Updated Safety & Antileukemic Activity Data of Sonrotoclax (BGB-11417), a Potent and Selective BCL2 Inhibitor, in Treatment-Naive Patients With Acute Myeloid Leukemia Unfit for Intensive Chemotherapy

PF477

Jake Shortt,[1] Chun Yew Fong,[2] Sophie Leitch,[3] Guru Subramanian Guru Murthy,[4] Shuh Ying Tan,[5] Xiaojun Huang,[6] Pau Montesinos,[7] Uwe Platzbecker,[8] Courtney DiNardo,[9] Si Cheng,[10] Vaibhav Mundra,[11] Rui Li,[10] Yu Liu,[12] Oleg Zernovak,[11] Amit Agarwal,[11] Andrew H. Wei[13]

[1]Monash Health and Monash University, Clayton, VIC, Australia; [2]Austin Health, Melbourne, VIC, Australia; [3]Te Whatu Ora, Health New Zealand, Waitemata, Auckland, New Zealand; [4]Medical College of Wisconsin, Milwaukee, WI, USA; [5]St Vincent's Hospital, Melbourne, VIC, Australia; [6]Peking University People's Hospital, Beijing, China; [7]Hospital Universitari I Politècnic La Fe, Valencia, Spain; [8]Universitaetsklinikum Leipzig Aör, Leipzig, Germany; [9]MD Anderson Cancer Center, Houston, TX, USA; [10]BeOne Medicines Ltd, Shanghai, China; [11]BeOne Medicines Ltd, San Carlos, CA, USA; [12]BeOne Medicines Ltd, Beijing, China; [13]Peter MacCallum Cancer Centre, Royal Melbourne Hospital, Melbourne, VIC, Australia

CONCLUSIONS

- Sonrotoclax + azacitidine combination treatment was generally well tolerated and demonstrated antileukemic activity in patients with TN unfit AML across all dose cohorts
 - DLTs occurred in four patients (grade 4 neutropenia, n=1; grade 4 thrombocytopenia, n=4)
 - The ORR was 74.7%; CR was achieved by 50.6% and CR/CRh by 59.5%
- The safety stopping criteria have not been met in any of the dose cohorts
 - Shorter sonrotoclax + azacitidine treatment schedules (<21 d) were well tolerated with a median RDI of >80%
- Exploratory exposure-response analysis in 14-d cohorts showed that antileukemic activity at exposures associated with an 80-mg dose was ≈2-fold lower than exposures associated with 160-mg or 320-mg dose
- Follow-up evaluation of 14-d dosing cohorts is ongoing in 80-mg, 160-mg, and 320-mg cohorts to determine the recommended phase 2 dose
- Data for patients with relapsed/refractory AML in this study are presented in poster PF491

INTRODUCTION

- Acute myeloid leukemia (AML), the most common acute form of leukemia in adults, has an aggressive disease course[1,2]
- Combination treatment with venetoclax, a B-cell lymphoma 2 (BCL2) inhibitor, and azacitidine has improved outcomes in treatment-naive patients with AML unfit for intensive chemotherapy (TN AML)[3]; however, relapse is common, and prognosis is suboptimal[4,5]
- Sonrotoclax (BGB-11417), a next-generation BCL2 inhibitor, is a more selective and pharmacologically potent inhibitor of BCL2 than venetoclax, with a shorter half-life and no drug accumulation[6]
- Updated safety and antileukemic activity data of sonrotoclax + azacitidine in TN AML from the phase 1b part of the BGB-11417-103 study are presented

METHODS

- BGB-11417-103 (NCT04771130) is an ongoing, phase 1b/2, global, dose-finding and -expansion study evaluating the safety and antileukemic activity of sonrotoclax + azacitidine in patients with AML, myelodysplastic syndromes (MDS), or MDS/myeloproliferative neoplasms (**Figure 1**)

Figure 1. BGB-11417-103 Study Design

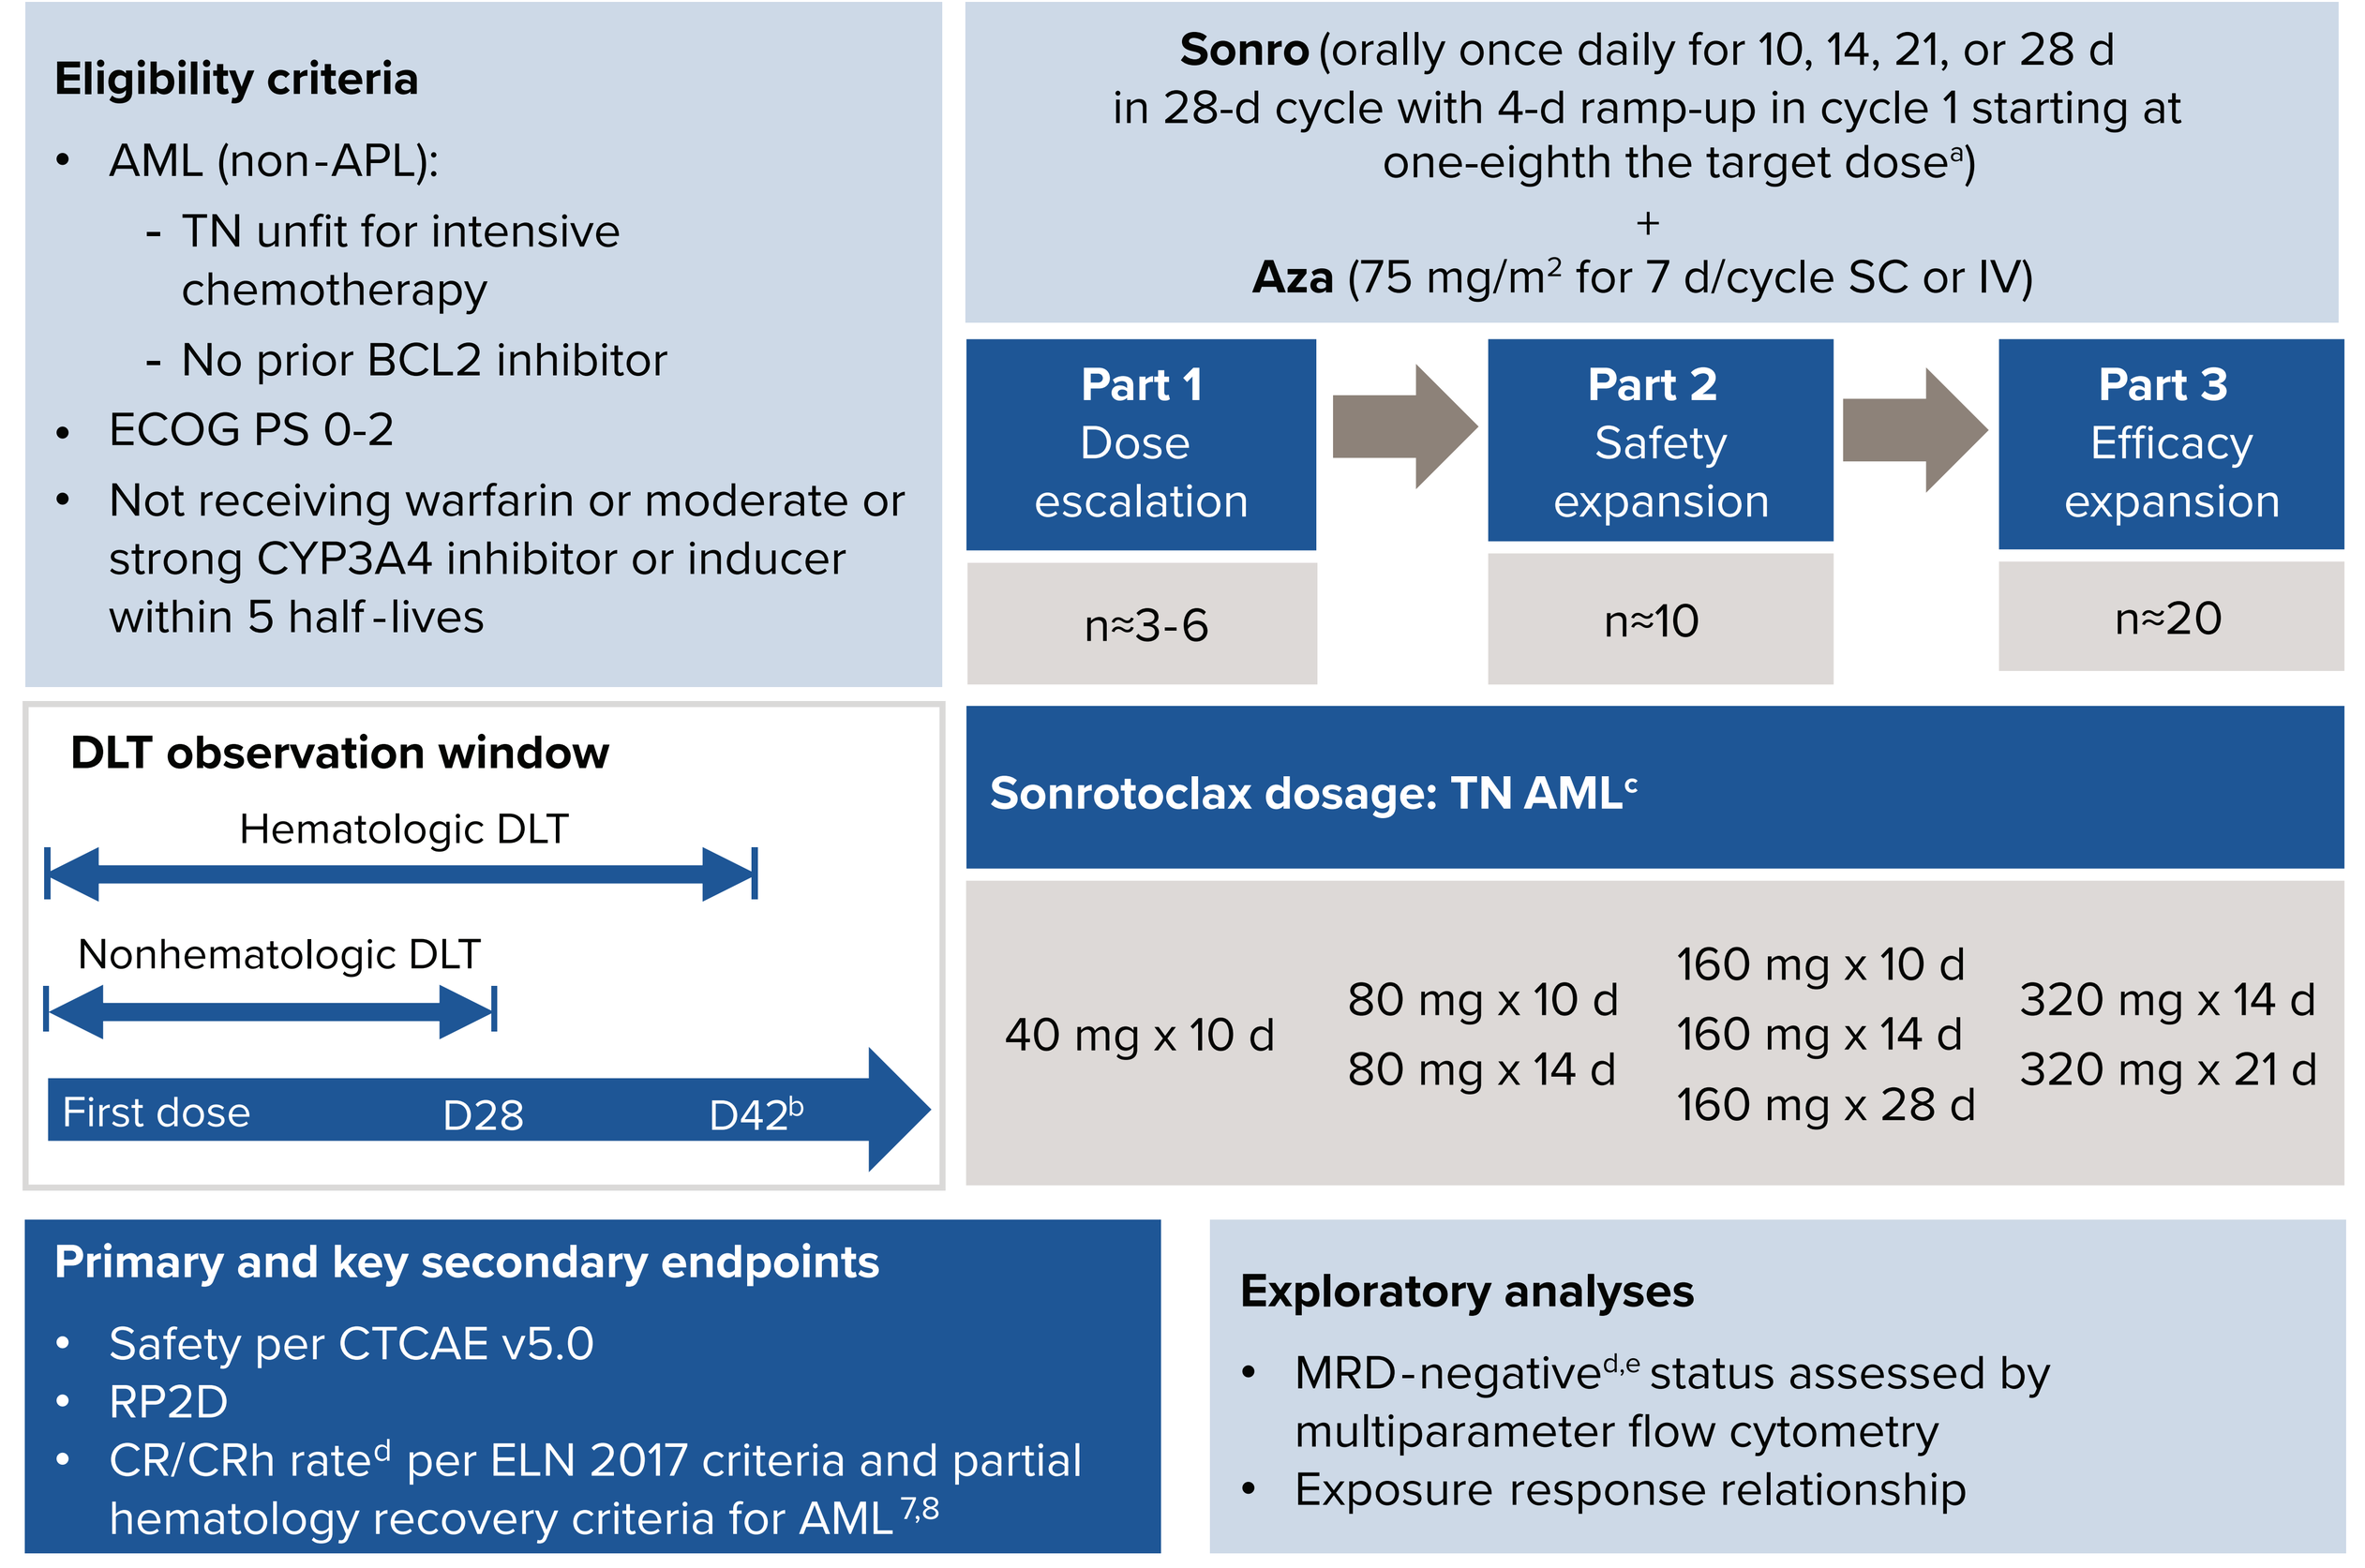
Eligibility criteria
AML (non-APL):
- TN unfit for intensive chemotherapy
- No prior BCL2 inhibitor
ECOG PS 0-2
Not receiving warfarin or moderate or strong CYP3A4 inhibitor or inducer within 5 half-lives
Sonro (orally once daily for 10, 14, 21, or 28 d in 28-d cycle with 4-d ramp-up in cycle 1 starting at one-eighth the target dose[a])
+
Aza (75 mg/m² for 7 d/cycle SC or IV)
Part 1 Dose escalation
n≈3-6
Part 2 Safety expansion
n≈10
Part 3 Efficacy expansion
n≈20
DLT observation window
Hematologic DLT
Nonhematologic DLT
First dose
D28
D42[b]
Sonrotoclax dosage: TN AML[c]
40 mg x 10 d
80 mg x 10 d
80 mg x 14 d
160 mg x 10 d
160 mg x 14 d
160 mg x 28 d
320 mg x 14 d
320 mg x 21 d
Primary and key secondary endpoints
Safety per CTCAE v5.0
RP2D
CR/CRh rate[d] per ELN 2017 criteria and partial hematology recovery criteria for AML[7,8]
Exploratory analyses
MRD-negative[d,e] status assessed by multiparameter flow cytometry
Exposure response relationship

[a]Initial 4-d ramp-up to mitigate potential risk of TLS. As a precautionary measure for TLS monitoring, patients were hospitalized during the ramp-up period. [b]Or C2 initiation. [c]Dose reductions were done first by reducing the number of dosing days/cycles of sonro. Once 10-d dosing was reached, aza dosing was reduced and then sonro daily dose. [d]Response and MRD status were assessed at end of C1 (C2 if remission not yet achieved) and then every three cycles until C13, then every six cycles. [e]MRD negative was defined as ≤1 residual leukemic blasts per 1,000 leukocytes or 10^{-3} at any time on the study.
Abbreviations: APL, acute promyelocytic leukemia; aza, azacitidine; BCL2, B-cell lymphoma 2; C, cycle, CYP3A4, cytochrome P450 3A4; D, day; DLT, dose-limiting toxicity; ECOG PS, Eastern Cooperative Oncology Group performance status; ELN, European LeukemiaNet; IV, intravenous; MRD, minimal residual disease; RP2D, recommended phase 2 dose; SC, subcutaneous; sonro, sonrotoclax; TLS, tumor lysis syndrome.

RESULTS

- As of January 10, 2025, 79 patients with TN AML were enrolled and treated with sonrotoclax + azacitidine; 19 (24.1%) remained on treatment (**Figure 2**)
- The median study follow-up was 7.7 months (m; range, 0.3-34.0 m); median age was 74 years (**Table 1**)
- The median number of study treatment cycles was four, and the median average cycle length was 34.0 days (d)
- The median relative dose intensity of sonrotoclax was >80%, except in the 160-mg × 28-d and 320-mg × 21-d cohorts

Figure 2. Patient Disposition

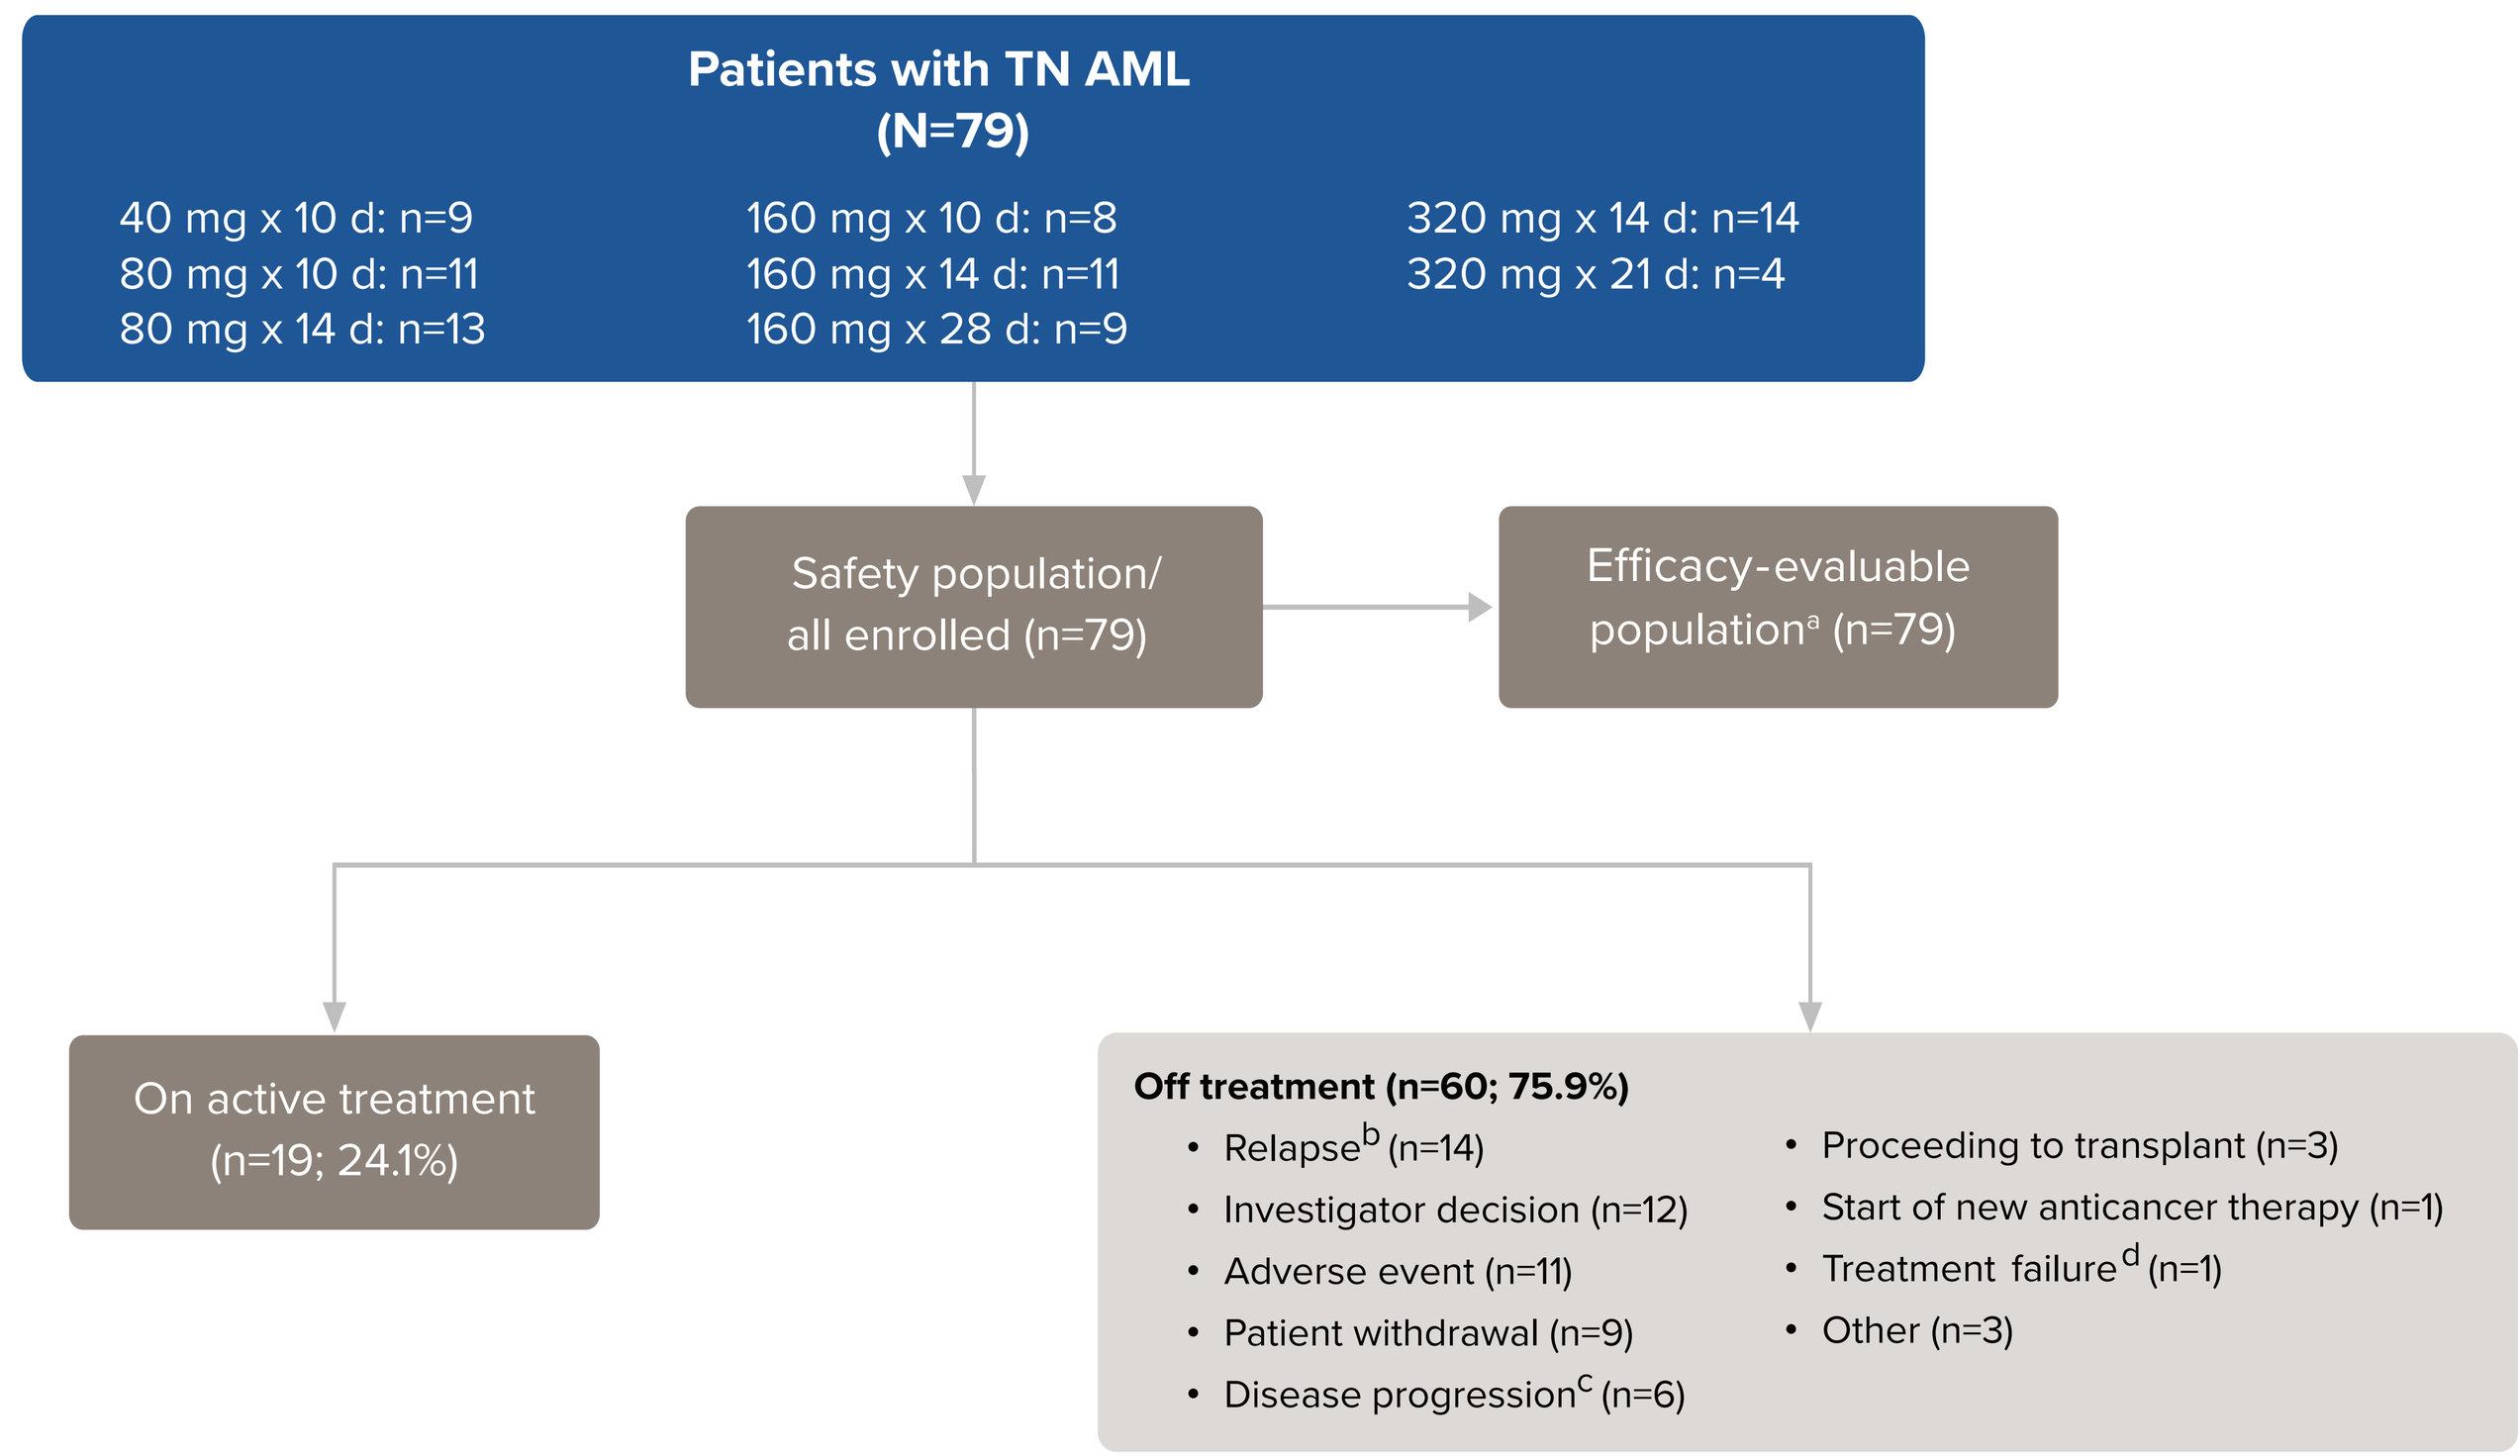
Patients with TN AML
(N=79)
40 mg x 10 d: n=9
80 mg x 10 d: n=11
80 mg x 14 d: n=13
160 mg x 10 d: n=8
160 mg x 14 d: n=11
160 mg x 28 d: n=9
320 mg x 14 d: n=14
320 mg x 21 d: n=4
Safety population/ all enrolled (n=79)
Efficacy-evaluable population[a] (n=79)
On active treatment (n=19; 24.1%)
Off treatment (n=60; 75.9%)
Relapse[b] (n=14)
Investigator decision (n=12)
Adverse event (n=11)
Patient withdrawal (n=9)
Disease progression[c] (n=6)
Proceeding to transplant (n=3)
Start of new anticancer therapy (n=1)
Treatment failure[d] (n=1)
Other (n=3)

Data cutoff: January 10, 2025. [a]Patients who (1) completed ≥1 treatment cycle (initiated the second cycle) or 42 days, whichever is earlier, or discontinued treatment during the first cycle or (2) had ≥1 response assessment. [b]Hematologic relapse (after CR/CRi) defined as bone marrow blasts ≥5%, reappearance of blasts in the blood, or development of extramedullary disease. [c]Defined as evidence of an increase in bone marrow blast percentage and/or in absolute blast counts in the blood, both per ELN 2017 response criteria. [d]Defined as no CR or CRi after six cycles of sonro + aza according to ELN 2017.
Abbreviations: CRi, CR with incomplete hematologic recovery; ELN, European LeukemiaNet.

Table 1. Baseline Patient Characteristics and Treatment Exposure in TN AML

	Sonro dose + aza								
	40 mg × 10 d (n=9)	80 mg × 10 d (n=11)	80 mg × 14 d (n=13)	160 mg × 10 d (n=8)	160 mg × 14 d (n=11)	160 mg × 28 d (n=9)	320 mg × 14 d (n=14)	320 mg × 21 d (n=4)	Total (N=79)
Follow-up, median (range), months	9.5 (0.5-38.8)	20.6 (0.3-43.4)	5.5 (0.6-10.4)	14.1 (1.4-35.1)	6.4 (1.1-8.6)	13.6 (5.1-26.9)	5.3 (3.5-15.1)	16.4 (8.8-22.0)	7.7 (0.3-43.4)
Age, median (range), years	72.0 (64-91)	77.0 (67-85)	74.0 (68-83)	78.0 (70-87)	71.0 (65-79)	70.0 (65-80)	73.0 (66-89)	76.0 (72-81)	74.0 (64-91)
Male, n (%)	6 (66.7)	5 (45.5)	7 (53.8)	6 (75.0)	6 (54.5)	7 (77.8)	12 (85.7)	3 (75.0)	52 (65.8)
AML type, n (%)									
De novo	5 (55.6)	11 (100)	12 (92.3)	6 (75.0)	8 (72.7)	6 (66.7)	11 (78.6)	3 (75.0)	62 (78.5)
Secondary	4 (44.4)	0	1 (7.7)	2 (25.0)	3 (27.3)	3 (33.3)	3 (21.4)	1 (25.0)	17 (21.5)
ELN 2017 AML risk stratification,[7] n (%)									
Favorable	0	2 (18.2)	4 (30.8)	1 (12.5)	2 (18.2)	1 (11.1)	1 (7.1)	1 (25.0)	12 (15.2)
Intermediate	4 (44.4)	4 (36.4)	6 (46.2)	2 (25.0)	3 (27.3)	3 (33.3)	6 (42.9)	3 (75.0)	31 (39.2)
Adverse	5 (55.6)	5 (45.5)	3 (23.1)	4 (50.0)	6 (54.5)	4 (44.4)	7 (50.0)	0	34 (43.0)
Positive genetic abnormality, n (%)[a]									
IDH1/ IDH2	1 (11.1)	1 (9.1)	2 (15.4)	1 (12.5)	2 (18.2)	0	1 (7.1)	0	8 (10.1)
FLT3	1 (11.1)	2 (18.2)	2 (15.4)	1 (12.5)	1 (9.1)	0	2 (14.3)	1 (25.0)	10 (12.7)
NPM1	0	3 (27.3)	3 (23.1)	1 (12.5)	0	0	1 (7.1)	1 (25.0)	9 (11.4)
TP53 aneuploidy or -17/abn(17p)	1 (11.1)	2 (18.2)	1 (7.7)	2 (25.0)	1 (9.1)	1 (11.1)	1 (7.1)	0	9 (11.4)
Treatment exposure									
No. of cycles, median (range)	4.0 (1.0-27.0)	15.0 (1.0-44.0)	4.0 (1.0-9.0)	8.5 (1.0-33.0)	3.0 (1.0-8.0)	5.0 (1.0-16.0)	3.0 (1.0-13.0)	13.0 (7.0-21.0)	4.0 (1.0-44.0)
Average cycle duration, median (range), days	32.0 (13.0-44.5)	30.0 (8.0-46.9)	29.3 (17.0-39.0)	34.4 (22.0-45.3)	39.5 (26.0-48.2)	35.1 (2.0-73.0)	36.0 (31.8-65.5)	32.4 (25.4-34.8)	34.0 (2.0-73.0)
Relative sonro dose intensity, median (range), %	100.0 (41.8-161.0)	91.5 (38.8-100.0)	100.0 (71.3-410.2)	92.6 (48.3-109.4)	90.2 (47.0-100.0)	63.3 (29.5-100.0)	83.1 (48.0-100.0)	64.4 (27.4-94.0)	90.0 (27.4-410.2)
Relative aza dose intensity, median (range), %	72.9 (35.2-101.2)	69.6 (37.2-100.4)	96.0 (83.0-100.8)	85.1 (52.3-100.2)	94.6 (61.8-102.1)	94.1 (44.3-100.2)	87.9 (43.3-99.8)	72.7 (47.5-87.4)	90.0 (35.2-102.1)

[a]As reported by investigator.
Abbreviations: aza, azacitidine; ELN, European LeukemiaNet; sonro, sonrotoclax.

- Treatment-emergent adverse event (TEAE) frequency and severity were similar across doses (**Table 2**)
- The most common any-grade and grade ≥3 TEAEs were neutropenia, infections and infestations, and thrombocytopenia (**Figure 3**)
- Tumor lysis syndrome (TLS) occurred in four patients (laboratory, n=2; clinical, n=2); all resolved in ≤4 d without sequelae (**Table 2**)
- Dose-limiting toxicities (DLTs) occurred in four patients; all were hematologic
- Twelve patients (15.2%) had a TEAE leading to death; two were treatment related (80 mg × 14 d, anemia; 160 mg × 14 d, neutropenic sepsis); the 30-d mortality rate was 3.8%
- Treatment discontinuation due to TEAEs occurred in 11 patients (13.9%)
 - The most common TEAE class leading to discontinuation of sonrotoclax (n=7, 8.9%) or azacitidine (n=7, 8.9%) was infections and infestations
- TEAEs leading to dose reduction occurred in 22 patients (27.8%) and 14 patients (17.7%) with sonrotoclax and azacitidine, respectively
 - The most common TEAE class leading to sonrotoclax (n=18, 22.8%) and azacitidine (n=12, 15.2%) dose reduction was neutropenia
- Shorter treatment schedules (<21 d) were better tolerated with relative dose intensities (RDIs) of >80%, and <30% of patients required sonrotoclax dose reduction (**Table 1**)

Table 2. TEAE Summary in TN AML

	Sonro dose + aza								
Patients, n (%)	40 mg × 10 d (n=9)	80 mg × 10 d (n=11)	80 mg × 14 d (n=13)	160 mg × 10 d (n=8)	160 mg × 14 d (n=11)	160 mg × 28 d (n=9)	320 mg × 14 d (n=14)	320 mg × 21 d (n=4)	Total (N=79)
Any TEAEs	9 (100)	11 (100)	13 (100)	8 (100)	11 (100)	9 (100)	14 (100)	4 (100)	79 (100)
Grade ≥3	9 (100)	10 (90.9)	13 (100)	8 (100)	11 (100)	9 (100)	13 (92.9)	4 (100)	77 (97.5)
Neutropenia[a]	9 (100)	9 (81.8)	12 (92.3)	8 (100)	9 (81.8)	8 (88.9)	12 (85.7)	4 (100)	71 (89.9)
Thrombocytopenia[b]	7 (77.8)	9 (81.8)	9 (69.2)	4 (50.0)	7 (63.6)	7 (77.8)	9 (64.3)	1 (25.0)	53 (67.1)
Infections and infestations	5 (55.6)	7 (63.6)	6 (46.2)	4 (50.0)	4 (36.4)	6 (66.7)	7 (50.0)	1 (25.0)	40 (50.6)
Serious TEAEs	8 (88.9)	10 (90.9)	10 (76.9)	7 (87.5)	7 (63.6)	8 (88.9)	9 (64.3)	2 (50.0)	61 (77.2)
Laboratory TLS	0	0	0	1 (12.5)	0	0	1 (7.1)	0	2 (2.5)
Clinical TLS	0	0	0	0	1 (9.1)	0	1 (7.1)	0	2 (2.5)
DLT, n/N (%)	0	2/10 (20.0)[c]	0	0	1/11 (9.1)[d]	0	1/13 (7.7)[d]	0	4/69 (5.8)
Led to death[e]	1 (11.1)	3 (27.3)	5 (38.5)	1 (12.5)	2 (18.2)	0	0	0	12 (15.2)
Led to discontinuation									
Aza	1 (11.1)	3 (27.3)	0	3 (37.5)	4 (36.4)	1 (11.1)	0	0	12 (15.2)
Sonro	2 (22.2)	3 (27.3)	0	2 (25.0)	4 (36.4)	1 (11.1)	0	0	12 (15.2)
Led to reduction									
Aza	3 (33.3)	6 (54.5)	0	1 (12.5)	0	2 (22.2)	1 (7.1)	1 (25.0)	14 (17.7)
Sonro	2 (22.2)	3 (27.3)	3 (23.1)	0	3 (27.3)	4 (44.4)	4 (28.6)	3 (75.0)	22 (27.8)
Led to interruption									
Aza	2 (22.2)	5 (45.5)	2 (15.4)	1 (12.5)	2 (18.2)	1 (11.1)	2 (14.3)	0	15 (19.0)
Sonro	1 (11.1)	5 (45.5)	6 (46.2)	2 (25.0)	2 (18.2)	6 (66.7)	4 (28.6)	0	26 (32.9)

[a]Neutropenia includes the terms *neutropenia, febrile neutropenia, neutrophil count decreased*, and *neutropenic sepsis*. [b]Thrombocytopenia includes the terms *thrombocytopenia* and *platelet count decreased*. [c]Grade 4 neutropenia, n=1; grade 4 thrombocytopenia, n=2. [d]Grade 4 thrombocytopenia. [e]Hospital-acquired pneumonia (80 mg × 10 d), neutropenic sepsis (160 mg × 10 d; related to disease), bronchopulmonary aspergillosis (80 mg × 10 d; related to disease), pulmonary sepsis without preceding confirmed pneumonia (40 mg × 10 d), metastatic squamous cell carcinoma (80 mg × 10 d), anemia (80 mg × 14 d; related to sonro, aza, and disease), coronary artery thrombosis (160 mg × 14 d), general physical health deterioration (80 mg × 14 d; related to disease), death from unknown cause (80 mg × 14 d; related to disease), neutropenic sepsis (160 mg × 14 d; related to sonro and aza), pneumonia (80 mg × 14 d; related to disease), and respiratory failure (80 mg × 14 d; related to disease).
Abbreviations: aza, azacitidine; DLT, dose-limiting toxicity; sonro, sonrotoclax; TEAE, treatment-emergent adverse event; TLS, tumor lysis syndrome.

Figure 3. TEAEs in ≥20% (All Grades) or ≥10% (Grade ≥3) of Patients With TN AML

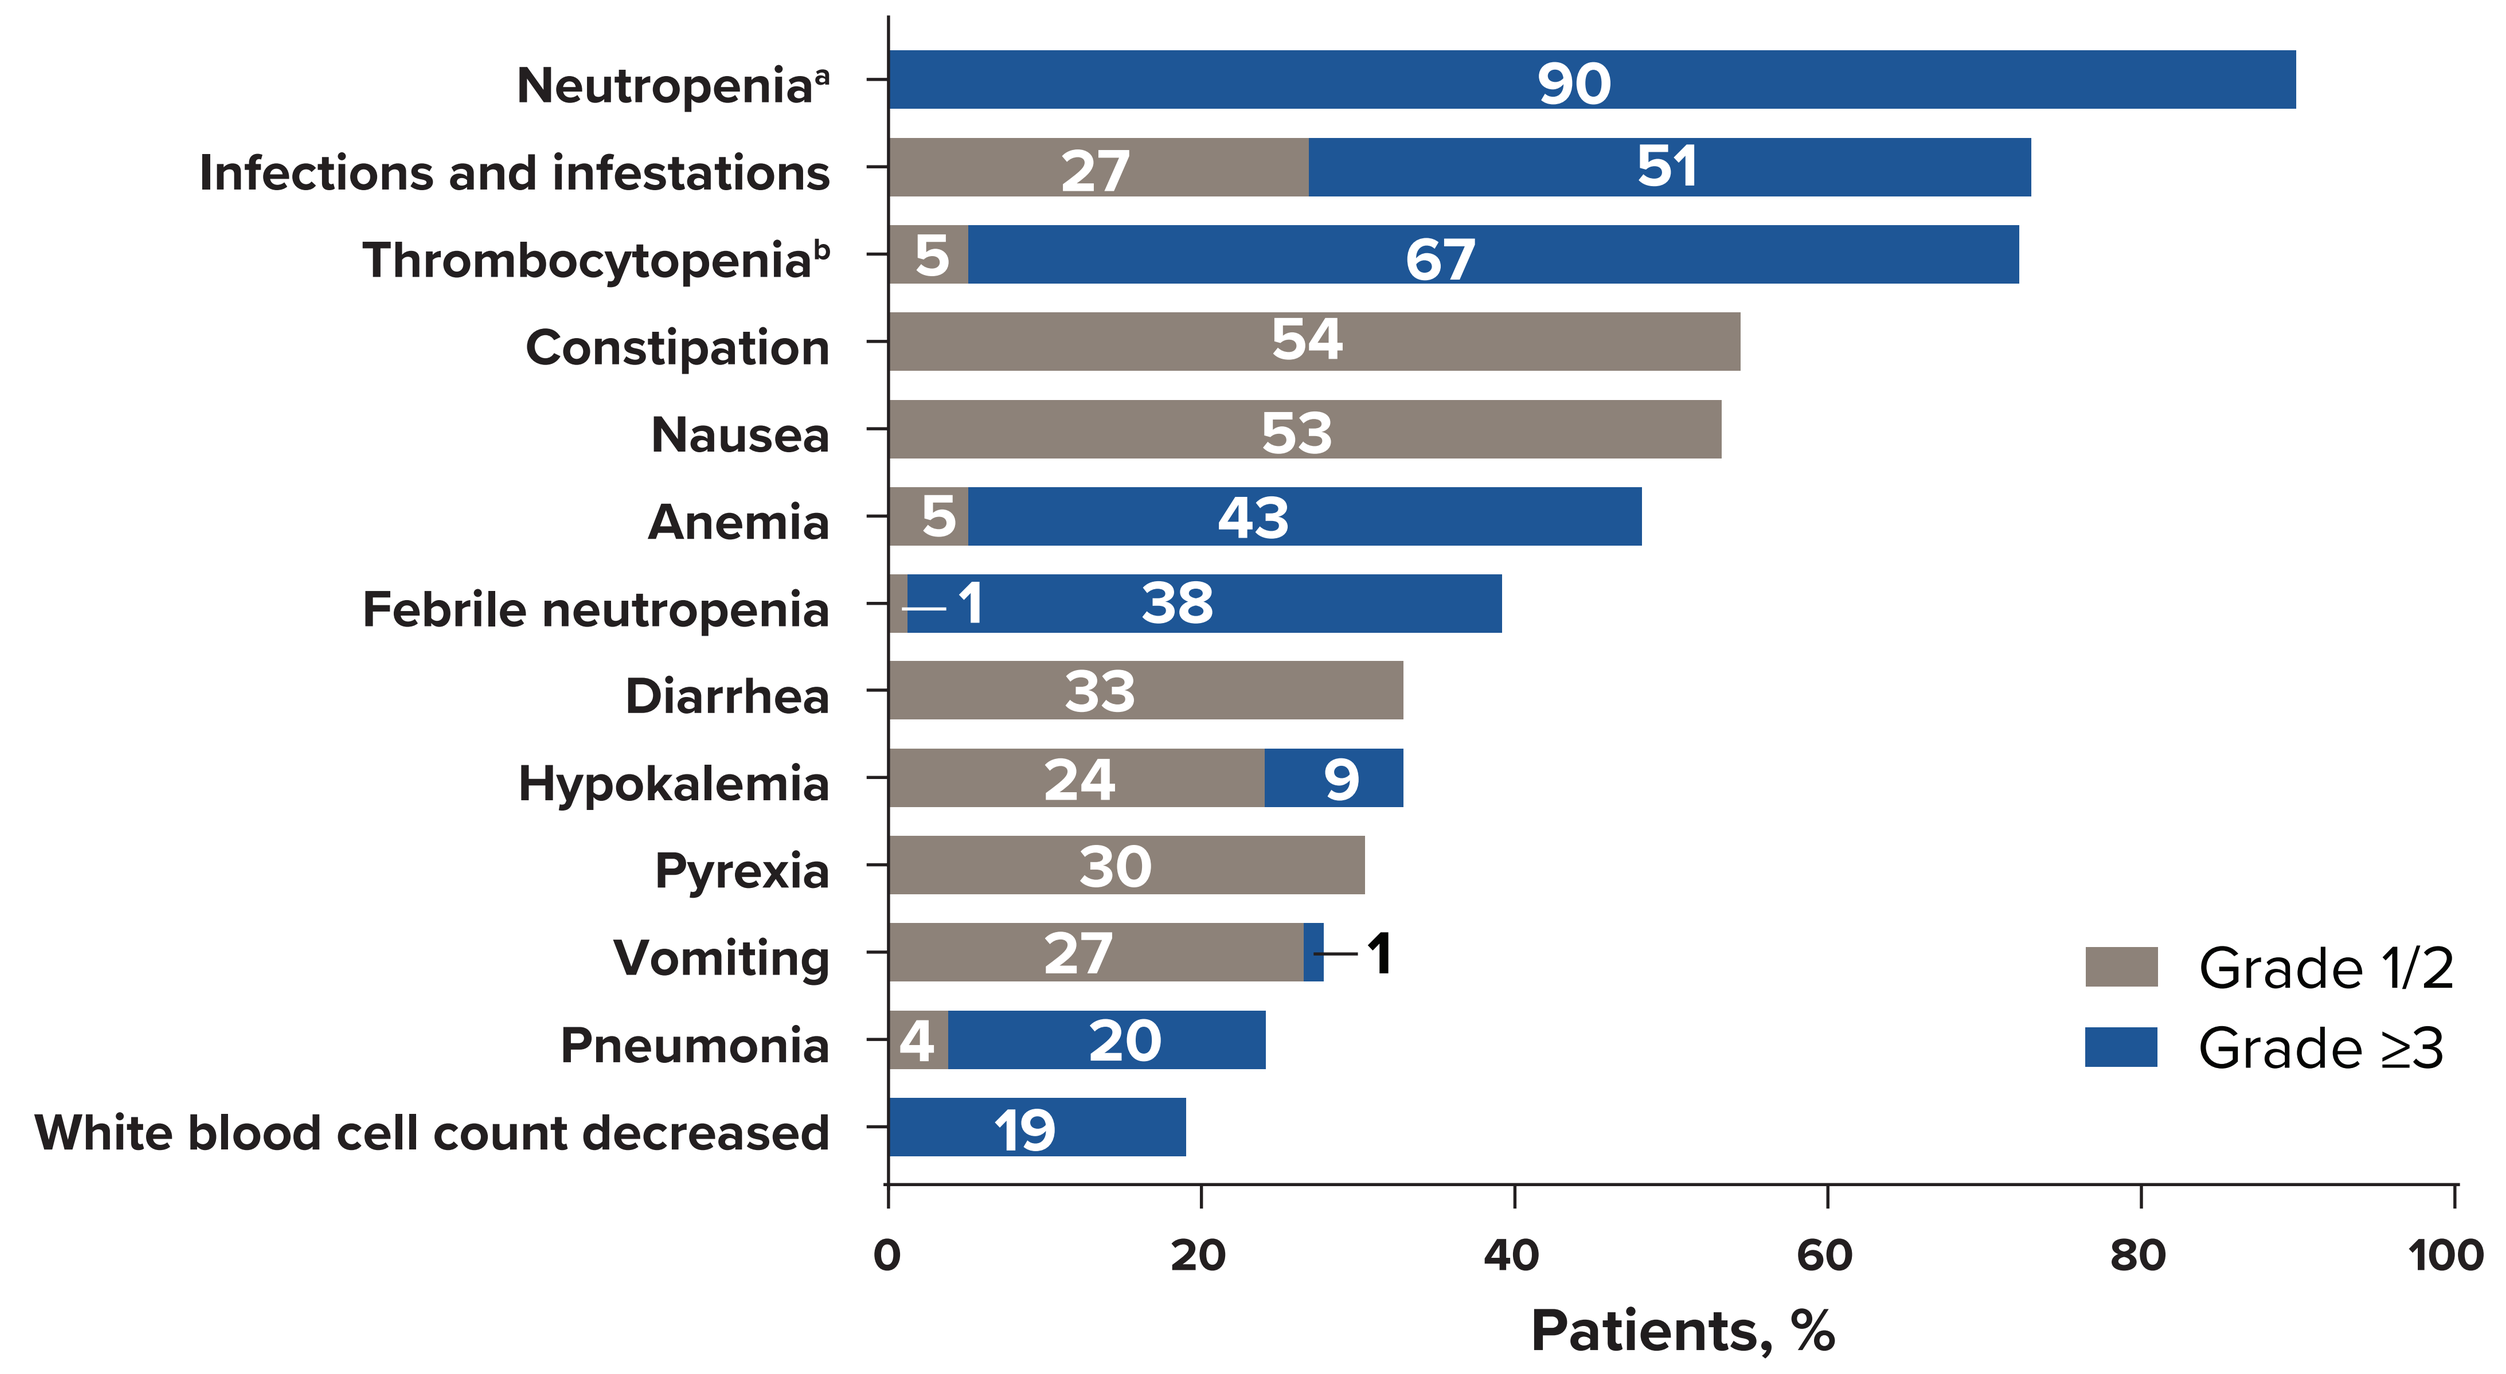
Neutropenia[a] 90
Infections and infestations 27 51
Thrombocytopenia[b] 5 67
Constipation 54
Nausea 53
Anemia 5 43
Febrile neutropenia 1 38
Diarrhea 33
Hypokalemia 24 9
Pyrexia 30
Vomiting 27 1
Pneumonia 4 20
White blood cell count decreased 19
0 20 40 60 80 100
Patients, %
Grade 1/2
Grade ≥3

[a]Neutropenia includes the terms *neutropenia, febrile neutropenia, neutrophil count decreased*, and *neutropenic sepsis*. [b]Thrombocytopenia includes the terms *thrombocytopenia* and *platelet count decreased*.
Abbreviation: TEAE, treatment-emergent adverse event.

- With a median follow-up of 7.7 m, the overall response rate (ORR) in all patients was 74.7% (**Figure 4A**)
 - Complete response (CR)/CR with partial hematologic recovery (CRh) was achieved in 59.5% (95% CI, 47.9%-70.4%) by a median of 1.3 m; CR was achieved in 50.6% (95% CI, 39.1%-62.1%) of patients by a median of 1.7 m (**Table 3** and **Figure 4B**)
 - In cohorts with the longest follow-up (40, 80, and 160 mg x 10 d), 75% of patients who achieved CR/CRh remained alive and progression free at 12 m since the first determination of response
- Minimal residual disease–negative status was achieved by 35.4% of patients (**Figure 4C**)

Table 3. Summary of Disease Responses in TN AML[a]

	Sonro dose + aza								
	40 mg × 10 d (n=9)	80 mg × 10 d (n=11)	80 mg × 14 d (n=13)	160 mg × 10 d (n=8)	160 mg × 14 d (n=11)	160 mg × 28 d (n=9)	320 mg × 14 d (n=14)	320 mg × 21 d (n=4)	Total (N=79)
CR, n (%)	4 (44.4)	8 (72.7)	5 (38.5)	4 (50.0)	5 (45.5)	4 (44.4)	6 (42.9)	4 (100)	40 (50.6)
Time to CR, median (range), months	1.3 (1.3-1.8)	1.8 (0.9-6.5)	1.7 (1.0-2.8)	2.6 (1.0-21.1)	1.2 (0.8-2.7)	3.0 (1.1-7.9)	1.3 (0.9-2.1)	7.6 (2.1-21.7)	1.7 (0.8-21.7)
By end of cycle 2, n (%)	4 (44.4)	6 (54.5)	4 (30.8)	2 (25.0)	5 (45.5)	2 (22.2)	6 (42.9)	1 (25.0)	30 (38.0)
CR/CRh, n (%)	5 (55.6)	8 (72.7)	7 (53.8)	5 (62.5)	6 (54.5)	5 (55.6)	7 (50.0)	4 (100)	47 (59.5)
Time to CR/CRh, median (range), months	1.3 (1.3-5.6)	1.4 (0.9-4.4)	1.7 (1.0-2.8)	1.2 (1.0-4.0)	1.0 (0.8-2.5)	1.2 (1.1-4.9)	1.2 (0.9-2.1)	4.1 (2.1-9.7)	1.3 (0.8-9.7)
CR/CRi, n (%)	6 (66.7)	8 (72.7)	7 (53.8)	6 (75.0)	6 (54.5)	6 (66.7)	10 (71.4)	4 (100)	53 (67.1)
Time to CR/CRi, median (range), months	1.3 (1.1-5.6)	1.4 (0.9-4.4)	1.7 (1.0-2.8)	1.1 (1.0-4.0)	1.0 (0.8-2.5)	1.5 (1.1-4.9)	1.5 (0.8-2.1)	1.8 (1.7-2.1)	1.3 (0.8-5.6)
MRD negative, n (%)	4 (44.4)	4 (36.4)	4 (30.8)	2 (25.0)	3 (27.3)	5 (55.6)	4 (28.6)	2 (50.0)	28 (35.4)
MRD NE/ND, n (%)	3 (33.3)	3 (27.3)	3 (23.1)	3 (37.5)	5 (45.5)	2 (22.2)	2 (14.3)	0	21 (26.6)

[a]Responses were determined using the ELN 2017 criteria and partial hematology recovery criteria for AML.
Abbreviations: aza, azacitidine; CR, complete response; CRh, CR with partial hematologic recovery; CRi, CR with incomplete hematologic recovery; MRD, minimal residual disease; ND, not done; NE, not estimable; sonro, sonrotoclax.

Figure 4. (A) ORR, (B) CR/CRh Rate, and (C) MRD Status in TN AML

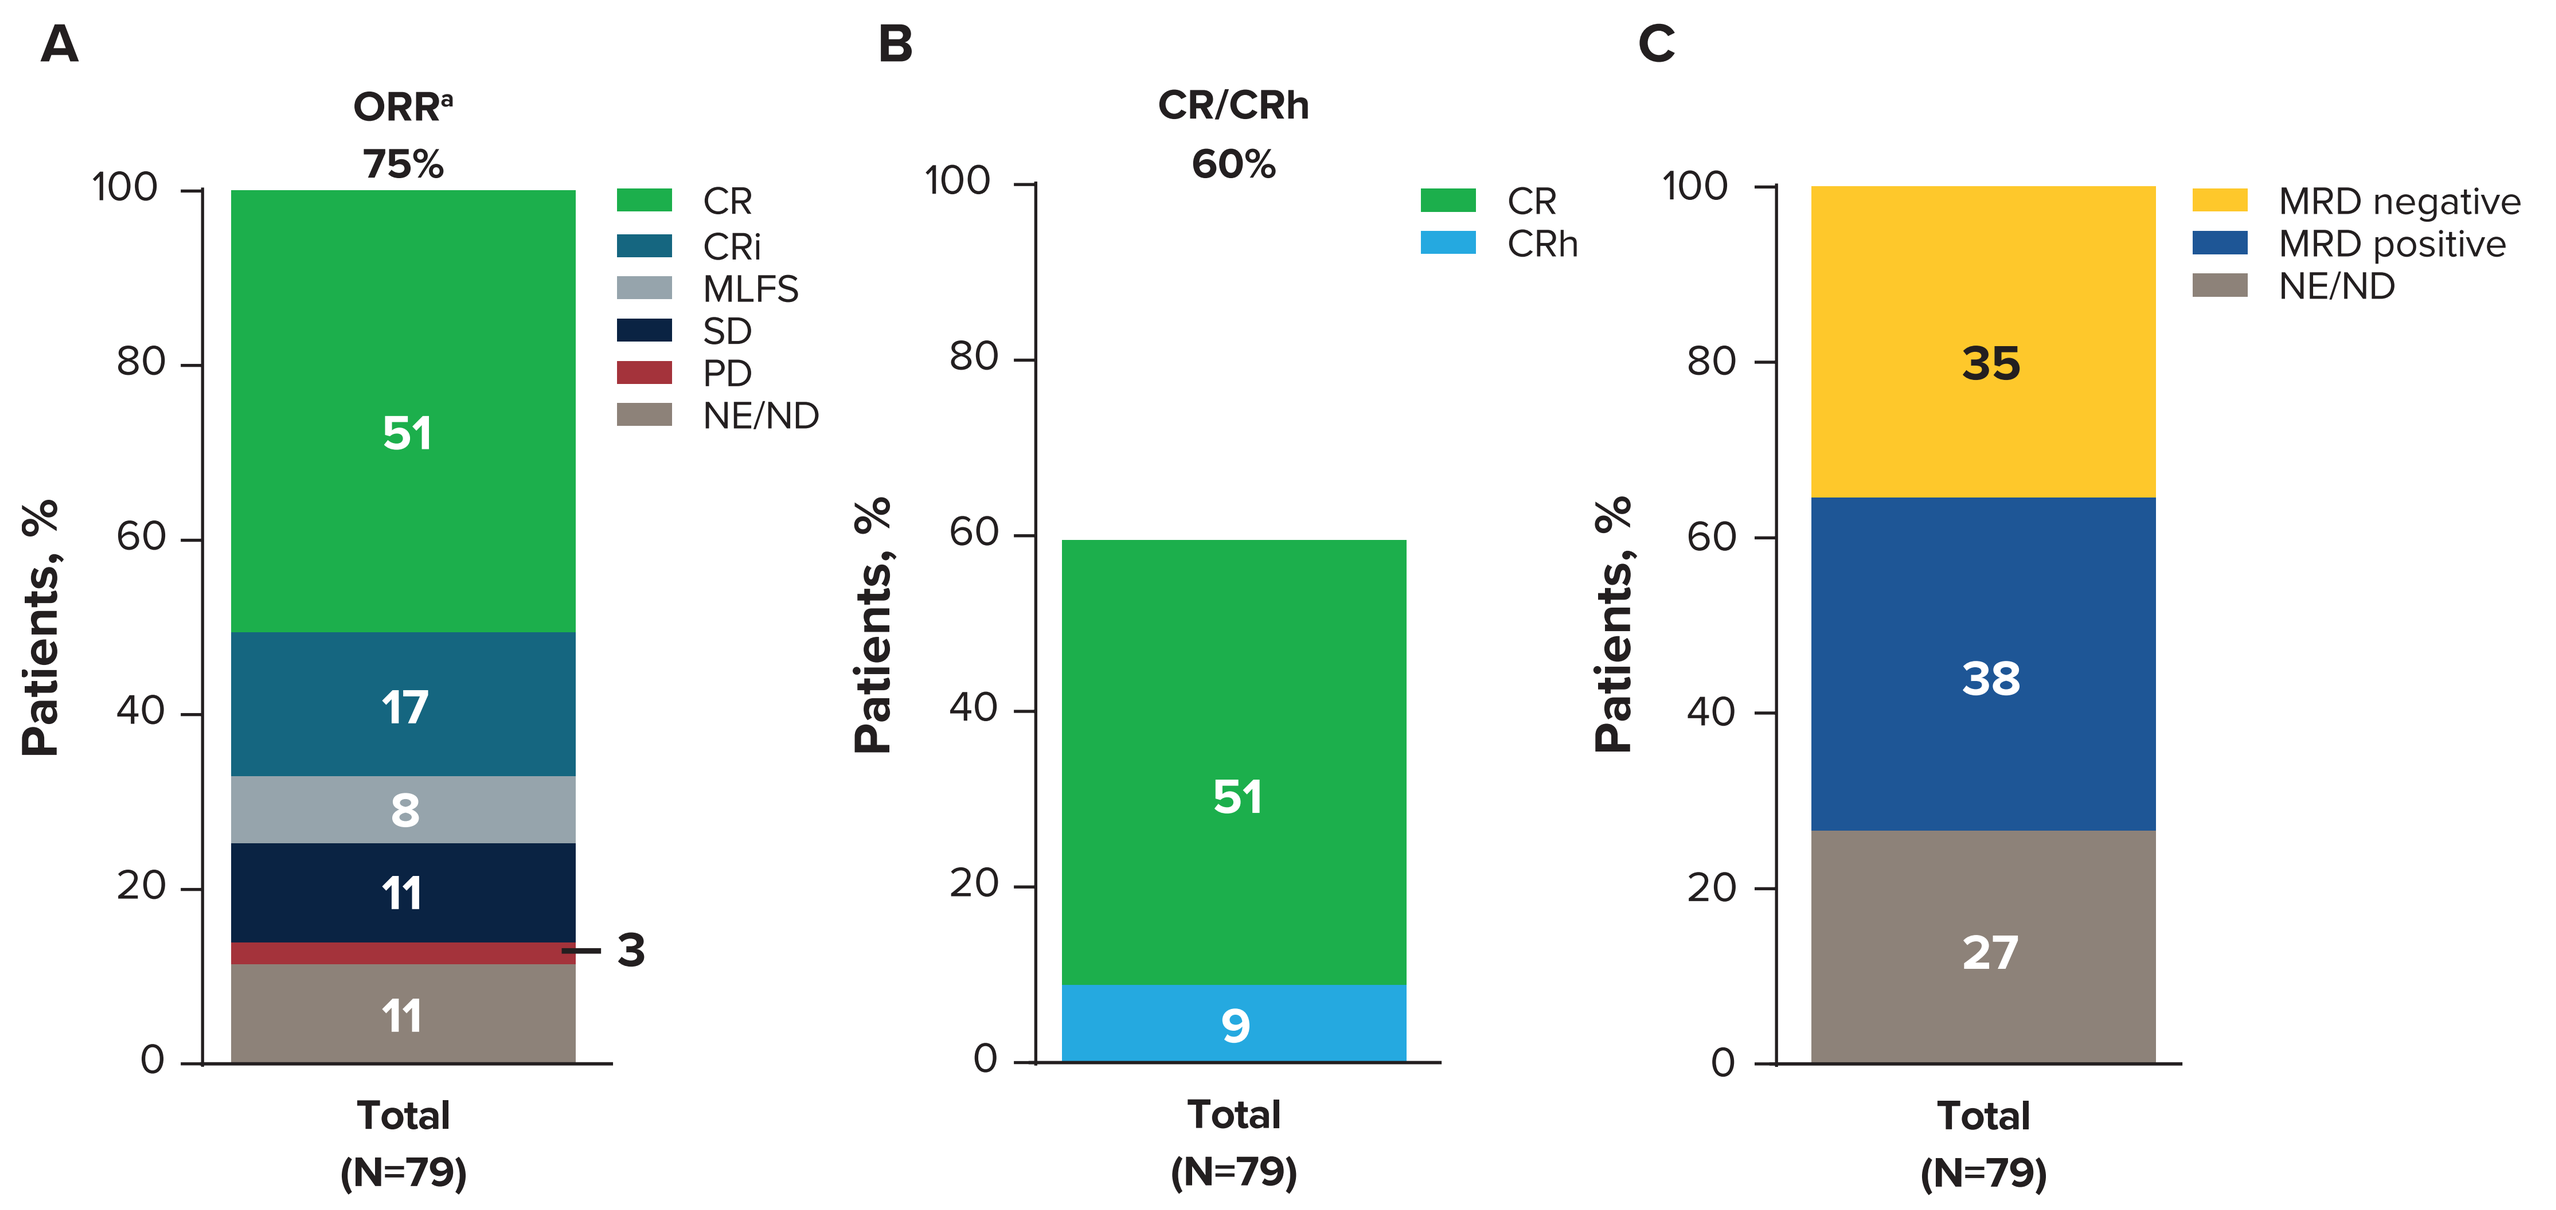
A
ORR[a]
75%
CR
CRi
MLFS
SD
PD
NE/ND
51
17
8
11
3
11
Patients, %
0 20 40 60 80 100
Total
(N=79)
B
CR/CRh
60%
CR
CRh
51
9
Patients, %
Total
(N=79)
C
MRD negative
MRD positive
NE/ND
35
38
27
Patients, %
Total
(N=79)

[a]ORR included CR, CRi, MLFS, and PR.
Abbreviations: CR, complete response; CRh, CR with partial hematologic recovery; CRi, CR with incomplete hematologic recovery; MLFS, morphologic leukemia-free state; MRD, minimal residual disease; ND, not done; NE, not evaluable; ORR, overall response rate; PD, progressive disease; SD, stable disease.

- Among 14-d cohorts with comparable follow-up, exploratory exposure-response analysis showed that the CR rate for the first tertile corresponding to pharmacokinetic exposure associated with the 80-mg dose was ≈2-fold lower than the CR rate for the second and third tertiles (**Figure 5**)

Figure 5. CR Rate by Sonrotoclax Exposure in 14-Day Cohorts

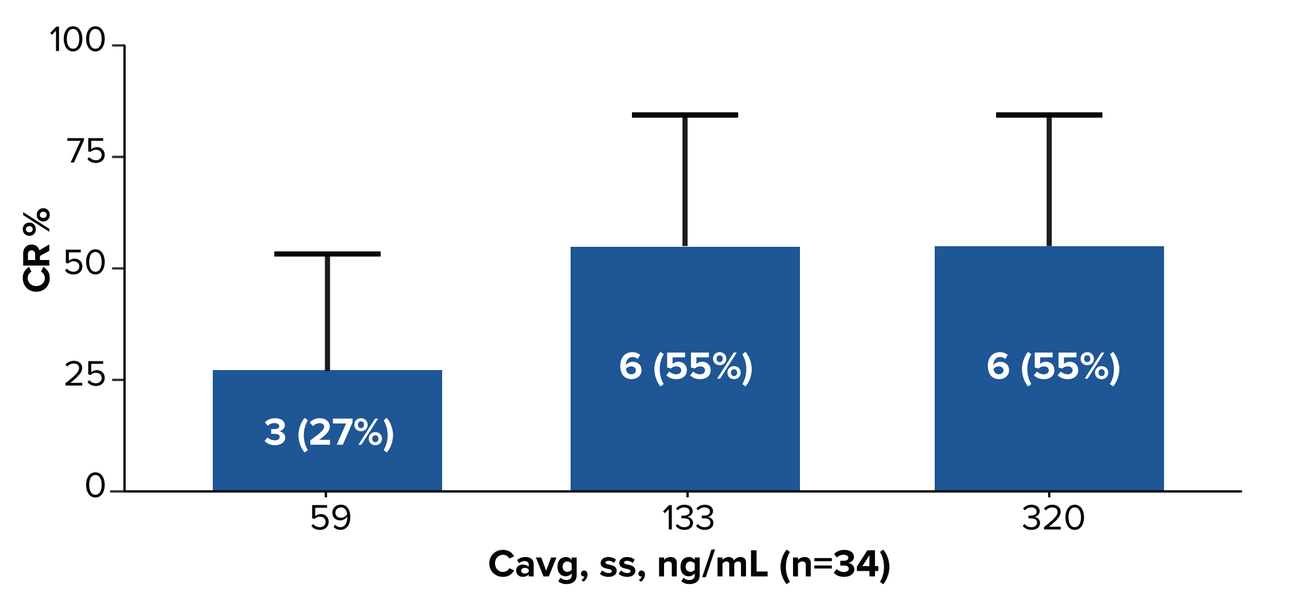
CR %
0 25 50 75 100
3 (27%)
6 (55%)
6 (55%)
59
133
320
Cavg, ss, ng/mL (n=34)

Median Cavg, ss for the 80-mg, 160-mg, and 320-mg dose levels was 60 ng/mL, 86 ng/mL, and 176 ng/mL, respectively.
Abbreviations: Cavg, ss, average sonrotoclax concentration at steady state; CR, complete response.

REFERENCES

1. Shimony S, et al. *Am J Hematol*. 2023;98(3):502-526.
2. Yi M, et al. *J Hematol Oncol*. 2020;13(1):72.
3. DiNardo CD, et al. *N Engl J Med*. 2020;383(7):617-629.
4. Thol F, Ganser A. *Curr Treat Options Oncol*. 2020:21(8):66.
5. Greiner J, et al. *Int J Mol Sci*. 2022:23(6):3304.
6. Guo Y, et al. *J Med Chem*. 2024;67(10):7836-7858.
7. Döhner H, et al. *Blood*. 2017;129(4):424-447.
8. Bloomfield CD, et al. *Blood Rev*. 2018;32(5):416-425.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines Ltd. Medical writing was provided by Brittany Gifford, PharmD, of Nucleus Global, an Inizio company, and supported by BeOne Medicines.

DISCLOSURES

JS: Consultant or advisory role: Novartis, BMS, Otsuka; Research funding: Astex/Taiho; Speakers bureau: Novartis, Mundipharma. **CYF:** Honoraria: Astellas; Consulting or advisory role: Amgen, Adaptive, Otsuka; Research funding: Jazz Pharma; Speakers bureau: Pfizer, AbbVie, Novartis, Servier. **SL:** Consulting or advisory role: BeOne Medicines Ltd; Speakers bureau: Pfizer; Travel, accommodations, expenses: BeOne Medicines Ltd, Pfizer, Janssen. **GSGM:** Honoraria: Cardinal Health, DAVA Oncology, Aptitude Health, Curio Science; Consulting or advisory board: BMS, BeOne Medicines Ltd, Pfizer, Daiichi Sankyo, Syndax, Gilead/Kite; Speakers bureau: Amgen, Rigel. **SYT:** Honoraria: Pfizer, Otsuka. **PM:** Consultant: Daiichi Sankyo, Jazz Pharma, AbbVie, Servier, Pfizer, Novartis, Kura Oncology, Syndax, Glycomimetics; Research funding, speakers bureau: Daiichi Sankyo, Jazz Pharma, AbbVie, Servier, Pfizer, Novartis. **UP:** Honoraria: Novartis, BMS, Janssen, AbbVie, Curis, Geron, Jazz Pharma, Akeso, Gilead, Servier; Consulting or advisory role: AbbVie, BMS, Janssen, Novartis, GSK, Hemavant; Speakers bureau: Novartis, BMS, Janssen, Jazz Pharma, Takeda, Sobi, Blueprint, AstraZeneca; Travel, accommodations, or expenses: AbbVie, Janssen. **CD:** Consulting or advisory role: AbbVie, Astellas, AZD, BMS, Daiichi Sankyo, Genmab, GSK, Immune Onc, Rigel, Schrodinger, Servier, Solu; Research funding: AbbVie, Astex, BMS, BeOne Medicines Ltd, Foghorn, Jazz Pharma, ImmuneOnc, Remix, Servier, Schrodinger. **SC, VM, RL, YL, OZ, AA:** Employment and may own stock: BeOne Medicines Ltd. **AHW:** Consultant: Servier, BeOne Medicines Ltd, AbbVie, Novartis; Research funding: Novartis, AbbVie, Servier, Janssen, BMS, Syndax, Astex, AstraZeneca, Amgen; Honoraria: Novartis, AstraZeneca, Astellas, Janssen, Amgen, Roche, Pfizer, AbbVie, Servier, Gilead, BMS, Shoreline, MacroGenics, Agios; Patents and royalties: Servier; Speakers bureau: AbbVie, Novartis, BMS, Servier, Astellas; Travel, accommodations, or expenses: Novartis, Servier. **XH:** No disclosures.

CORRESPONDENCE: Jake Shortt, jake.shortt@monashhealth.org

Presented at the EHA2025 Congress; June 12-15, 2025; Milan, Italy