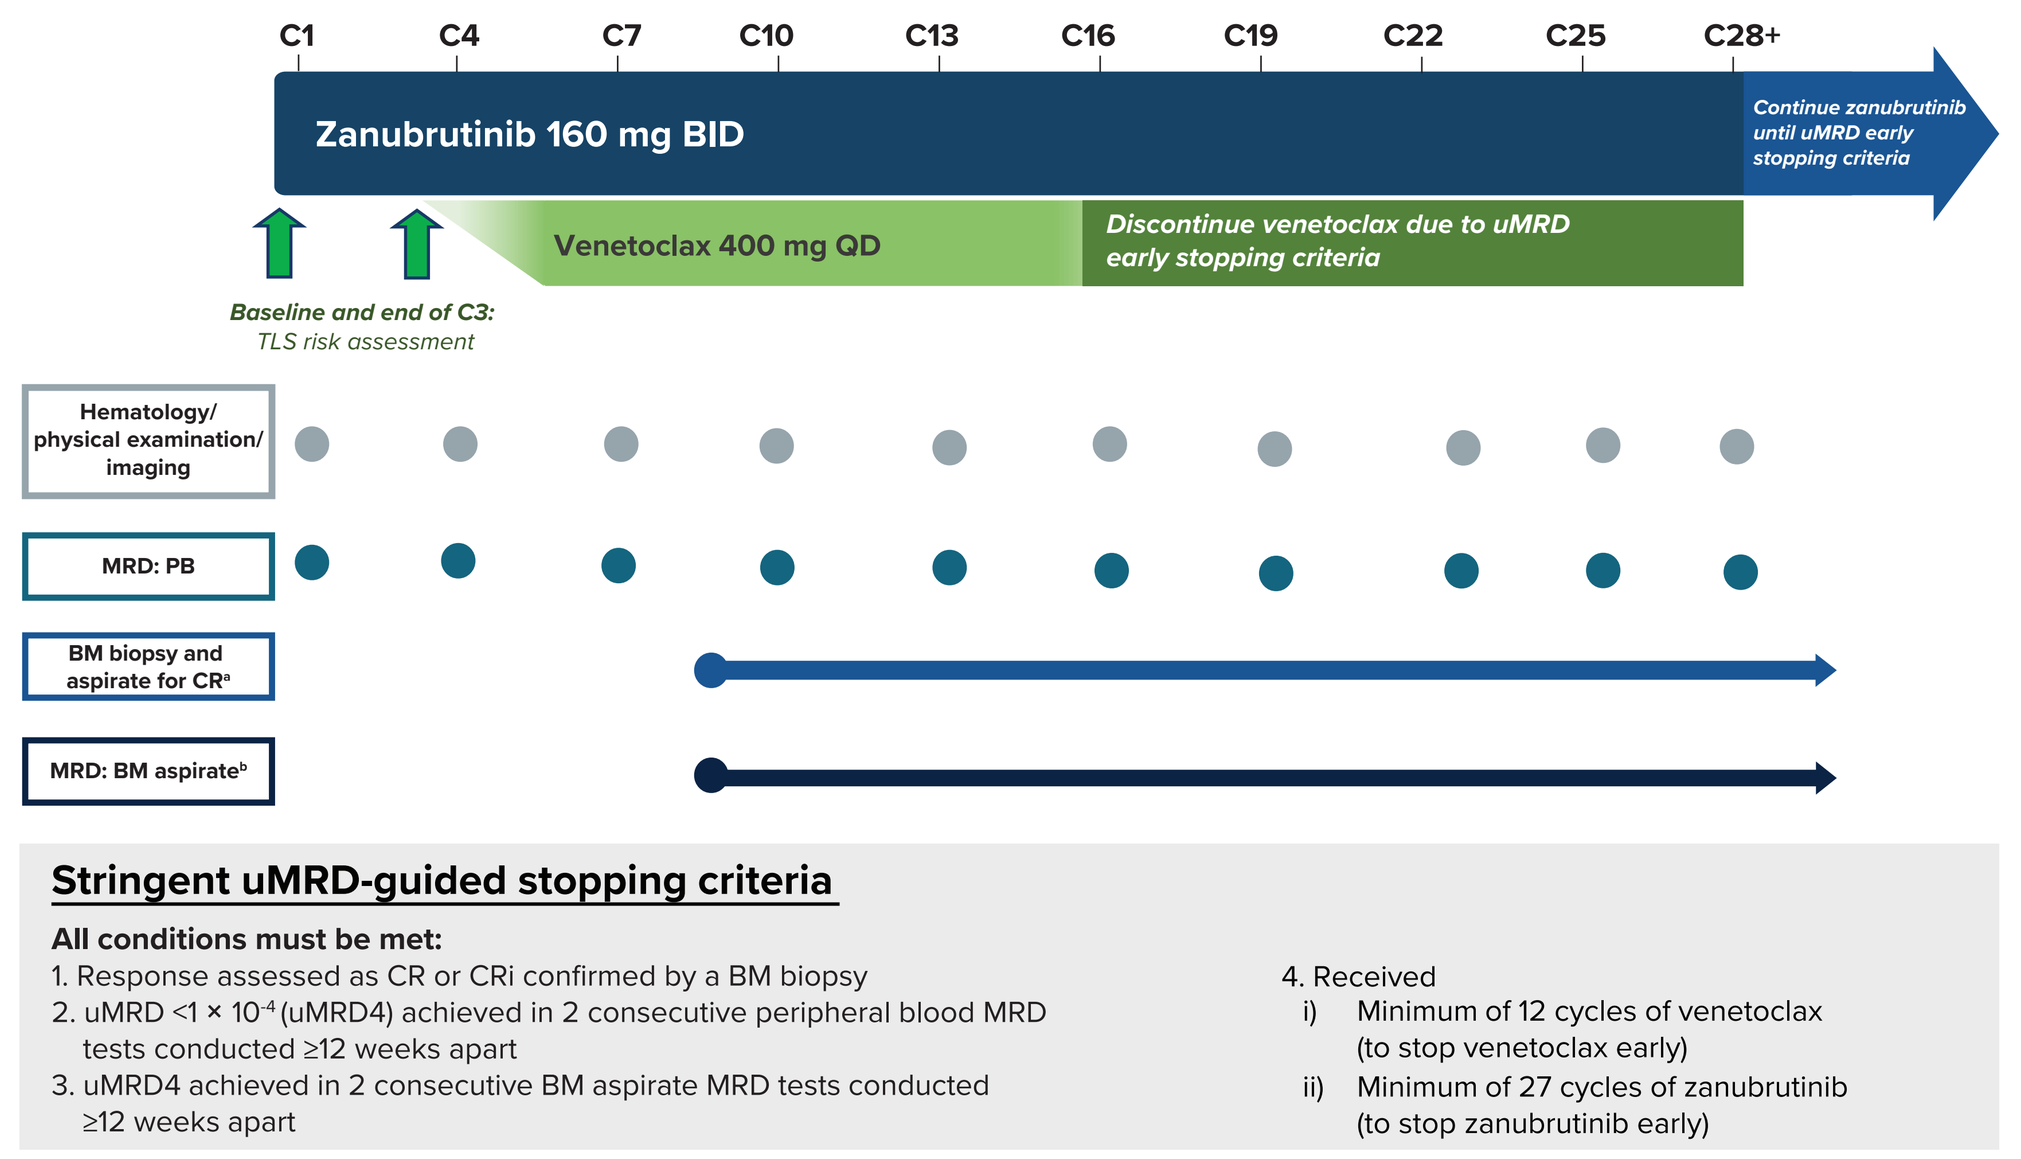
C1 C4 C7 C10 C13 C16 C19 C22 C25 C28+
Zanubrutinib 160 mg BID
Continue zanubrutinib until uMRD early stopping criteria
Venetoclax 400 mg QD
Discontinue venetoclax due to uMRD early stopping criteria
Baseline and end of C3: TLS risk assessment
Hematology/physical examination/imaging
MRD: PB
BM biopsy and aspirate for CRa
MRD: BM aspirateb
Stringent uMRD-guided stopping criteria
All conditions must be met:
1. Response assessed as CR or CRi confirmed by a BM biopsy
2. uMRD <1 × 10⁻⁴ (uMRD4) achieved in 2 consecutive peripheral blood MRD tests conducted ≥12 weeks apart
3. uMRD4 achieved in 2 consecutive BM aspirate MRD tests conducted ≥12 weeks apart
4. Received
i) Minimum of 12 cycles of venetoclax (to stop venetoclax early)
ii) Minimum of 27 cycles of zanubrutinib (to stop zanubrutinib early)

[a]BM biopsy and aspirate were required to confirm a suspected CR/CRi (BM biopsy collection timepoint not defined per protocol), starting after cycle 9 and then annually if needed. [b]Patients with confirmed CR/CRi and 2 consecutive PB-uMRD results ≥12 weeks apart.
Abbreviations: BID, twice daily; BM, bone marrow; C, cycle; CR, complete response; CRi, complete response with incomplete bone marrow recovery; MRD, measurable residual disease; PB, peripheral blood; QD, once daily; TLS, tumor lysis syndrome; uMRD, undetectable measurable residual disease. uMRD4, undetectable measurable residual disease (<1 CLL cell in 10,000 leukocytes at 10^{-4} sensitivity by 8-color flow cytometry).

RESULTS

Disposition and baseline characteristics

- Between November 2019 and July 2022, 114 patients were enrolled into SEQUOIA Arm D
- As of September 16, 2024, 85 patients remained on zanubrutinib monotherapy
 - Zanubrutinib was discontinued in patients mainly due to adverse events (n=9; 8%), uMRD early stopping criteria met (n=8; 7%) and progressive disease (n=6; 5%)
 - Venetoclax was discontinued primarily due to completion of its 24 cycles, per protocol (n=87; 76%), uMRD early stopping criteria met (n=8; 7%) and adverse events (n=7; 6%)
- Baseline demographic and disease characteristics are shown in **Table 1**

Table 1. Baseline Demographics and Clinical Characteristics

	With del(17p) and/or *TP53*mut (n=66)	Without del(17p) and *TP53*mut (n=47)	All patients (N=114)[a]
Age, median (range), years	66 (26-87)	67 (36-80)	67 (26-87)
≥65 years, n (%)	36 (55)	32 (68)	68 (60)
Male, n (%)	34 (52)	29 (62)	64 (56)
ECOG PS 0-1, n (%)	64 (97)	47 (100)	112 (98)
CIRS >6	10 (15)	11 (23)	21 (18)
CrCl, mL/min, median (range)	73 (25-253)	82 (41-355)	76 (25-355)
SLL, n (%)	3 (5)	3 (6)	6 (5)
Binet stage C, n (%)[b]	30 (48)	16 (36)	46 (43)
Bulky disease, n (%)			
LDi ≥5 cm	29 (44)	19 (40)	49 (43)
LDi ≥10 cm	5 (8)	1 (2)	6 (5)
Median time from initial diagnosis, months	19.3	42.2	28.5
TP53 mutated, n (%)	49 (74)	0	49 (43)
del(17p), n (%)	59 (89)	0	59 (52)
del(17p) and *TP53* mutated, n (%)	42 (64)	0	42 (37)
IGHV unmutated, n (%)[c]	56 (85)	30 (64)	86 (75)
Complex karyotype, n (%)			
≥3 abnormalities	33 (50)	14 (30)	47 (41)
≥5 abnormalities	24 (36)	2 (4)	26 (23)

[a]One patient had a missing *TP53* result (via central laboratory). [b]Binet Stage was assessed at study entry in patients with CLL. [c]Four patients had a missing IGHV result, one due to missed sample collection and three due to insufficient quantity of sample.
Abbreviations: CIRS, Cumulative Illness Rating Scale; CrCl, creatinine clearence; ECOG PS, Eastern Cooperative Oncology Group performance status; IGHV, immunoglobulin heavy-chain variable region; LDi, longest diameter; mut, mutation.

Efficacy

Best overall response

- The rates of CR/CRi were similar regardless of del(17p)/*TP53* mutational status: 47% with del(17p) and/or *TP53* mutation and 49% without del(17p) and *TP53* mutation (**Figure 3**)

Figure 3. Best ORR

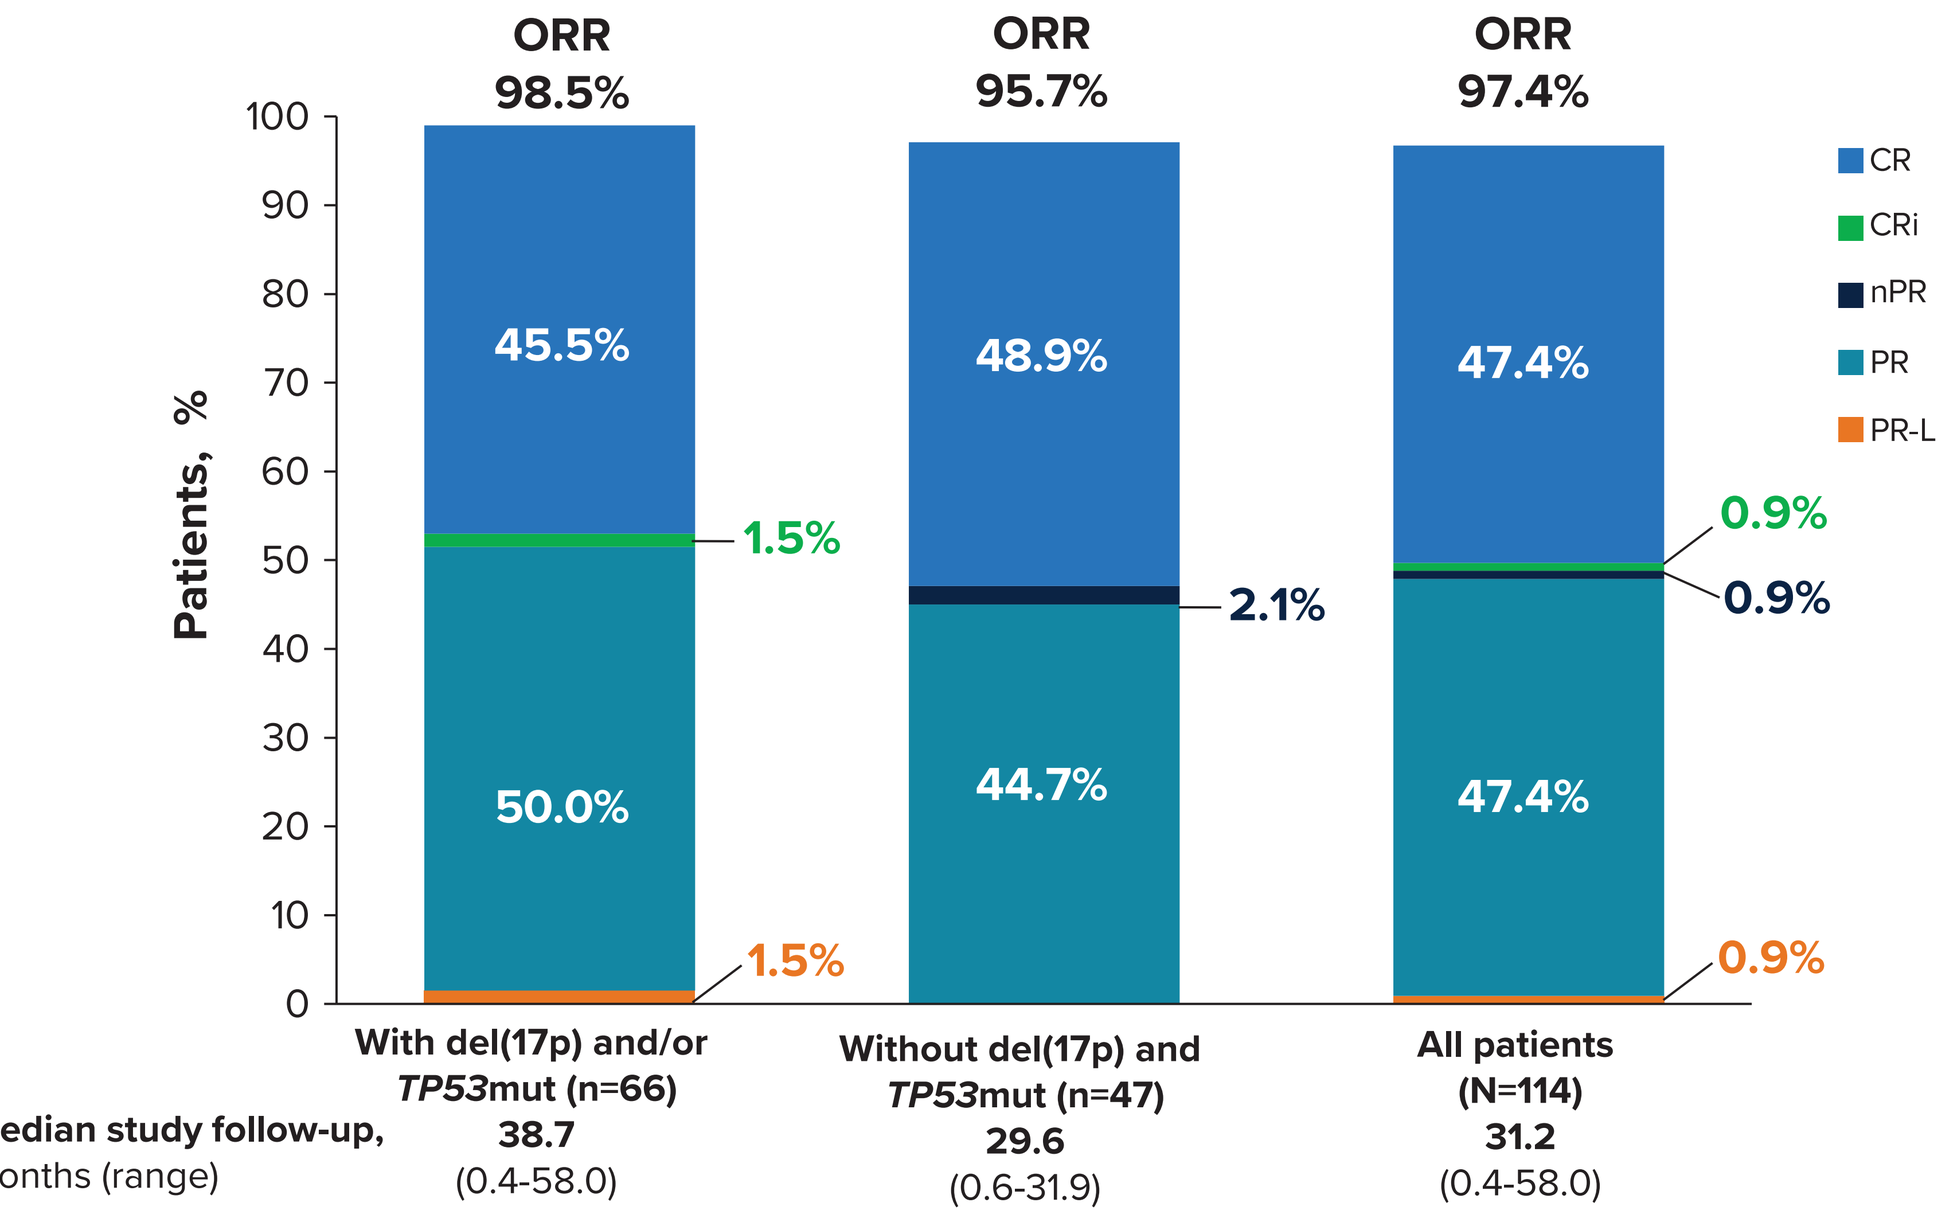
ORR 98.5%
ORR 95.7%
ORR 97.4%
Patients, %
45.5%
1.5%
50.0%
1.5%
48.9%
2.1%
44.7%
47.4%
0.9%
0.9%
47.4%
0.9%
CR
CRi
nPR
PR
PR-L
With del(17p) and/or TP53mut (n=66)
Without del(17p) and TP53mut (n=47)
All patients (N=114)
Median study follow-up, months (range)
38.7 (0.4-58.0)
29.6 (0.6-31.9)
31.2 (0.4-58.0)

Abbreviations: CR, complete response; CRi, complete response with incomplete hematopoietic recovery; mut, mutation; nPR, nodular partial response; ORR, overall response rate; PR, partial response; PR-L, partial response with lymphocytosis.

uMRD in peripheral blood

- Median time to first perhiperal blood (PB)-uMRD was 19 (range, 3-47) months in patients with del(17p) and/or *TP53* mutation and 11 (range, 6-25) months in patients without del(17p) and *TP53* mutation
- Best PB-uMRD in the peripheral blood was similar regardless of mutational status (**Figure 4**)
- The rate of PB-uMRD increased from cycle 16 and cycle 28 in both subgroups (**Table 2**)

Table 2: Best uMRD in Peripheral Blood[a]

	With del(17p) and/or *TP53*mut (n=66)	Without del(17p) and *TP53*mut (n=47)
Best PB-uMRD, n (%)		
By cycle 16	14 (21)	20 (43)
By cycle 28	32 (49)	28 (60)

[a]Best uMRD in peripheral blood was defined as achieving uMRD in the peripheral blood at ≥1 timepoint.
Abbreviations: mut, mutation; PB, peripheral blood; uMRD, undetectable minimal residual disease.

Figure 4: Best uMRD in Peripheral Blood[a]

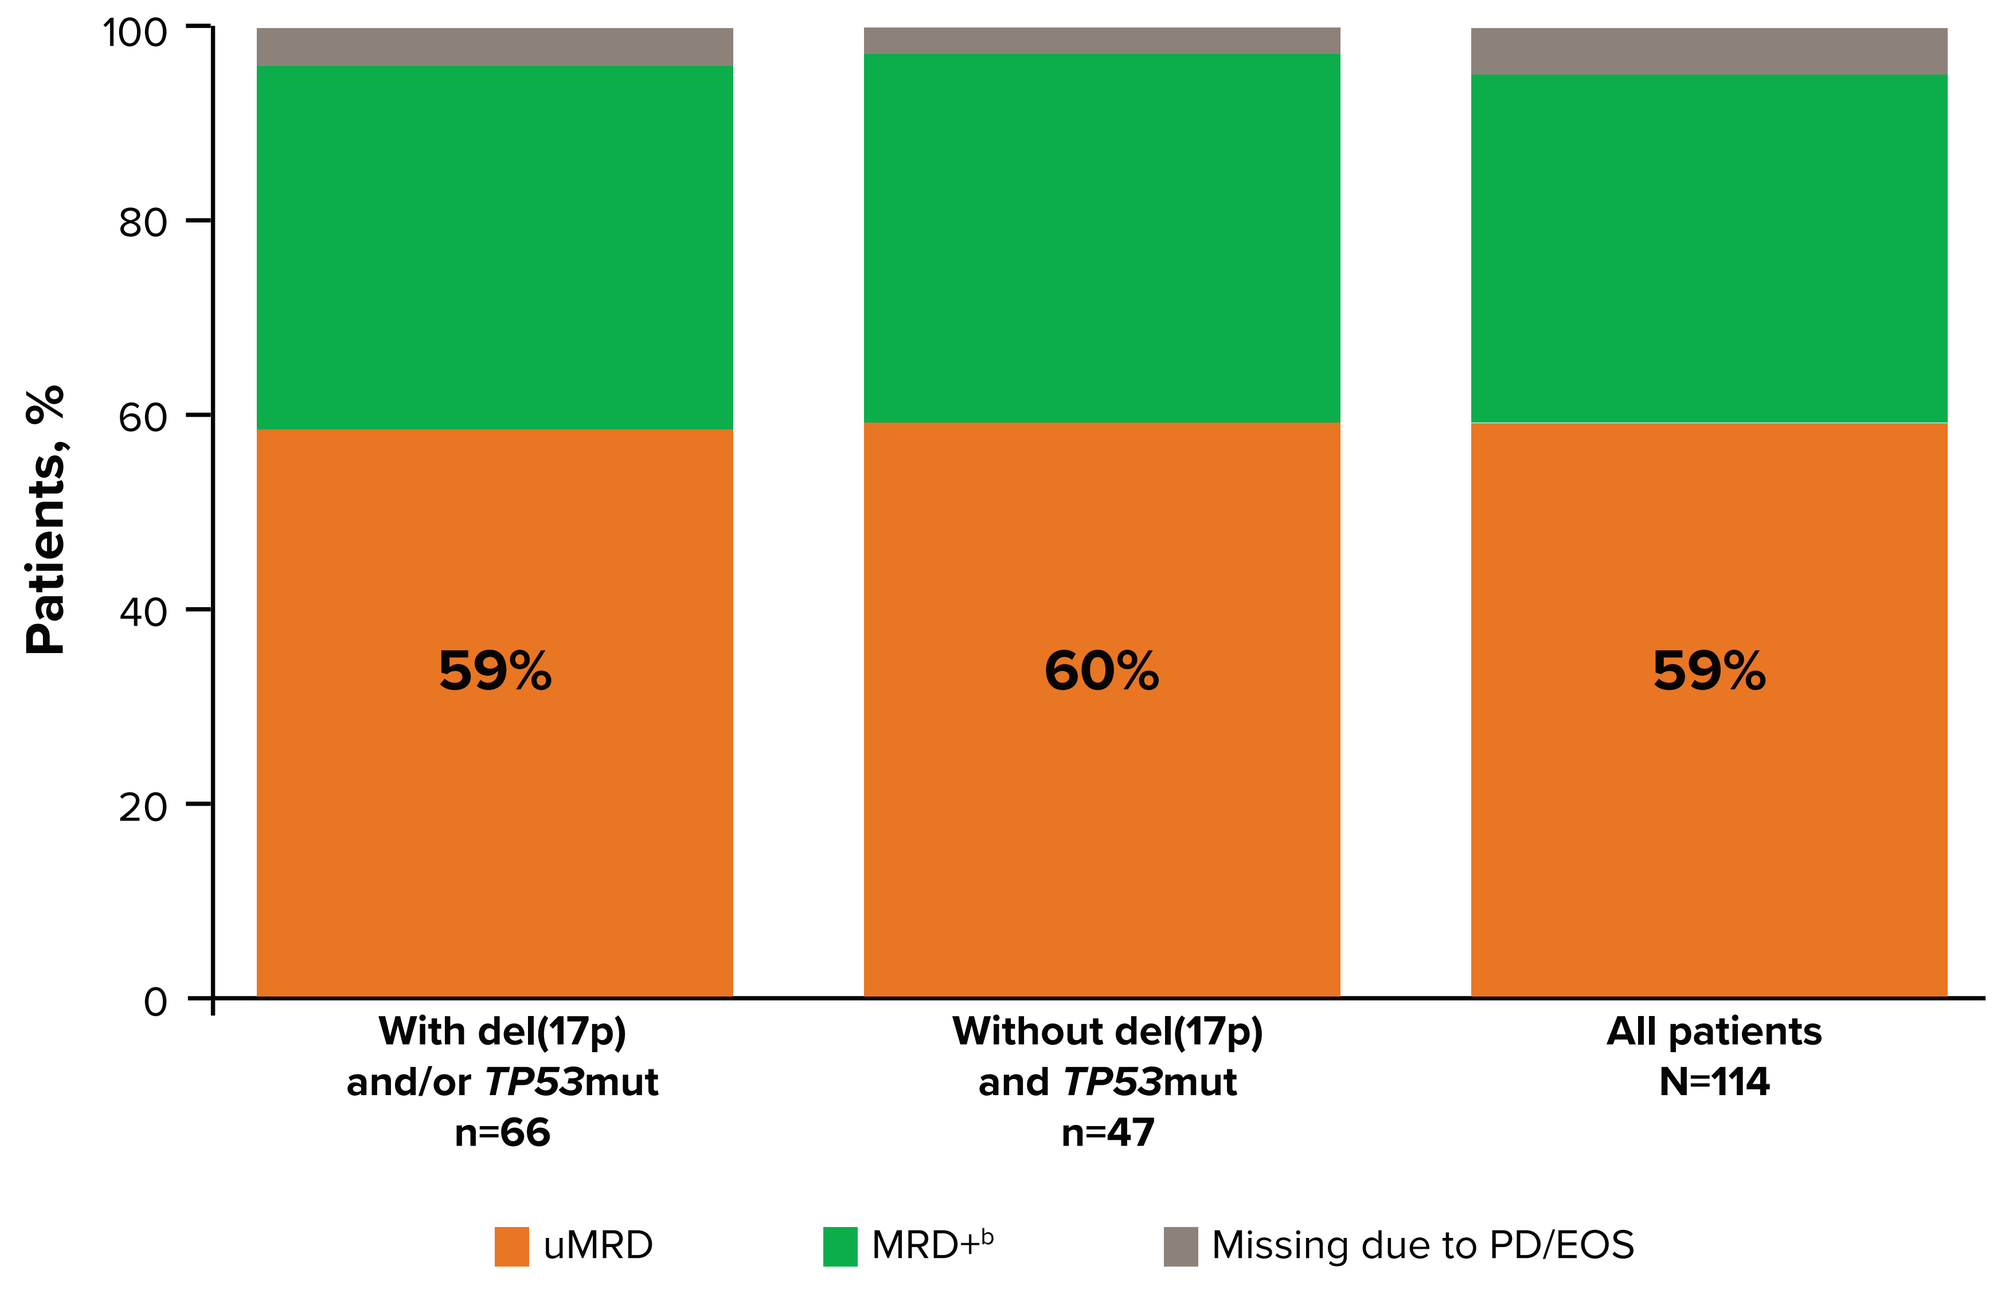
Patients, %
59%
60%
59%
With del(17p) and/or TP53mut n=66
Without del(17p) and TP53mut n=47
All patients N=114
uMRD
MRD+b
Missing due to PD/EOS

[a]Best uMRD in peripheral blood was defined as achieving uMRD in the peripheral blood at ≥1 timepoint. [b]MRD $\geq 1 \times 10^{-4}$
Abbreviations: EOS, end of study; mut, mutation; PB, peripheral blood; PD, progressive disease; uMRD, undetectable minimal residual disease.

PFS

- With a median follow-up of 31.2 (range, 0.4-58.0) months in all patients, the median PFS was not reached; the 24-month PFS rate was 92% (95% CI, 85-96) (**Figure 5A**)
- The median follow-up was 38.7 (range, 0.4-58.0) months in patients with del(17p) and/or *TP53* mutation and 29.6 (range, 0.6-31.9) months in patients without del(17p) and *TP53* mutation
 - The 24-month PFS rate (95% CI) was 94% (85-98) and 89% (76-95), respectively (**Figure 5B**)
- Of the 11 patients who discontinued after meeting stringent uMRD-guided stopping criteria, only one patient with del(17p) has progressed

Figure 5. PFS

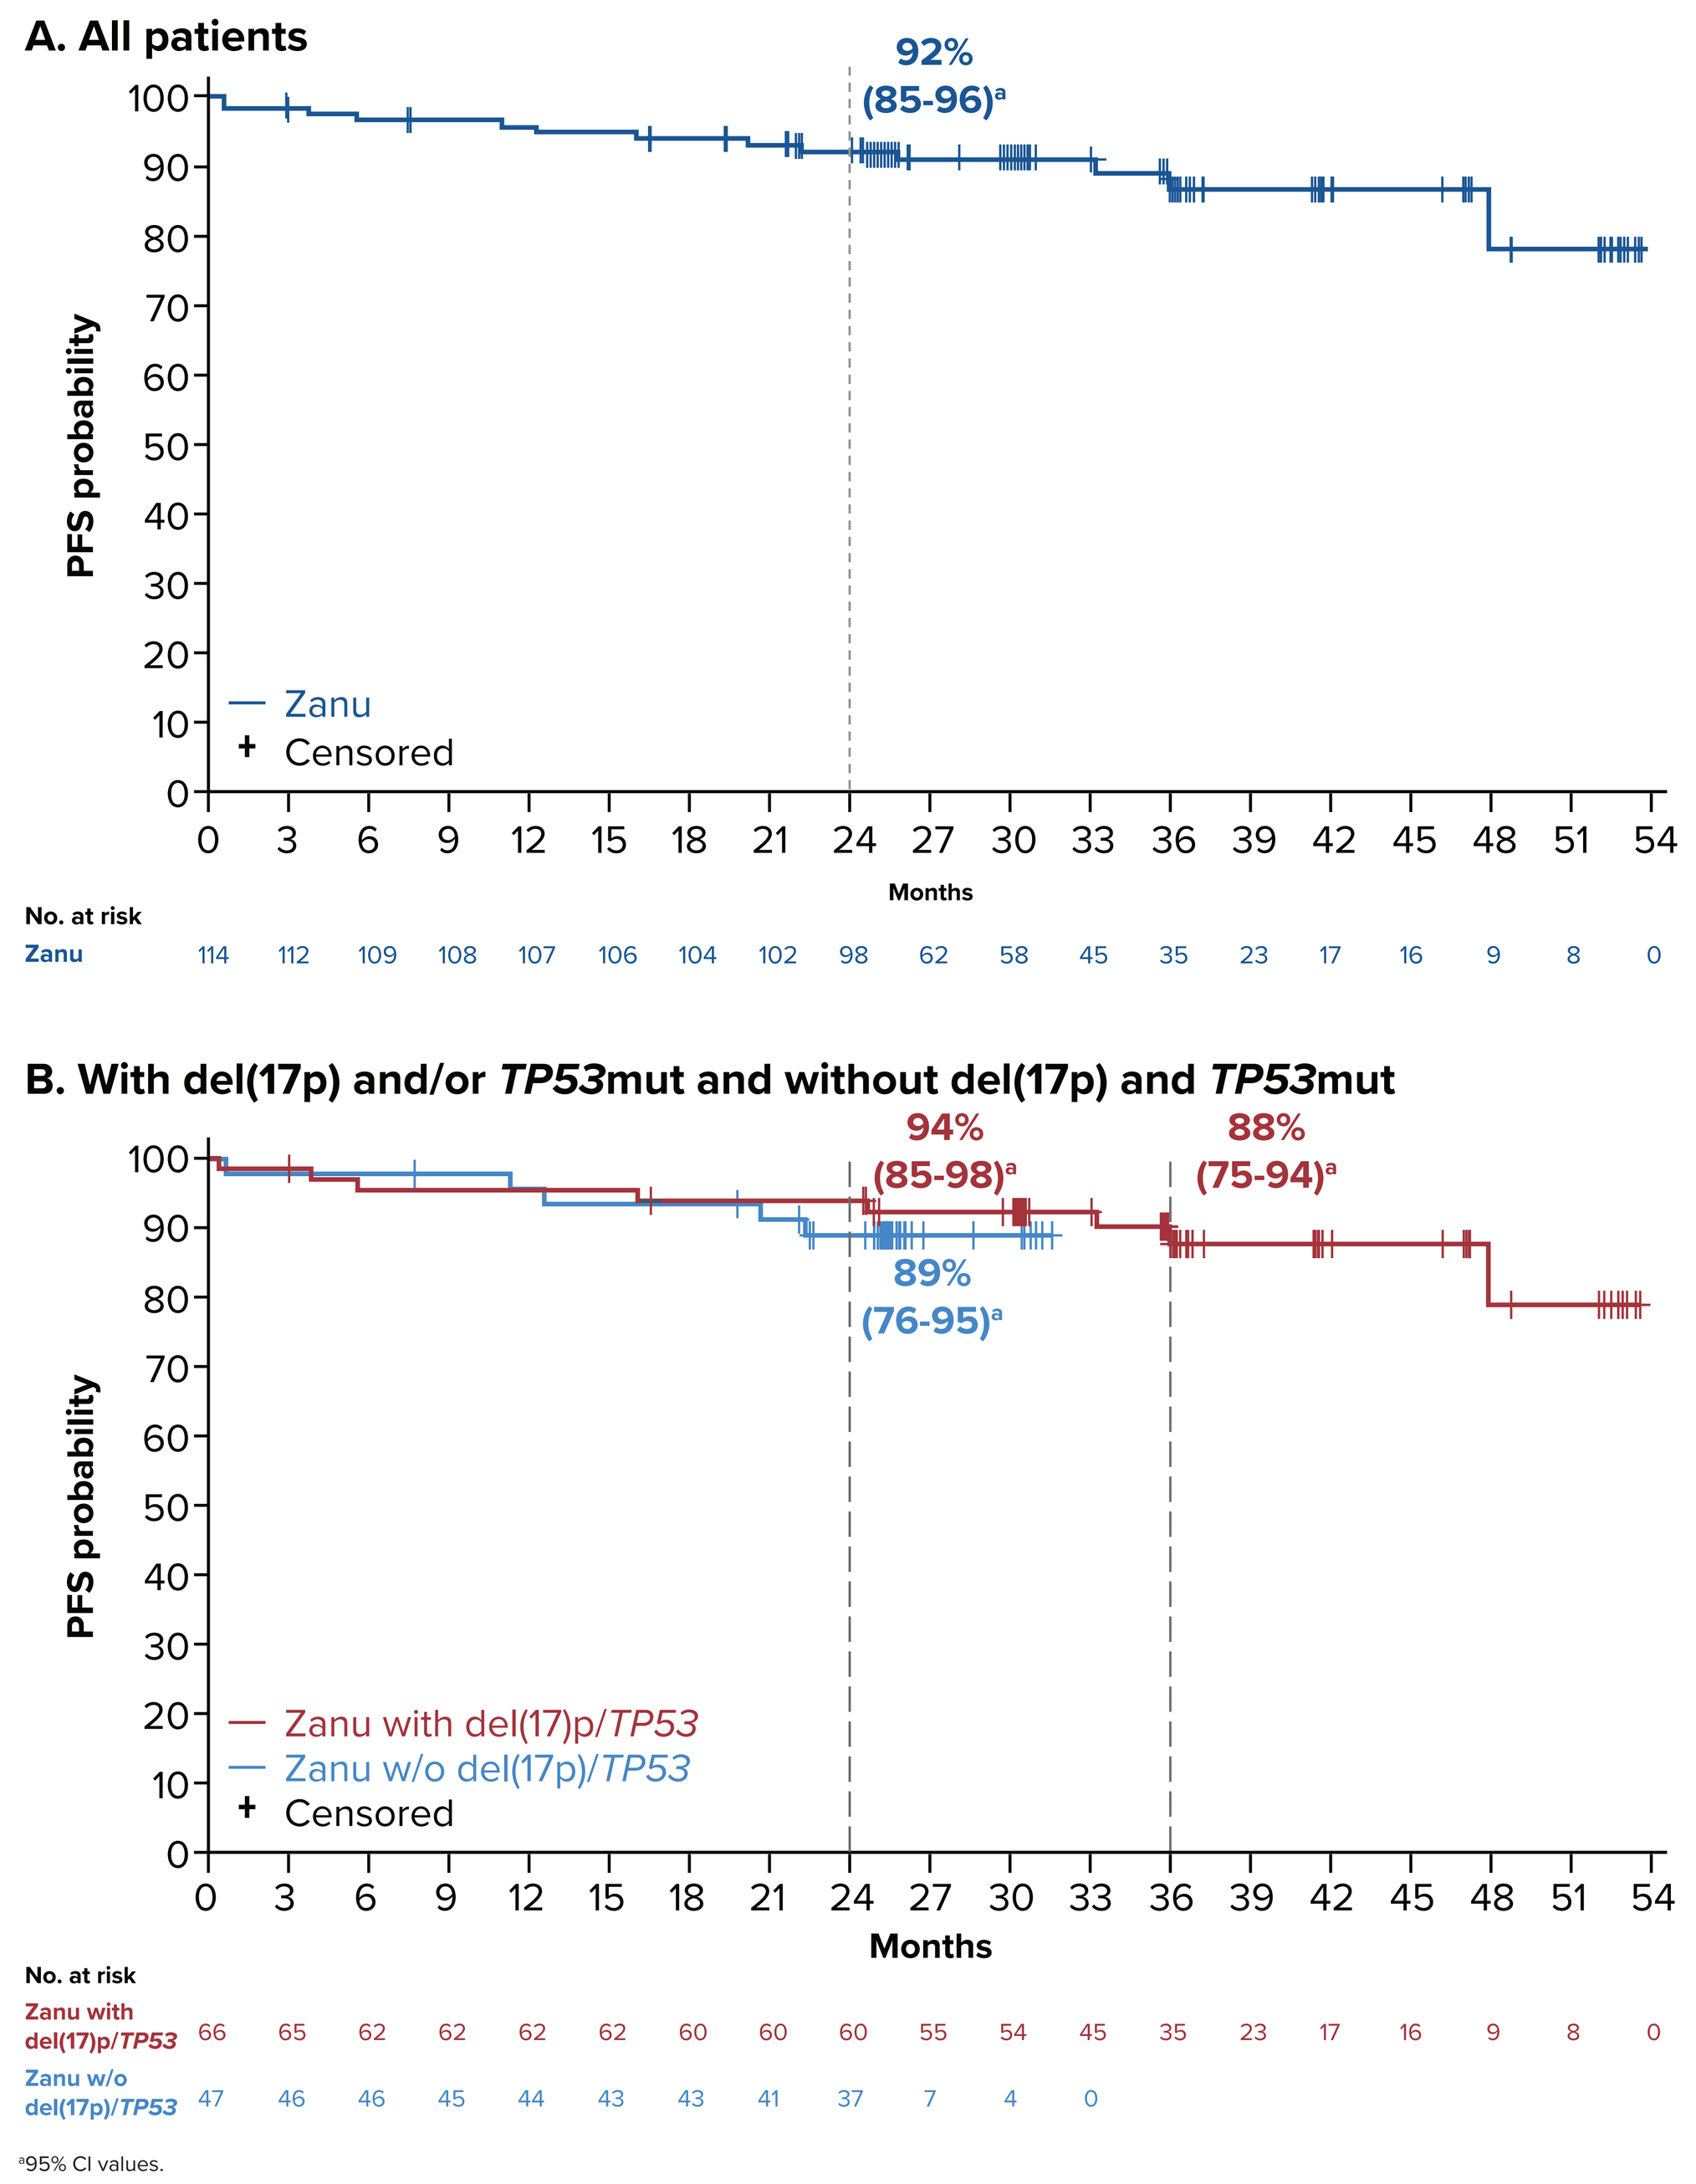
A. All patients
92% (85-96)a
PFS probability
Zanu
Censored
Months
No. at risk
Zanu 114 112 109 108 107 106 104 102 98 62 58 45 35 23 17 16 9 8 0
B. With del(17p) and/or TP53mut and without del(17p) and TP53mut
94% (85-98)a
88% (75-94)a
89% (76-95)a
PFS probability
Zanu with del(17p)/TP53
Zanu w/o del(17p)/TP53
Censored
Months
No. at risk
Zanu with del(17p)/TP53 66 65 62 62 62 62 60 60 60 55 54 45 35 23 17 16 9 8 0
Zanu w/o del(17p)/TP53 47 46 46 45 44 43 43 41 37 7 4 0

[a]95% CI values.
Abbreviations: ITT, intention-to-treat; mut, mutation; PFS, progression-free survival; w/o, without; Zanu, zanubrutinib.

Safety

- The most common treatment-emergent adverse events (TEAEs) and TEAEs of special interest are presented in **Figure 6**
- Five deaths occurred in this study due to adverse events[a]; no COVID-19-related deaths occurred

Figure 6. TEAEs and TEAEs of Special Interest

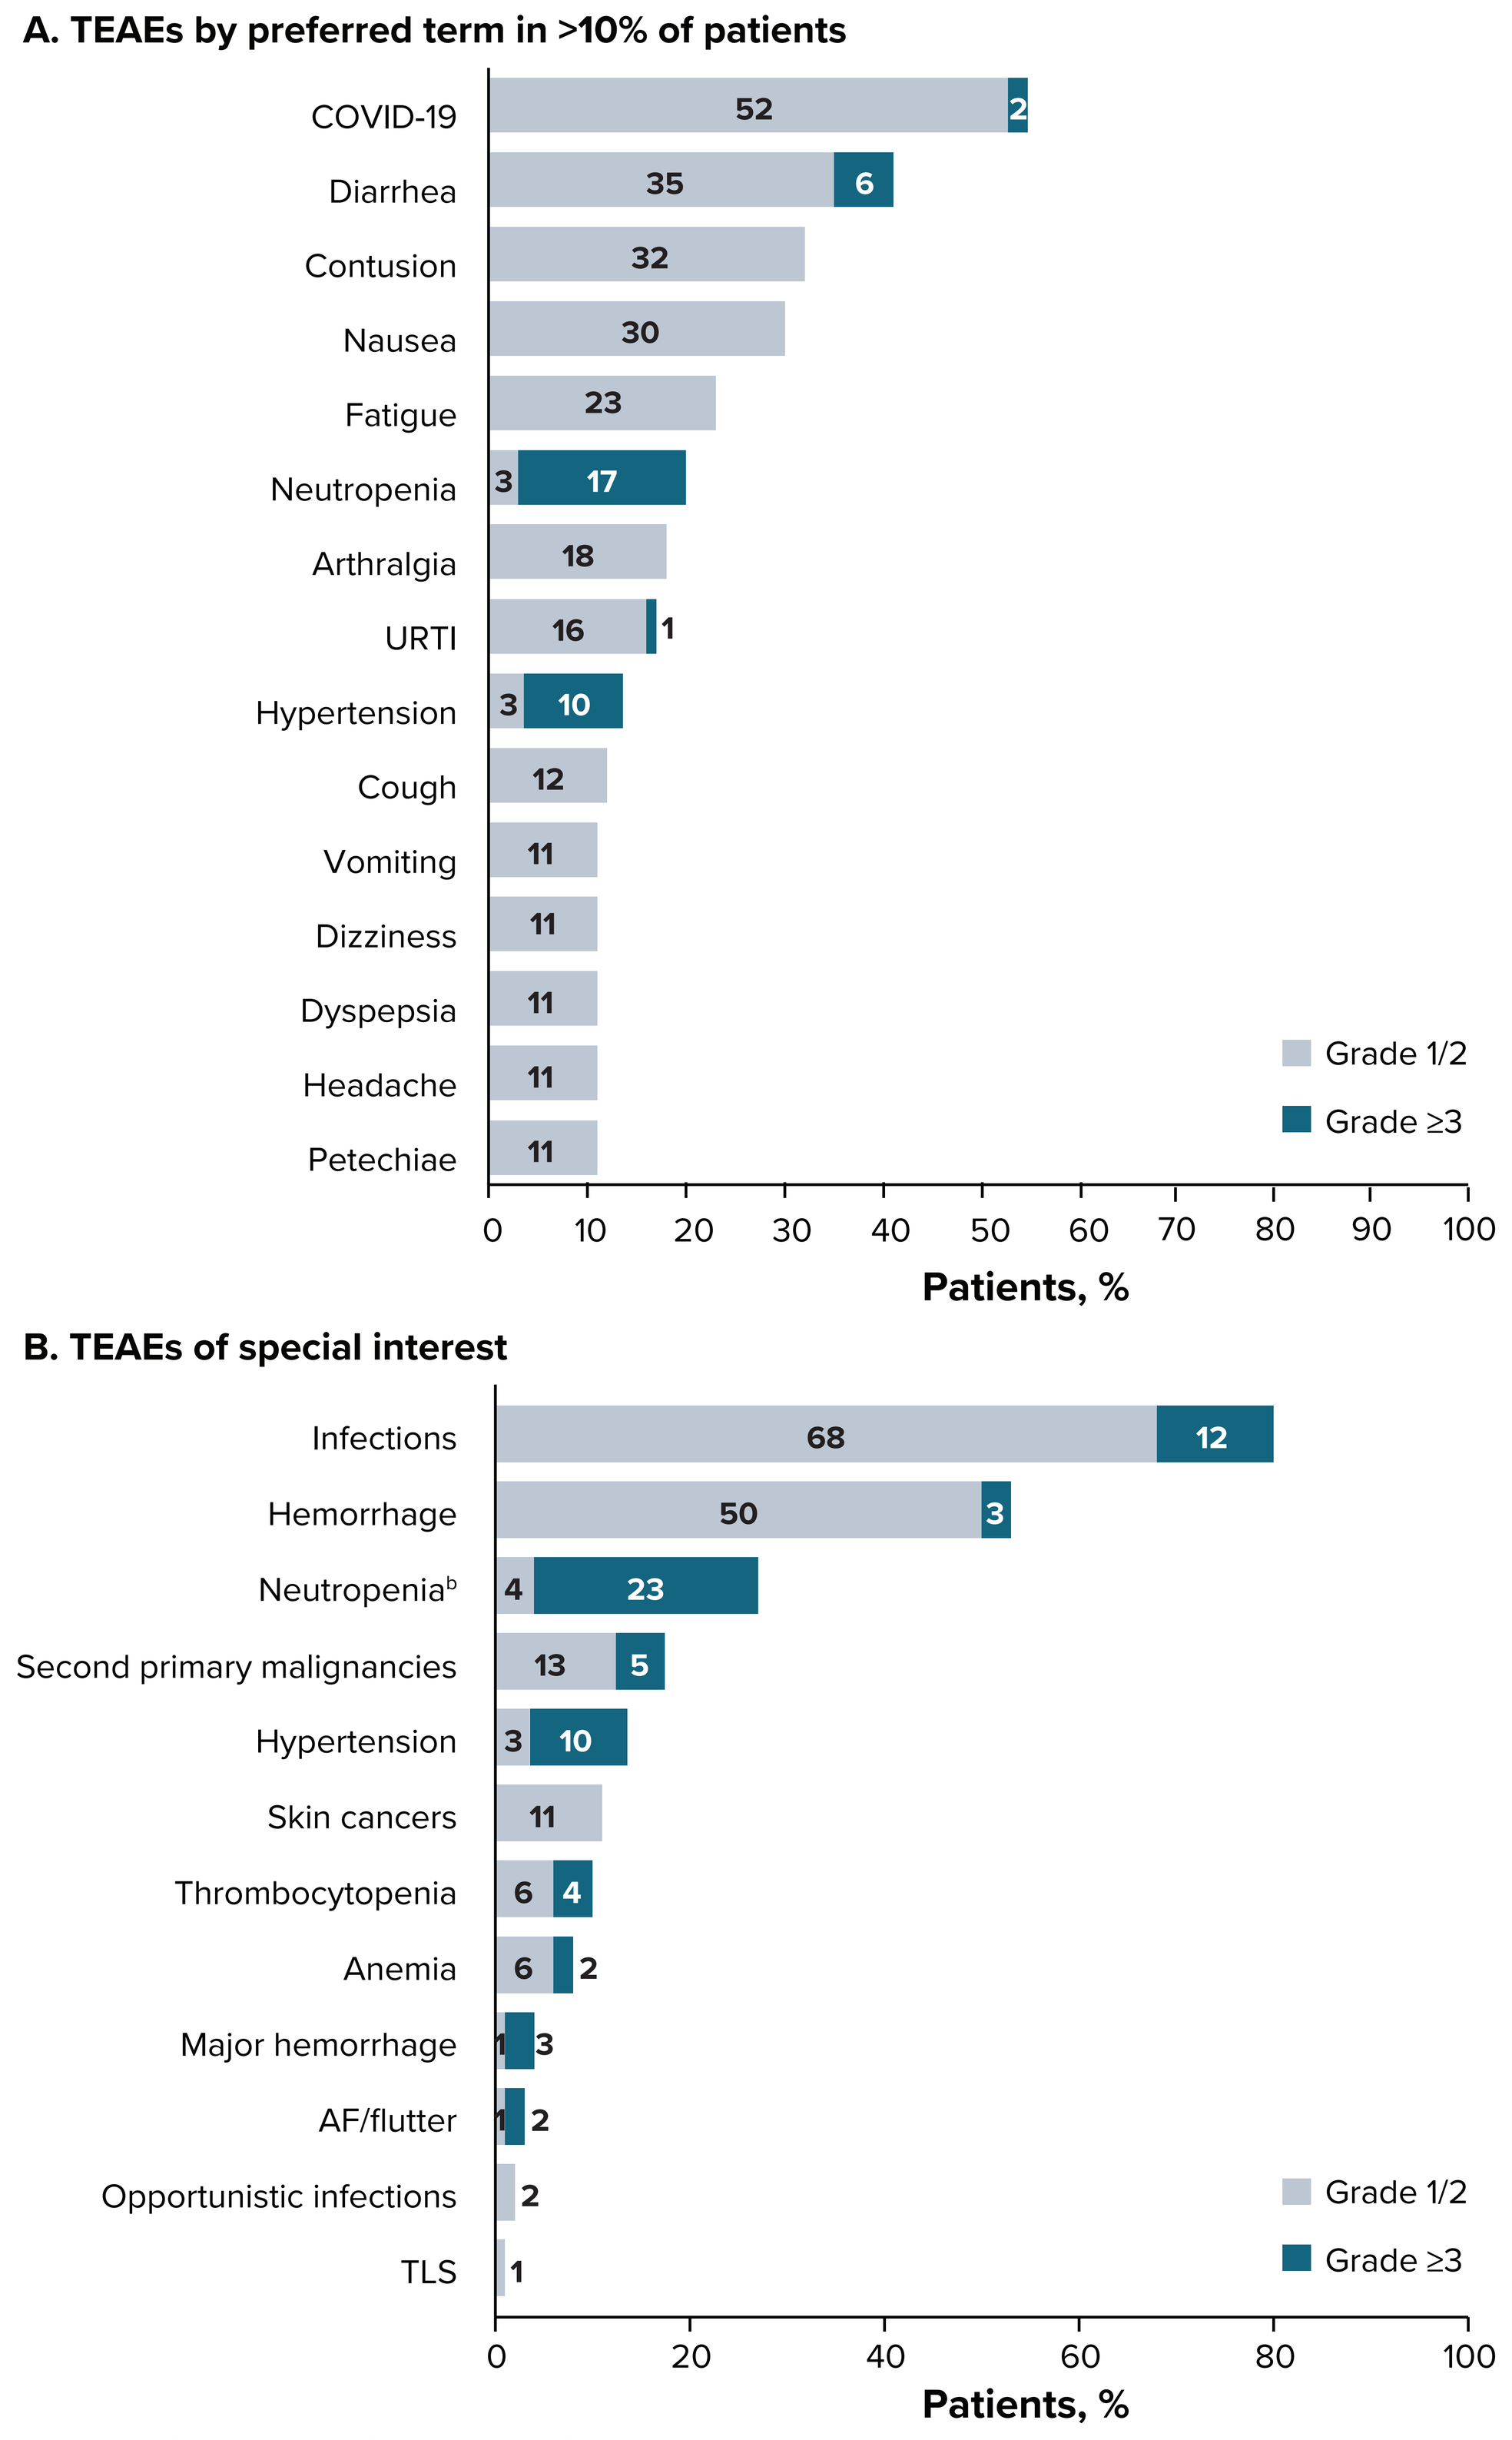
A. TEAEs by preferred term in >10% of patients
COVID-19 52 2
Diarrhea 35 6
Contusion 32
Nausea 30
Fatigue 23
Neutropenia 3 17
Arthralgia 18
URTI 16 1
Hypertension 3 10
Cough 12
Vomiting 11
Dizziness 11
Dyspepsia 11
Headache 11
Petechiae 11
Grade 1/2
Grade ≥3
Patients, %
B. TEAEs of special interest
Infections 68 12
Hemorrhage 50 3
Neutropeniab 4 23
Second primary malignancies 13 5
Hypertension 3 10
Skin cancers 11
Thrombocytopenia 6 4
Anemia 6 2
Major hemorrhage 1 3
AF/flutter 1 2
Opportunistic infections 2
TLS 1
Grade 1/2
Grade ≥3
Patients, %

[a]One patient experienced a fatal road traffic accident leading to intracranial hemorrhage and intra-abdominal hemorrhage. One patient experienced death due to pneumonia and septic shock. Other TEAEs leading to death included lung carcinoma, gallbladder carcinoma, and intracranial hemorrhage in a patient with concomitant direct oral anticoagulant use and prior zanubrutinib discontinuation. [b]Included neutropenia, neutrophil count decreased and agranulocytosis.
Abbreviations: AEs, adverse event; AF, atrial fibrillation; TEAE, treatment-emergent adverse events; TLS, tumor lysis syndrome; URTI, upper respiratory tract infection.

REFERENCES

1. Guo Y, et al. *J Med Chem.* 2019;62(17):7923-7940.
2. Brukinsa (zanubrutinib). Prescribing information. BeiGene USA; 2024.
3. Brukinsa (zanubrutinib). Summary of product characteristics. BeiGene Ireland Limited; 2024.
4. Brown JR, et al. *Blood.* 2024;144(26):2706-2717.
5. Tam CS, et al. *Haematologica.* 2021;106(9):2354-2363.
6. Brown JR et al. *N Engl J Med.* 2025. 392(8):748-762.
7. Tam CS, et al. *Blood.* 2022;139(22):3278-3289.
8. Shadman M, et al. *J Clin Oncol.* 2025;43(7):780-787.
9. Tam CS, et al. *Lancet Oncol.* 2022;23(8):1031-1043.
10. Hallek M, et al. *Blood.* 2008;111(12):5446–5456.
11. Cheson BD, et al. *J Clin Oncol.* 2012;30(23):2820-2822.
12. Cheson BD, et al. *J Clin Oncol.* 2014;32(27):3059-3967.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers.

This study was sponsored by BeOne Medicines Ltd. Medical writing support was provided by Manoshi Nath, MSc, of Nucleus Global, an Inizio company, and supported by BeOne Medicines.

DISCLOSURES

MS: Consultant: AbbVie, Genentech, AstraZeneca, Genmab, Janssen, BeOne Medicines Ltd, BMS, MorphoSys/Incyte, Kite Pharma, Lilly, Fate Therapeutics, Nurix, Merck; Research funding: Mustang Bio, Genentech, AbbVie, BeOne Medicines Ltd, AstraZeneca, Genmab, Morphosys/Incyte, Vincerx; Stock: Koi Biotherapeutics; Employment: BMS (spouse). **TM:** Honoraria: BeOne Medicines Ltd, AstraZeneca, Sobi, Roche, Janssen, AbbVie, Lilly; Consultant: AbbVie, BeOne Medicines Ltd, Sobi, Alexion, Novartis, Janssen, AstraZeneca, Lilly, Roche; Research grants: Janssen, AbbVie; Travel, accommodations, or expenses: Alexion, BeOne Medicines Ltd, AbbVie, Janssen, AstraZeneca; Advisory board: AbbVie, BeOne Medicines Ltd, AstraZeneca, Janssen. **SM:** Consultant, membership on an entity's board of directors or advisory committee, research funding and/or speakers bureau: AstraZeneca, BeOne Medicines Ltd, Lilly, Janssen Pharmaceuticals, Juno/BMS, AbbVie, Genentech. **ML:** Honoraria: Janssen, AbbVie; Advisory board: Janssen, Sobi, AbbVie, Recordati. **MT:** Consulting or advisory role: Lilly, Roche, Jansen-Cilag; Travel, accomodations, expenses: Jansen-Cilag, Takeda. **TR:** Janssen, Gilead, AstraZeneca; Research funding: Lilly, Janssen, AstraZeneca, Gillead, Glaxo; Travel, accomodations, expenses: AstraZeneca. **IWF:** Employment: OneOncology; Research grants (all payments made to institution): AbbVie, AstraZeneca, BeOne Medicines Ltd, BMS, Celgene, City of Hope National Medical Center, Epizyme, Fate Therapeutics, Genentech, Gilead Sciences, IGM Biosciences InnoCare Pharma, Incyte, Janssen, Kite Pharma, Loxo, Marker Therapeutics, Merck, MorphoSys, Myeloid Therapeutics, Novartis, Nurix, Pfizer, Roche, Seattle Genetics, TG Therapeutics, Vincerx Pharma, 2seventy bio; Consultant fees (all payments made to physician): Abbvie, BeOne Medicines Ltd, Genentech, Genmab, KITE, Vincerx; Board of Directors or advisory committee: OneOncology, Vincerx Adv Committee. **JRB:** Consultant: Abbvie, Acerta/AstraZeneca, Alloplex Biotherapeutics, BeOne Medicines Ltd, BMS, EcoR1, Galapagos NV, Genentech/Roche, Grifols Worldwide Operations, InnoCare Pharma Inc, Kite Pharma, Loxo/Lilly, Magnet Biomedicine, Merck, Pharmacyclics; Research funding: BeOne Medicines Ltd, Gilead, iOnctura, Loxo/Lilly, MEI Pharma, TG Therapeutics; Royalties: UpToDate; Data Safety Monitoring Board for Grifols Therapeutics. **PG:** Honoraria: AbbVie, AstraZeneca, BeOne Medicines Ltd, BMS, Janssen, Galapagos, Lilly/Loxo, MSD, Roche; Research funding: AbbVie, AstraZeneca, BMS, Janssen. **EF:** Consulting or advisory role: AbbVie, BeOne Medicines Ltd, Johnson & Johnson, Gilead, AstraZeneca; Travel, accomodations, expenses: BeOne Medicines Ltd, Johnson & Johnson, Gilead. **CST:** Honoraria: AbbVie, Janssen, BeOne Medicines Ltd, AstraZeneca. **WJ:** Honoraria and consulting or advisory role: BeOne Medicines Ltd, AstraZeneca, Janssen. **WJ:** Consultant and research funding: AbbVie, AstraZeneca, BeOne Medicines Ltd, Jansen Cilag, Lilly, Roche, Takeda. **LX:** Employment and may own stock: BeOne Medicines Ltd. **TT:** Employment and may own stock: BeOne Medicines Ltd. **SA:** Employment, may own stock, travel, accommodations, or expenses: BeOne Medicines Ltd. **JH:** Employment: BeOne Medicines Ltd, Genentech; Stocks or other ownership: BeOne Medicines Ltd, Roche. **AT:** Consulting or advisory role, honoraria, and travel, accomodations, expenses: AbbVie, BeOne Medicines Ltd, Lilly, AstraZeneca, J&J.

CORRESPONDENCE: Mazyar Shadman, mshadman@fredhutch.org

Presented at the EHA2025 Congress; June 12-15, 2025; Milan, Italy
Data originally presented at the 2025 ASCO Annual Meeting; May 30-June 3, 2025; Chicago, IL, USA. Abstract 7009